Chemoablation of Melanoma with Intralesional Rose Bengal (PV-10)

S. S. Agarwala [1], J. Thompson [2], M. Smithers [3], M. Ross [4], B. Coventry [5], D. Minor [6], C. Scoggins [7], P. Hersey [8] and E. Wachter [9]

[1] St Luke's Hospital & Health Network, Bethlehem, PA USA; [2] Sydney Melanoma Unit, Sydney, NSW AUS; [3] Princess Alexandra Hospital, Woolloongabba, QLD AUS; [4] M. D. Anderson Cancer Center, Houston, TX USA; [5] Royal Adelaide Hospital, Adelaide, SA AUS; [6] California Pacific Medical Center, San Francisco, CA USA; [7] University of Louisville, Louisville, KY USA; [8] Newcastle Melanoma Unit, Waratah, NSW AUS; [9] Provectus Pharmaceuticals, Inc., Knoxville, TN USA

Abstract No. 9060
ASCO Annual '09 Meeting
29 May - 2 June 2009

For additional information:
Eric Wachter
wachter@pvct.com

ABSTRACT

Background: Intralesional rose bengal (PV-10, a sterile 10% solution in saline) can elicit selective ablation of solid tumors and an apparent bystander response in untreated lesions. In phase 1 testing (study PV-10-MM-01) in 20 subjects with AJCC Stage III (19 subjects) or IV (1 subject) melanoma, a single injection of PV-10 into 1-20 lesions led to durable objective response (OR) at 12-24 weeks in 40% of subjects (20% CR + 20% PR by modified RECIST) and locoregional disease control (CR + PR + SD) in 75% of subjects. Untreated bystander lesions achieved an OR in 15% of subjects, and all subjects with an OR of their injected lesions achieved disease control of their bystander lesions. PV-10 was well tolerated, with only one Grade 3 adverse event (photosensitivity reaction). The most common AEs were transient pain at the treatment site, followed by local inflammation or infection.

Methods: Expanded phase 2 testing commenced in late 2007 in up to 80 subjects with measurable Stage III or IV melanoma (study PV-10-MM-02). After an initial treatment of 1-20 cutaneous, subcutaneous or nodal lesions, new or incompletely responsive lesions could be retreated at weeks 8, 12 or 16, with follow-up to 52 weeks. An additional up to 2 lesions, including visceral lesions, remained untreated for assessment of bystander response. Seven centers in Australia and the USA enrolled subjects, with enrollment completed May 2009. A modified Fleming design allowed interim assessment of safety and OR at weeks 4 and 24, respectively, after treatment of the 20th and 40th subjects. The primary end-point is OR of injected lesions in the intent-to-treat population; secondary endpoints include OR of bystander lesions and PFS.

Results: Interim safety data for the first 40 subjects treated is comparable to phase 1, with transient mild to moderate locoregional pain, vesicles or edema most common; Grade 3 AEs have been rare, with no Grade 4 or 5 AEs attributed to PV-10. Interim efficacy for the first 40 subjects is also comparable to that of phase 1.

Conclusions: The safety and efficacy profile of intralesional therapy with PV-10 compares favorably with available therapeutic options for this patient population.

BACKGROUND

PV-10 is a sterile, non-pyrogenic solution of Rose Bengal disodium (10% RB)

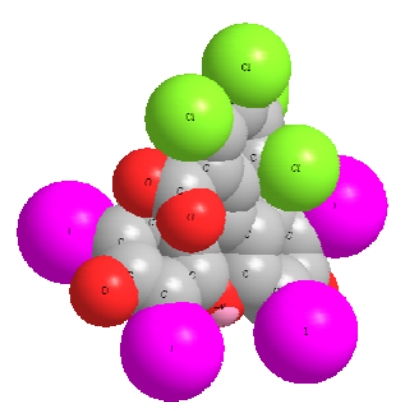

- ***RB is a Fluorescein derivative attributed to Gnehm in 1882***
- ***Prior Human Use of RB***
 - IV hepatic diagnostic, ^{131}I radiolabeled RB: Robengatope®
 - Topical ophthalmic diagnostic: Rosettes® and Minims®
- ***Established Safety History***
 - Not metabolized, short half-life (ca 30 min), excretion via bile
- ***In Non-Clinical Testing PV-10 Targeted Neoplastic Tissue***
 - Murine xenograft/ homograft and spontaneous tumors in companion animals
 - Prolonged retention in tumors
 - Lysosomal accumulation within cancer cells leading to acute necrosis
 - Minimal toxicity in normal tissue
 - Selective chemoablation of injected tumors
- ***PV-10 May Elicit Bystander Effect in Non-Injected Tumors***
 - Ablation reduces tumor burden and exposes tumor antigens to host
 - Apparent immune-mediated response
 - Possible systemic benefit
- ***Phase 1 Clinical Testing***
 - Single injection into 1–20 lesions in 20 subjects with AJCC Stage III/IV melanoma
 - Intralesional dosing at 50% of calculated lesion volume
 - 1–3 additional lesions untreated to assess bystander response
 - 12–24 weeks observation
 - ORR assessed by modified RECIST
 - AEs generally mild to moderate grade – predominantly locoregional
 - Preliminary efficacy
 - Injected lesions: ORR = 40% (locoregional disease control in 75% of subjects)
 - Bystander lesions: ORR = 15% (locoregional disease control in 55% of subjects)

