A Phase 1 Study of Oncolytic Immunotherapy of Metastatic Neuroendocrine Tumours using Intralesional Rose Bengal Disodium: Cohort 1 Results

TJ Price [1], G Cehic [1], EA Wachter [2], I Kirkwood [3], R Sebben [1], L Leopardi [1], J Reid [1], A Alawawdeh [1], SJ Neuhaus [1] and G Maddern [1]

[1] The Queen Elizabeth Hospital, Woodville, SA AUS; [2] Provectus Biopharmaceuticals, Inc., Knoxville, TN USA; [3] Royal Adelaide Hospital, Adelaide, SA AUS.

2019 AGTIG ASM – Australasian Gastro-Intestinal Trials Group
21st Annual Scientific Meeting – 21-23 August 2019 – Adelaide

Background

Neuroendocrine tumours (NET) associated with the gastrointestinal tract are frequently indolent but troublesome as a result of endocrine secretory properties and a propensity for metastasis to the liver, nodes and lungs. Metastatic NET (mNET) located in the midgut and liver often secrete vasoactive products, giving rise to "Carcinoid Syndrome" (e.g., flushing, diarrhoea, wheezing, abdominal cramps and peripheral oedema). These symptoms are the focus of a validated quality of life instrument (EORTC QLQ-GI.NET21). Chromogranin A (CgA) is a sensitive serum biomarker for disseminated disease, while somatostatin receptor (SSTR) expression by ^{68}Ga-DOTATATE PET provides a means for radiologic assessment of tumour viability.

Treatment options for mNET include surgical resection, chemoablation, and systemic somatostatin analogues (e.g., octreotide, lantreotide) or radio-labelled analogues (e.g., Lutate/Lutathera®, ^{177}Lu DOTA-octreotate). There remains, however, a need for additional options for mNET patients. A paradigm shift in anti-cancer therapy has occurred over the last decade with the introduction of immunotherapy treatments, but immunotherapy has demonstrated limited anti-tumour activity in NET [1,2].

Rose bengal disodium (PV-10) is an oncolytic immunotherapy [3-6] undergoing clinical development for solid tumours (e.g., cutaneous melanoma, metastatic uveal melanoma, and hepatocellular carcinoma) [7-14].

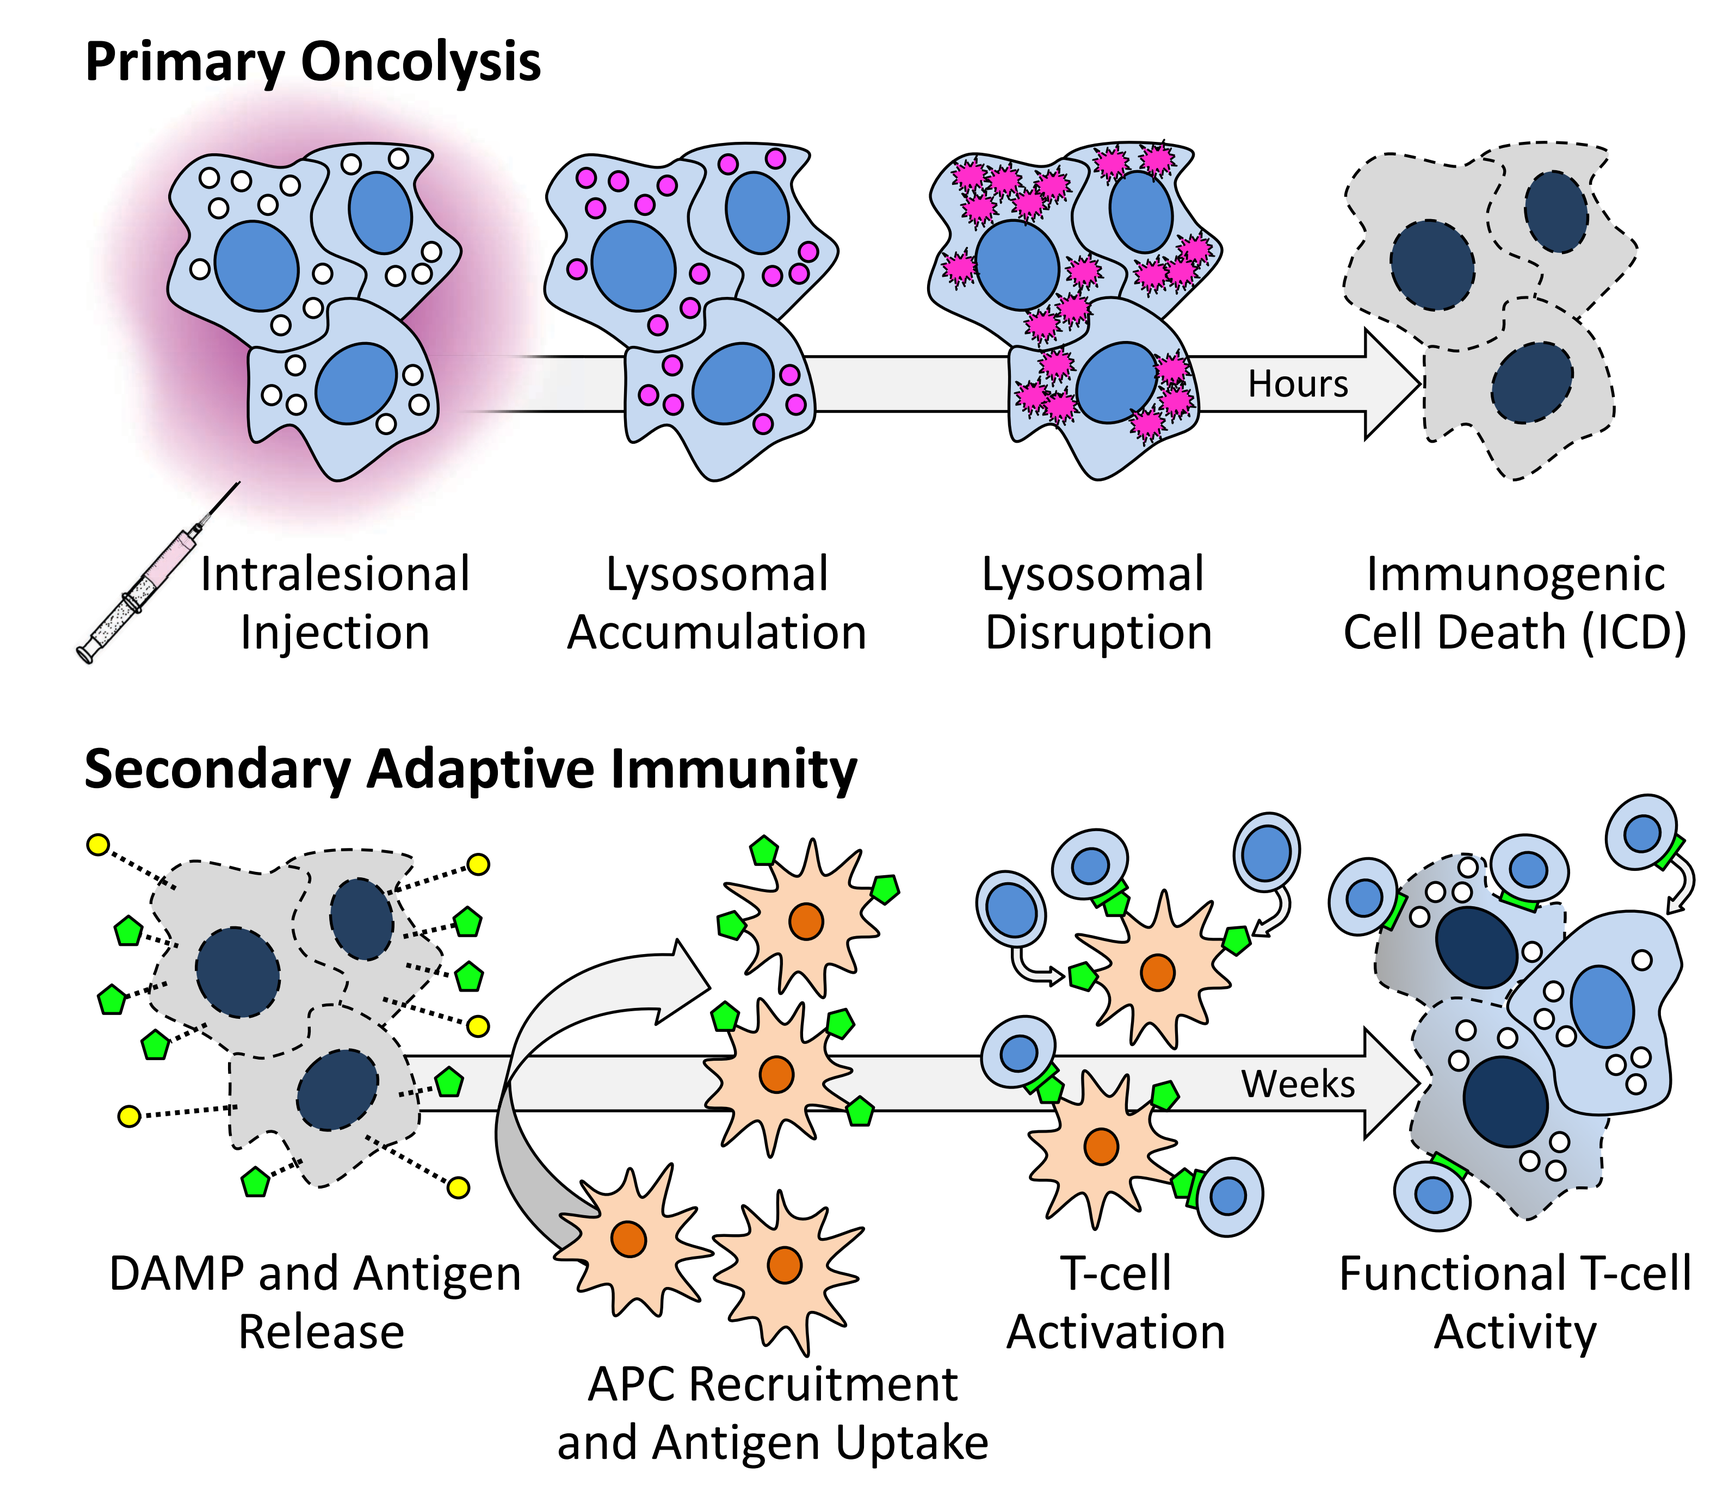
Primary Oncolysis
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Hours
Immunogenic Cell Death (ICD)
Secondary Adaptive Immunity
DAMP and Antigen Release
APC Recruitment and Antigen Uptake
T-cell Activation
Weeks
Functional T-cell Activity

1. Strosberg et al., ASCO GI 2019. **2.** Yarchoan et al., NEJM 2017; 377: 2500. **3.** Wachter et al., Proceedings of SPIE 2002; 4620: 143. **4.** Liu et al., Oncotarget 2016; 7: 37893. **5.** Qin et al., Cell Death and Disease 2017; 8: e2584. **6.** Liu et al., PLoS ONE 2018; 13: e0196033. **7.** Thompson et al., Melanoma Res 2008; 18: 405. **8.** Thompson et al., Annals Surg Oncol 2015; 22: 2135. **9.** Lippey et al., J Surg Oncol 2016; 114: 380. **10.** Foote et al., J Surg Oncol 2017; 115: 891. **11.** Read et al., J Surg Oncol 2018; 117: 579. **12.** Agarwala et al., ASCO 2019 (abstract 9559). **13.** Goldfarb et al., CIO 2017. **14.** Patel et al., ISOO Biennial Conference 2019.

Methods

This single-centre phase 1 study (protocol **PV-10-NET-01**, ClinicalTrials.gov Identifier **NCT02693067**) is evaluating the safety, tolerability, and reduction of biochemical markers and symptoms resulting from percutaneous intralesional administration of PV-10 in 12 subjects with progressive mNET with hepatic lesions not amenable to resection or other potentially curative therapy. Target lesion(s) are defined by the interventional radiologist and must be 1.0 - 3.9 cm in longest diameter.

There are two sequential dose escalation cohorts (up to 6 subjects in each) based on number of discrete interventions. Cohort 1 subjects receive PV-10 to a single hepatic lesion per treatment cycle, and can receive PV-10 to additional uninjected hepatic lesions ≥6 weeks after prior injection. Cohort 2 subjects may receive injection of multiple lesions per treatment cycle.

Cohort	Number of Subjects	PV-10 Dose per Lesion Volume	Maximum Number of Lesions Injected per Treatment Cycle	Maximum PV-10 Dose per Treatment Cycle
1	6	0.5 mL / cm^3	1 in a single segment	15 mL
2	6	0.5 mL / cm^3	1 or more	15 mL

Disease evaluations are performed at screening, week 6, and months 3 and 6 after PV-10 injection.

The Primary Endpoint is Safety:

- Safety will be established in Cohort 1 if no more than 1 of the 6 subjects experiences a dose-limiting toxicity (DLT), defined as onset of any CTCAE Grade 3 or greater non-haematological (excluding fatigue) or Grade 4 haematological toxicity within 28 days of PV-10 administration that is persistent for 14 days or longer. If 2 or more subjects experience a DLT, then PV-10 injection will be judged to be intolerable.

Secondary Endpoints:

- Objective response rate (ORR) of injected Target and bystander lesions; ORR assessed by contrast-enhanced CT.
- Target lesion SSTR expression by ^{68}Ga-DOTATATE PET; standardised uptake value (SUV) used as a surrogate for tumour viability.
- Changes in biochemical response (serum biomarker CgA).
- Symptom assessment using patient-reported outcome (QLQ-GI.NET21).
- Changes in peripheral blood mononuclear cells (PBMCs) vs baseline.

Subject Characteristics

Cohort 1 (N = 6) has Fully Enrolled:

- 4 of 6 subjects male
- Median age 65 yrs (range 47-72)
- Primary site: small intestine (N = 3) , pancreas (N = 2), caecal (N = 1)
- Grade: Gd1 (N = 5), Gd2 (N = 1)
- All subjects received prior SSA and PRRT
- Median CgA at baseline was 645 (range 30-2819)

Results: Clinical and Biomarker Outcomes

Study Treatments and Safety:

- To date 1 subject in Cohort 1 has received 4 PV-10 treatment cycles, 1 has received 2 cycles, and 4 have received a single cycle
- Median dose/cycle = 2.1 mL PV-10, range = 1.0 – 5.8 mL
- Toxicity has been acceptable, including pain post-procedure, carcinoid flare and nausea
- LFTs have remained stable

Clinical and Biomarker Outcomes:

- Overall QOL score was stable for 5 of 6 subjects
- CgA response: 5 stable, 1 progression
- One subject with "carcinoid pellagra" had rash resolution
- PBMC data not currently analysed (insufficient data at cutoff date)

Results: CT and PET/CT Assessment

Rose bengal disodium is a tetraiodinated fluorescein derivative (4,5,6,7-tetrachloro-2',4',5',7'-tetraiodofluorescein disodium). The presence of 4 iodides facilitates visualization on CT during administration and follow-up.

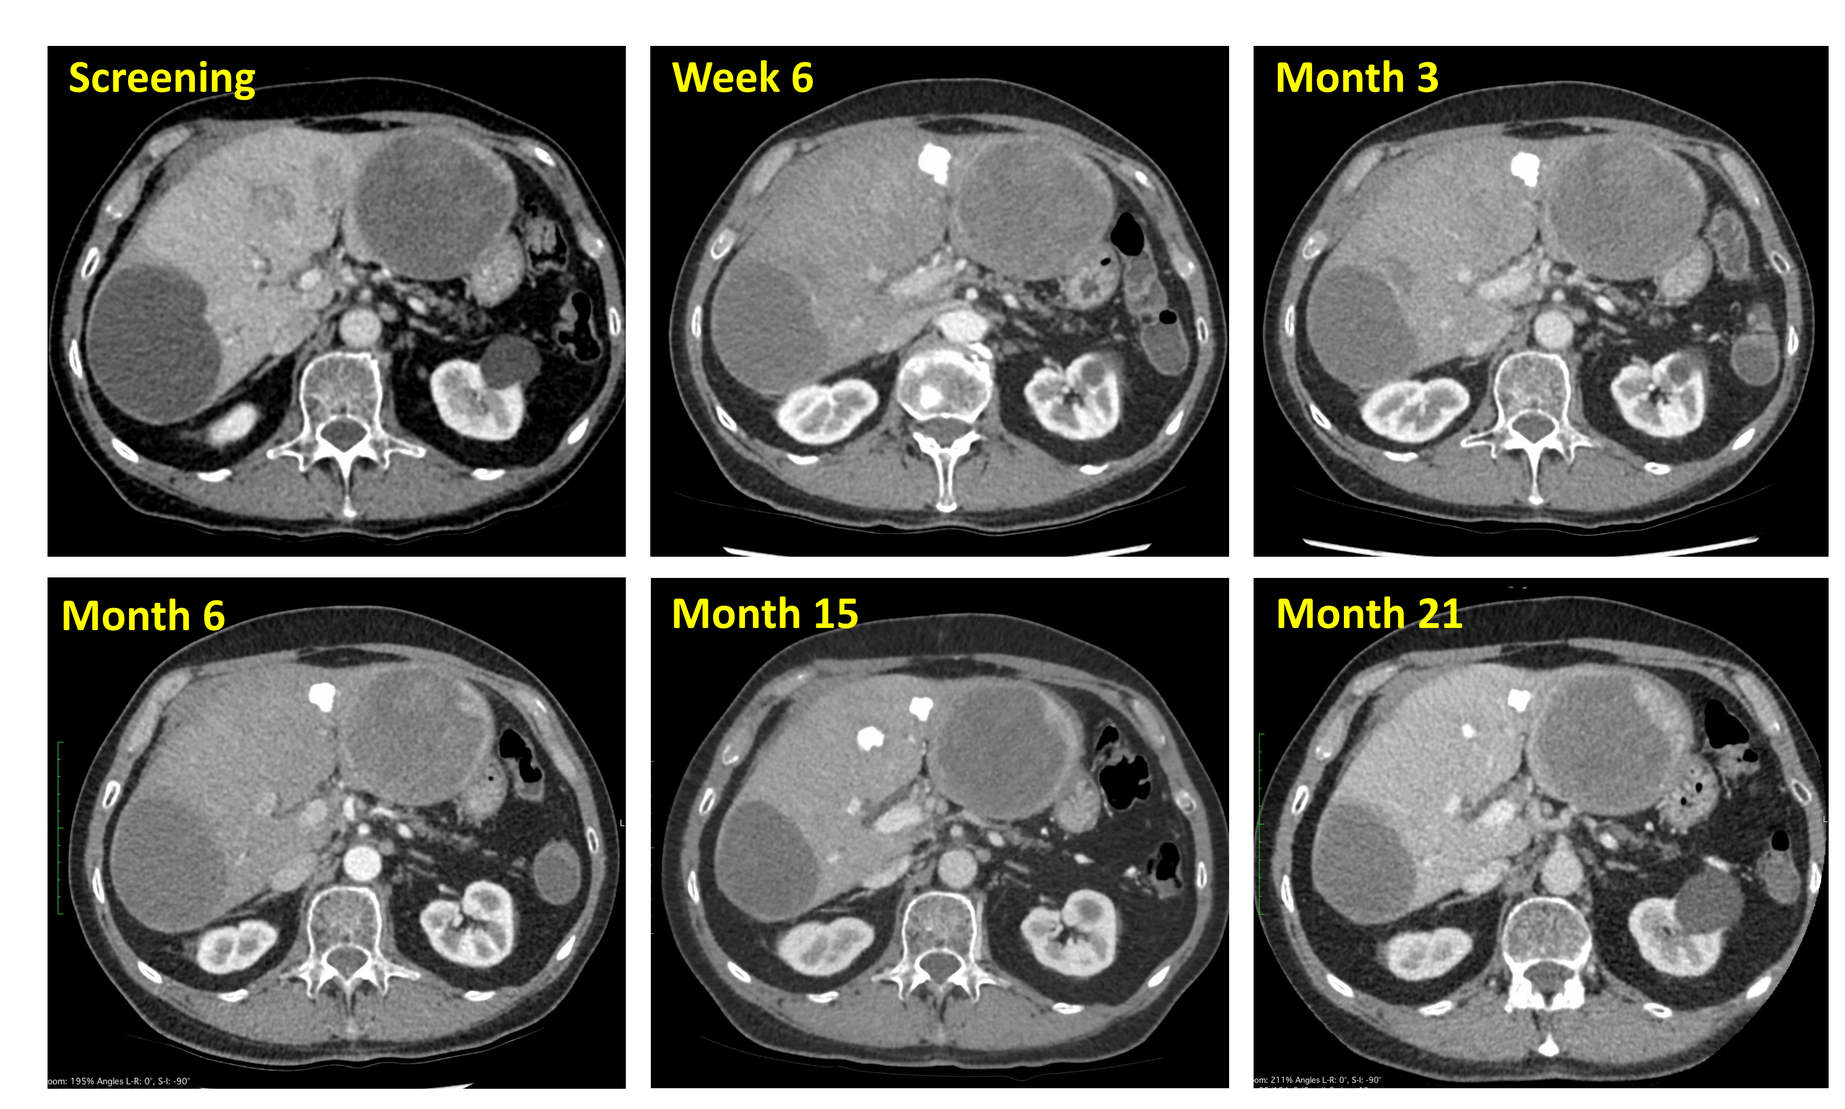
Screening
Week 6
Month 3
Month 6
Month 15
Month 21

Subject 0101: Male age 63, multifocal hepatic mNET (small intestine primary) with symptomatic progression on 30 mg Lutate (^{177}Lu DOTA-octreotate). First tumour injected with 5.3 mL PV-10 on 5 Apr 2017. Follow-up at 6 weeks to 21 months illustrates retention of PV-10 in injected tumour. Retention in adjacent tumour (injected in 3rd treatment cycle on 8 Jun 2018) evident in Month 15 and 21 scans.

Retention of PV-10 at Injection Site

Visit	Date	ROI (cm^3)	SUV_{MAX} (HU)	SUV_{SUM} (HU)
Screening	03 Mar 2017	--	--	--
Week 6	15 May 2017	5.8	936	1,126k
Month 3	11 Jul 2017	4.7	875	858k
Month 6	19 Oct 2017	4.0	819	793k
Month 14	08 Jun 2018	2.1	722	416k
Month 20	23 Nov 2018	1.9	699	374k
Month 25	13 May 2019	1.5	762	272k

Measurements using spherical ROI on non-contrast CT

Fused PET/CT illustrates concordance between injected lesion and local response (**Subject 0101**).

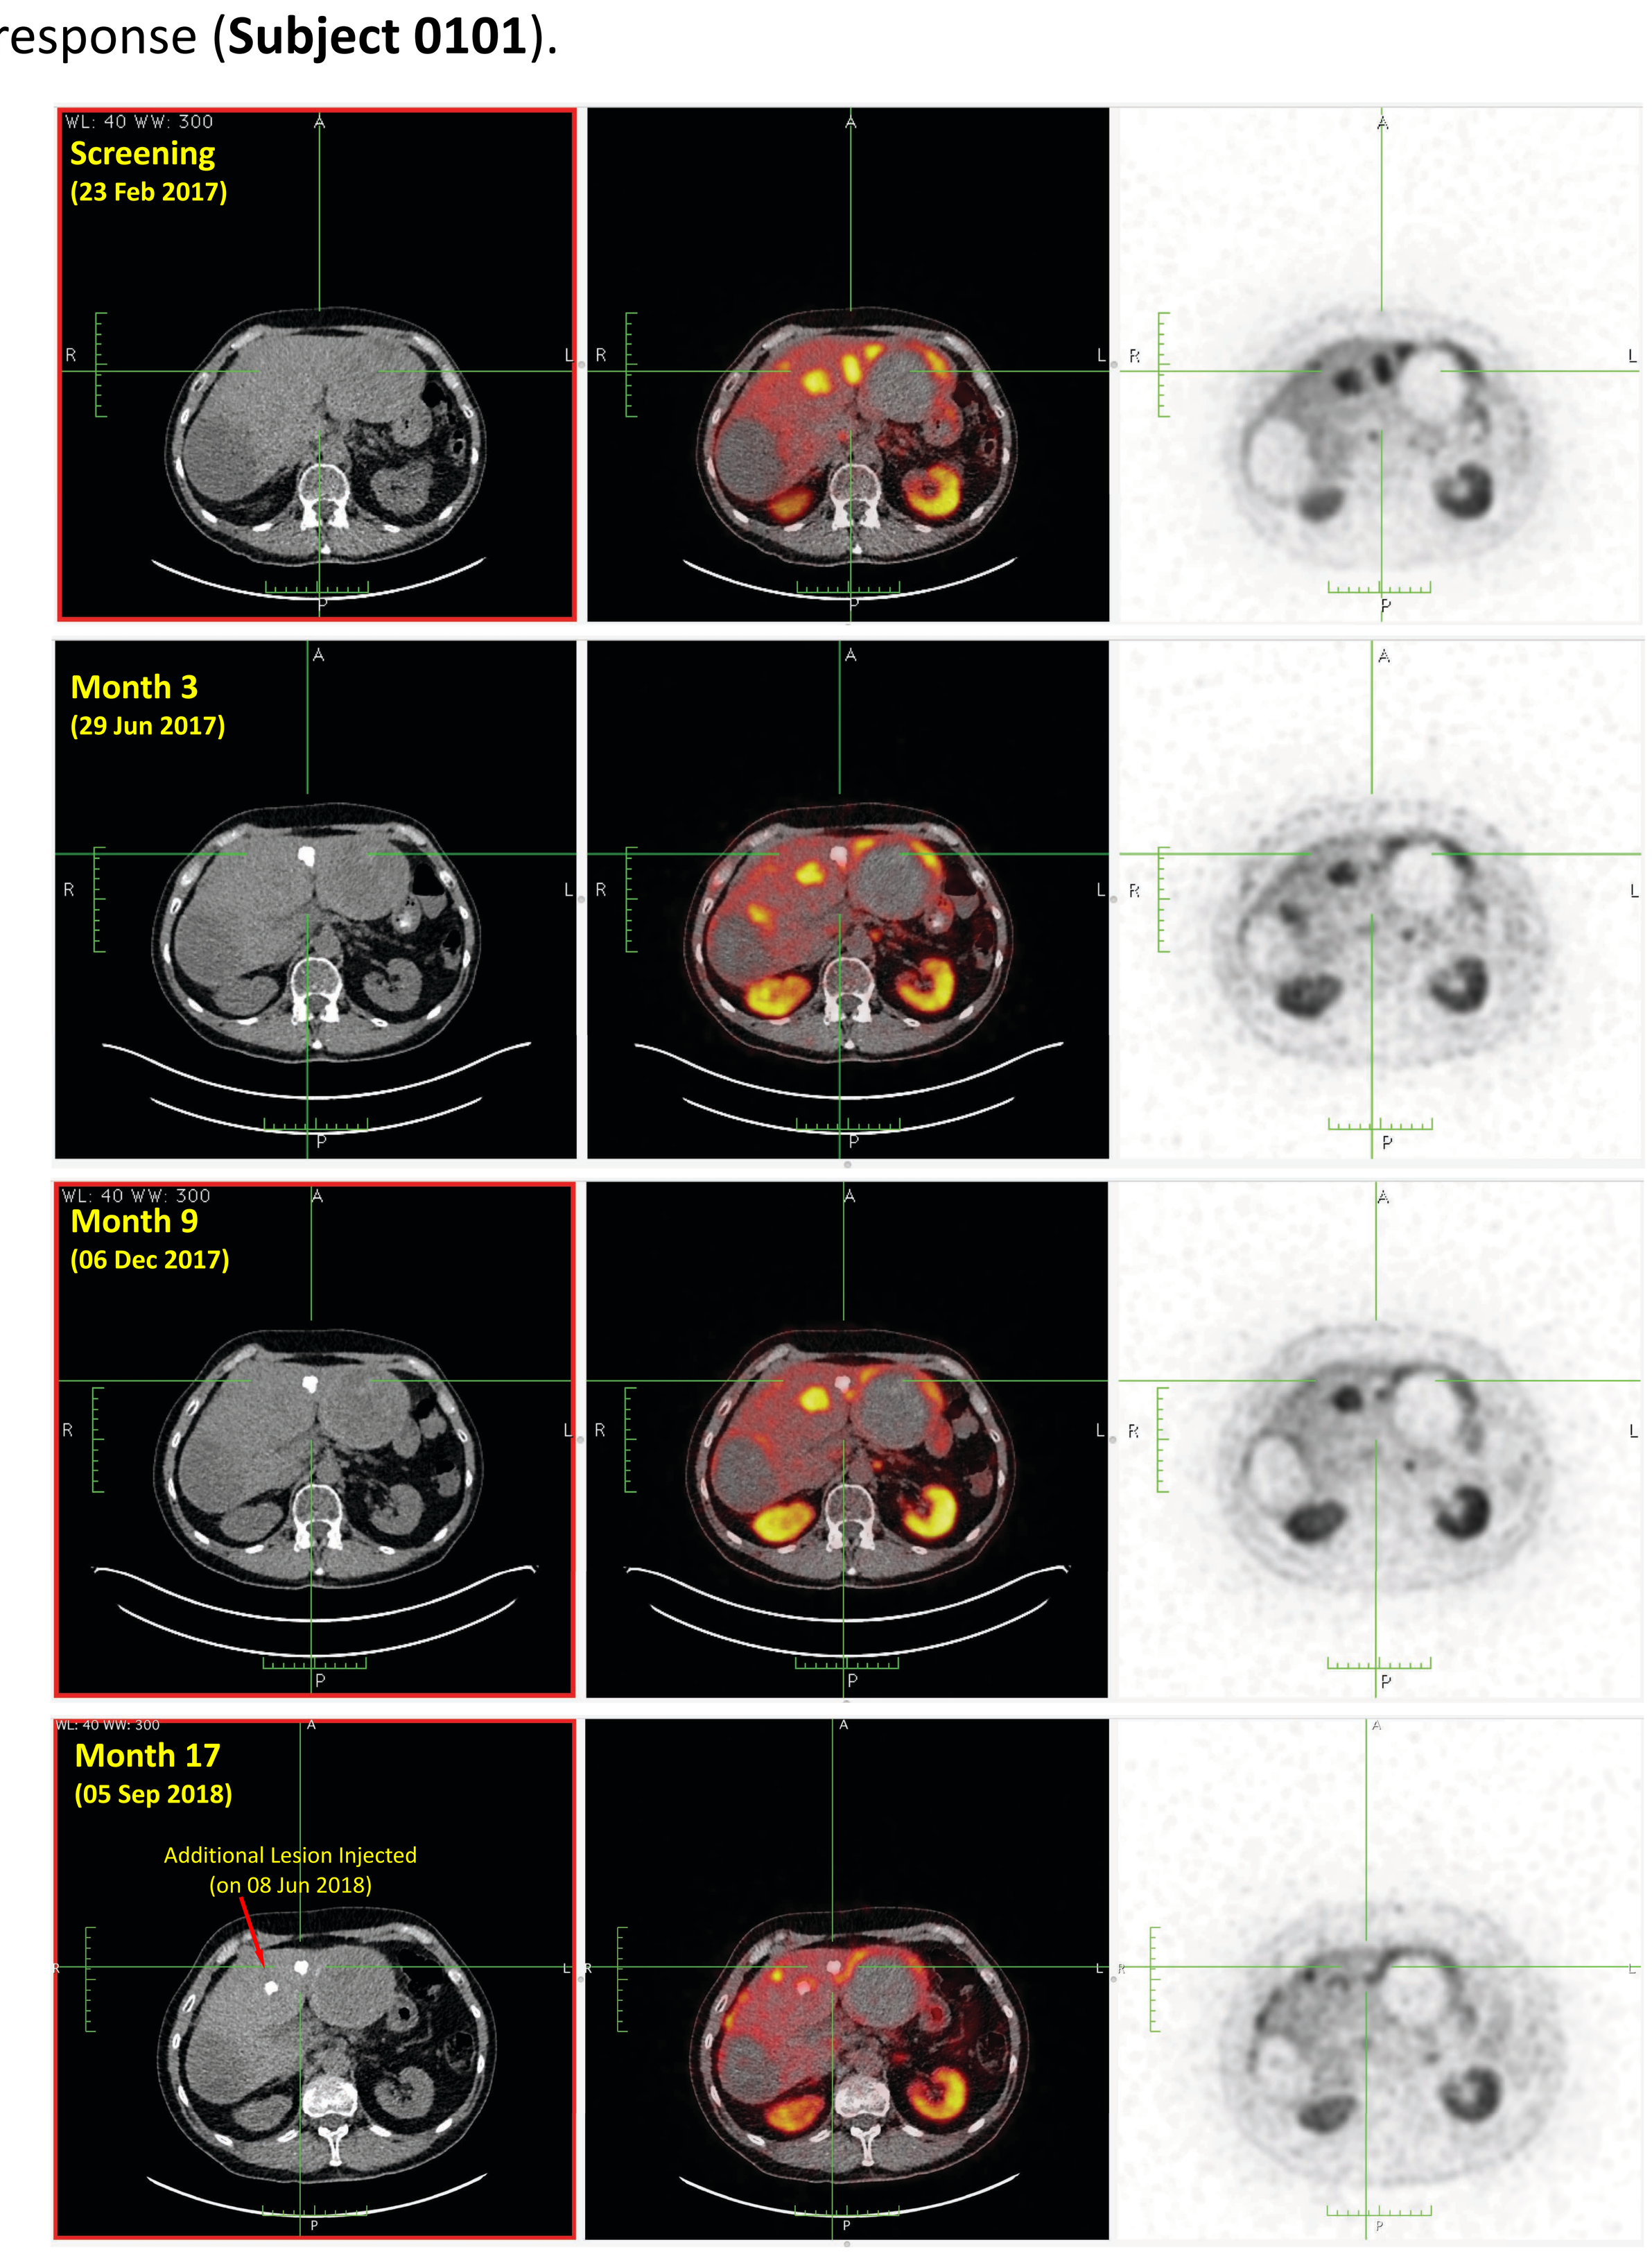
Screening
(23 Feb 2017)
Month 3
(29 Jun 2017)
Month 9
(06 Dec 2017)
Month 17
(05 Sep 2018)
Additional Lesion Injected
(on 08 Jun 2018)

Results: Assessment of Change is SSTR

Maximum intensity projection (**MIP**) illustrates local and systemic response.

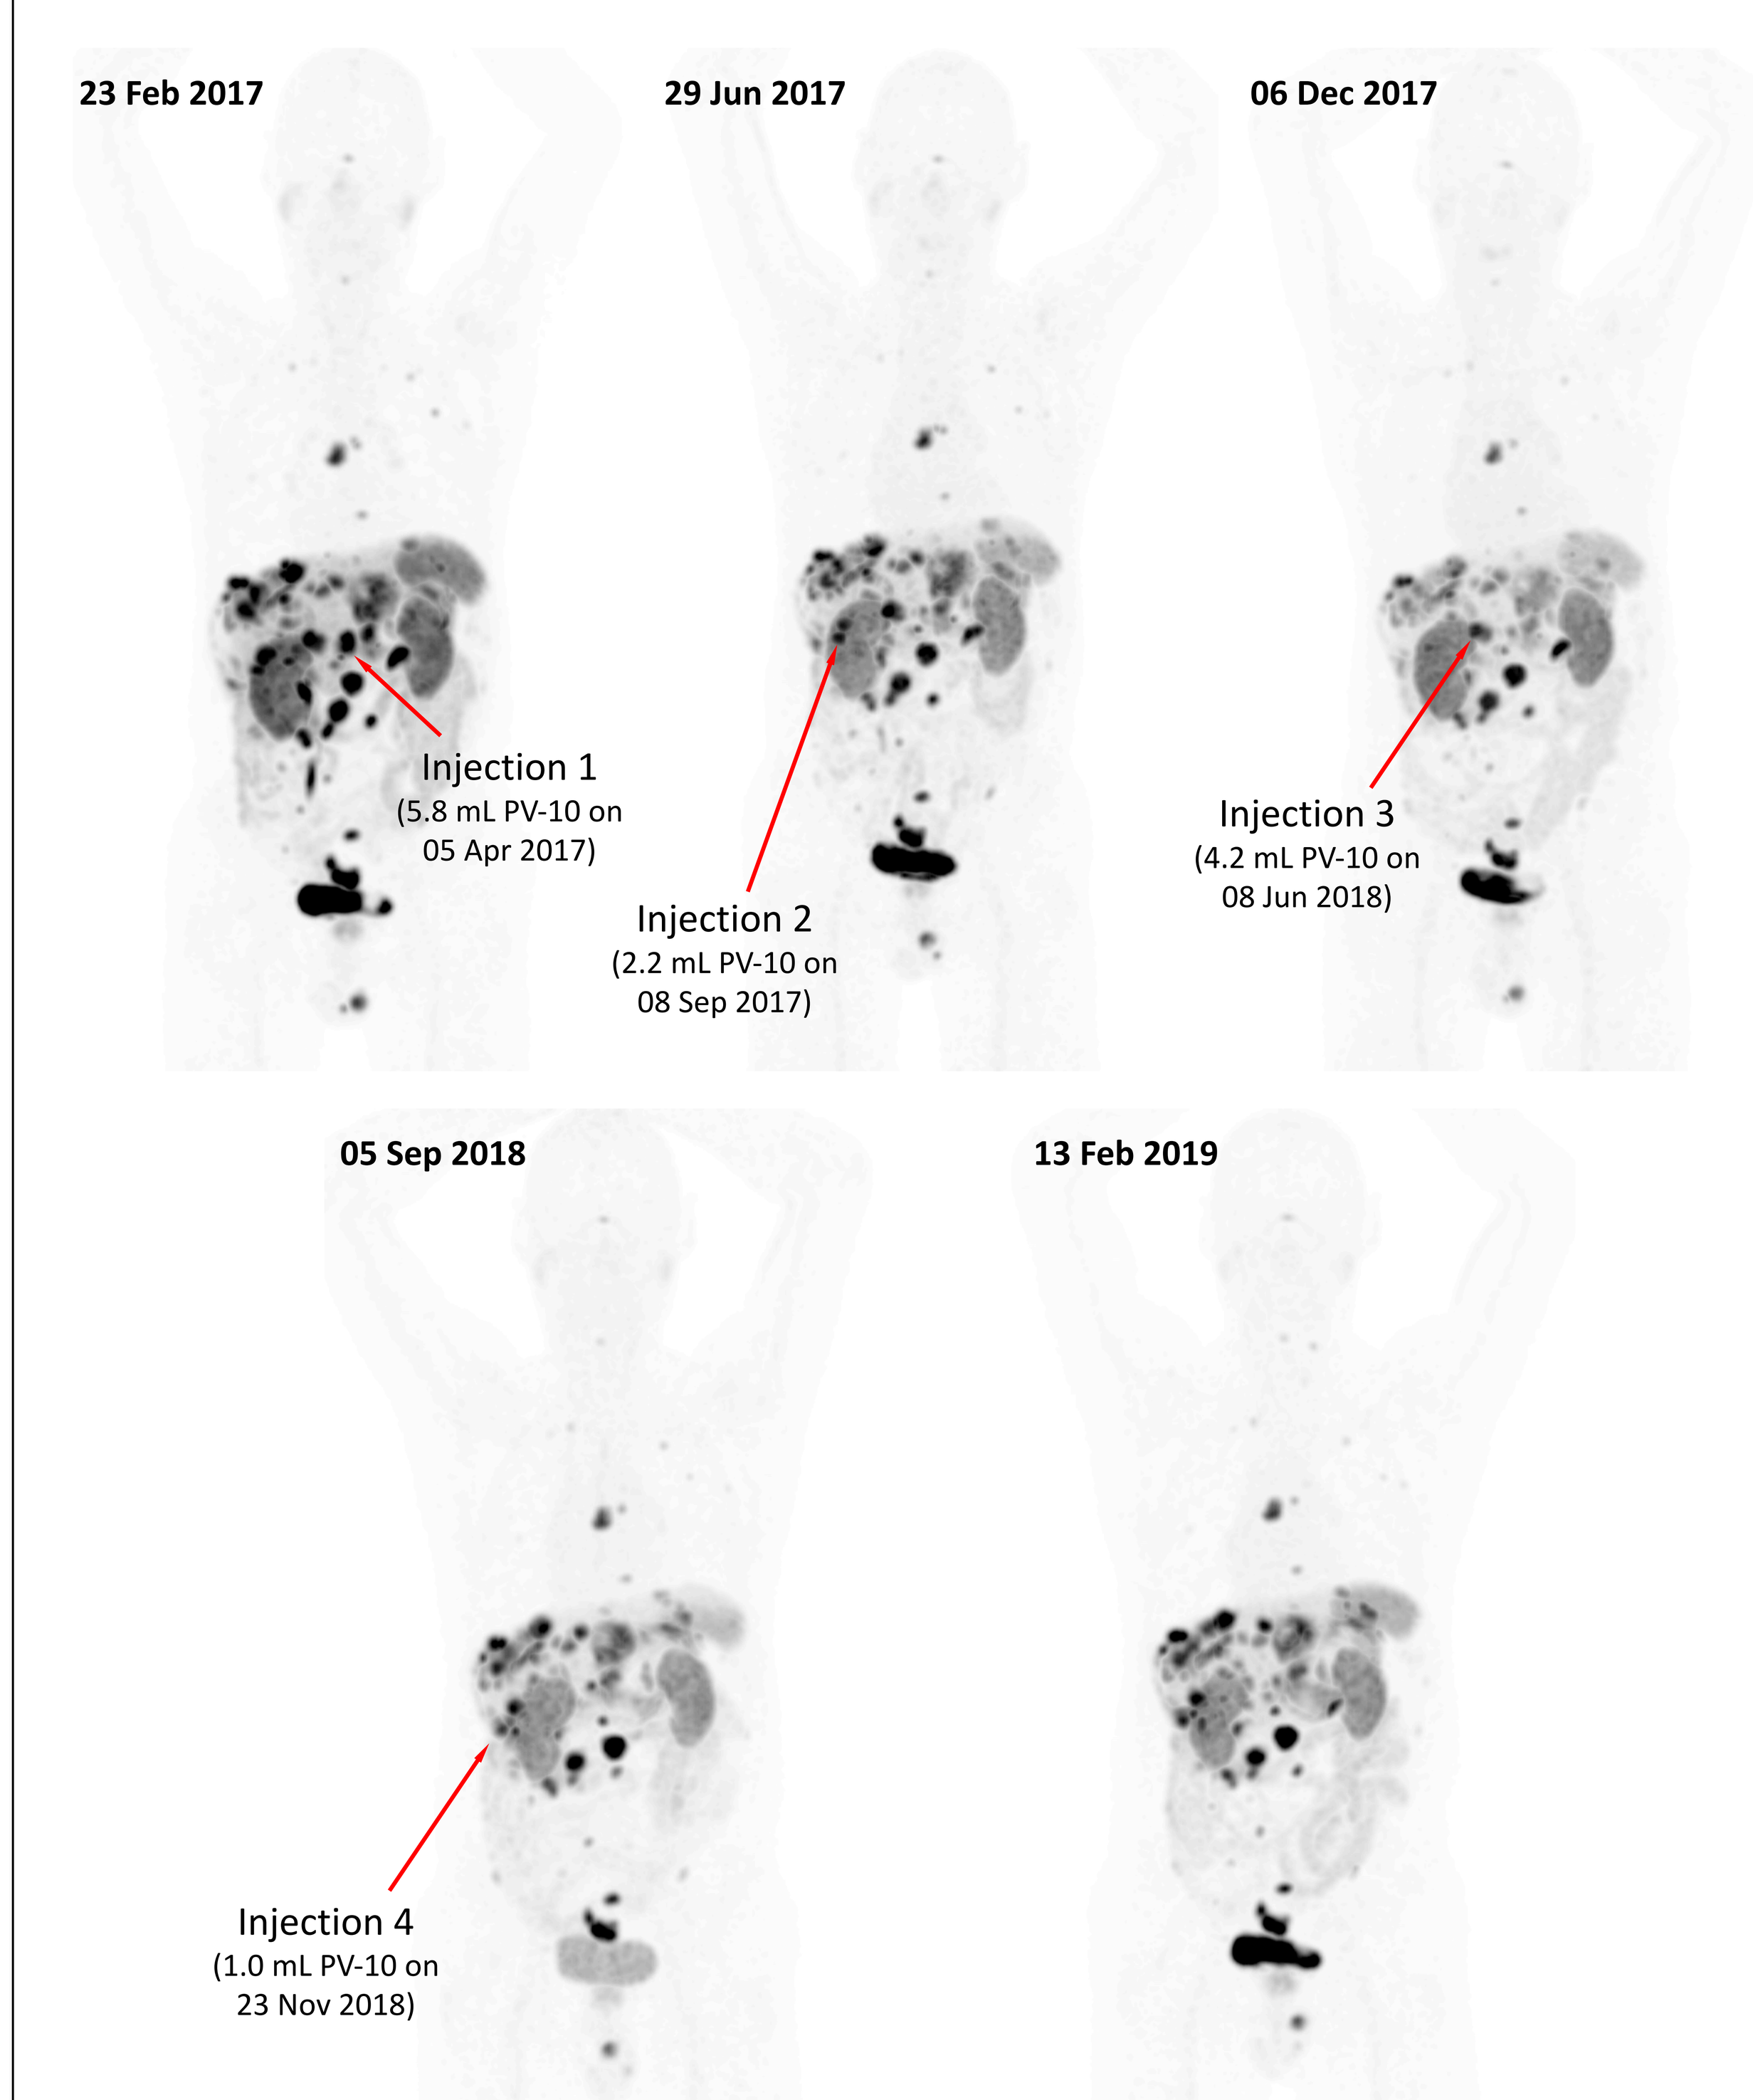
23 Feb 2017
29 Jun 2017
06 Dec 2017
Injection 1
(5.8 mL PV-10 on 05 Apr 2017)
Injection 2
(2.2 mL PV-10 on 08 Sep 2017)
Injection 3
(4.2 mL PV-10 on 08 Jun 2018)
05 Sep 2018
13 Feb 2019
Injection 4
(1.0 mL PV-10 on 23 Nov 2018)

Subject 0101: Anterior MIP – Ga-DOTATATE PET

Results: Objective Response

Objective Response (Cohort 1):

- ORR in injected lesions is 50% (progression in 1 subject), with overall disease control of 83%
- Response follow-up is ongoing for 3 of 6 subjects in Cohort 1

Conclusions

- PV-10 elicited no safety concerns in hepatic mNET, with a safety profile consistent with percutaneous intralesional delivery to other hepatic malignancies [13,14]
- PV-10 is readily imaged during administration and follow-up
- Encouraging single-agent activity is evident in injected lesions
- PV-10 may yield systemic disease control through T cell activation
- Combination of PV-10 with checkpoint inhibition (CI) may overcome lack of CI activity in NET [1] (an immunologically "cold" class [2])
- Enrolment to Cohort 2 of the single-agent study is underway (2 of 6 subjects have received at least one cycle of PV-10 in Cohort 2)
- Future development options include:
 - Expansion of current single-agent study to open a phase 1 expansion cohort at multiple centres in AUS and USA
 - Initiation of a phase 2 combination study with anti-PD-1 or anti-PD-L1 antibodies (pharma partner needed for CI supply and co-sponsorship)
 - Academic co-sponsorship with regional or global consortium

For additional information: Timothy.Price@sa.gov.au
wachter@pvct.com