PHASE 2 OVERVIEW

Protocol PV-10-MM-02

- ***Study Design***
 - Open label, single-arm trial
 - 80 subjects with AJCC Stage III/IV melanoma
 - **Treatment of 1–10 Target Lesions and up to 10 Non-Target Lesions**
 - Target Lesions must be ≥ 0.2 cm diameter
 - Biopsy confirmation of at least one Target Lesion
 - Single intralesional dosing at 50% of calculated lesion volume
 - **Observe up to 1–2 untreated Bystander Lesions**
 - Typically small or difficult to access
 - Biopsy confirmation of each Bystander Lesion
 - **Retreatment (new or partially-responsive lesions) allowed at weeks 8, 12 or 16**
 - **Observe for 52 weeks**
- ***Outcome Assessment***
 - Modified RECIST assessed on Target and Bystander Lesions
 - Progression Free Survival
 - Modified Fleming design
 - Interim Safety Assessment 28 days after 20th and 40th subject treated
 - Interim Efficacy Assessment 24 weeks after 20th and 40th subject treated
- ***Study Status: Enrollment commenced Aug 2007, completed May 2009***
- ***N = 40 Interim Safety and Efficacy Assessments complete***
 - AJCC Stage III (35 subjects) and Stage IV (5 Subjects) metastatic melanoma
 - 26 males / 14 females (all subjects white) , median age 74.5 yrs (range 37–92 yrs)
 - Disease history: median 33.6 months from 1^0 diagnosis to study enrollment
 - Treatment history: Sx (40 subjects), nodal biopsy (24), ILI/ILP (7), XRT (7), amputation (3), IFN (2), vaccine (2), systemic chemo (1), investigational agents (1)
 - Study lesions: extremity (27 subjects lower and 4 upper extremity), torso (6), H&N (5)
 - Lesions treated per subject: median 8 (range 1–20)
 - Number of treatment cycles per subject: median 2 (range 1–3)
 - PV-10 dose per treatment: mean 2.5 mL, median 1.4 mL (range 0.2–15.0 mL)
 - Cumulative PV-10 dose per subject: mean 4.6 mL, median 2.9 mL (range 0.4–25.6 mL)

ADVERSE EVENTS

Adverse Events At Least Possibly Related to Treatment
Protocol PV10-MM-02 – Initial 28 Days Post First Treatment, All Subjects (N = 40)

System Organ Class[1] / Preferred Term[1]	Adverse Events (by CTCAE Grade) 1	2	3	4	Total	%
Nervous system disorders						
Headache	3	1	0	0	4	10%
Gastrointestinal disorders						
Diarrhoea	3	0	0	0	3	8%
Abdominal pain	1	0	0	0	1	2%
Skin and subcutaneous tissue disorders						
Rash generalized	0	1	0	0	1	2%
Skin discolouration	0	1	0	0	1	2%
Renal and urinary disorders						
Polyuria	2	0	0	0	0	5%
General disorders and administration site conditions						
Injection site pain	9	13	2	0	24	60%
Injection site vesicles	6	5	1	0	12	30%
Injection site oedema	6	5	0	0	11	28%
Injection site swelling	4	3	0	0	7	18%
Injection site erythema	2	2	0	0	4	10%
Injection site warmth	1	1	0	0	2	5%
Injection site inflammation	0	1	0	0	1	2%
Injection site cellulitis	0	0	1	0	1	2%
Injection site infection	0	1	0	0	1	2%
Injection site skin (flap) necrosis	0	0	1	0	1	2%
Injection site irritation	1	0	0	0	1	2%
Injection site pruritus	1	0	0	0	1	2%
Injection site urticaria	1	0	0	0	1	2%
Injection site reaction	1	0	0	0	1	2%
Injection site photosensitivity reaction	1	0	0	0	1	2%
Lethargy	2	0	0	0	2	5%

[1] System Organ Class and Preferred Term are based on the MedDRA version 11.1 dictionary

EXAMPLE CLINICAL RESPONSE

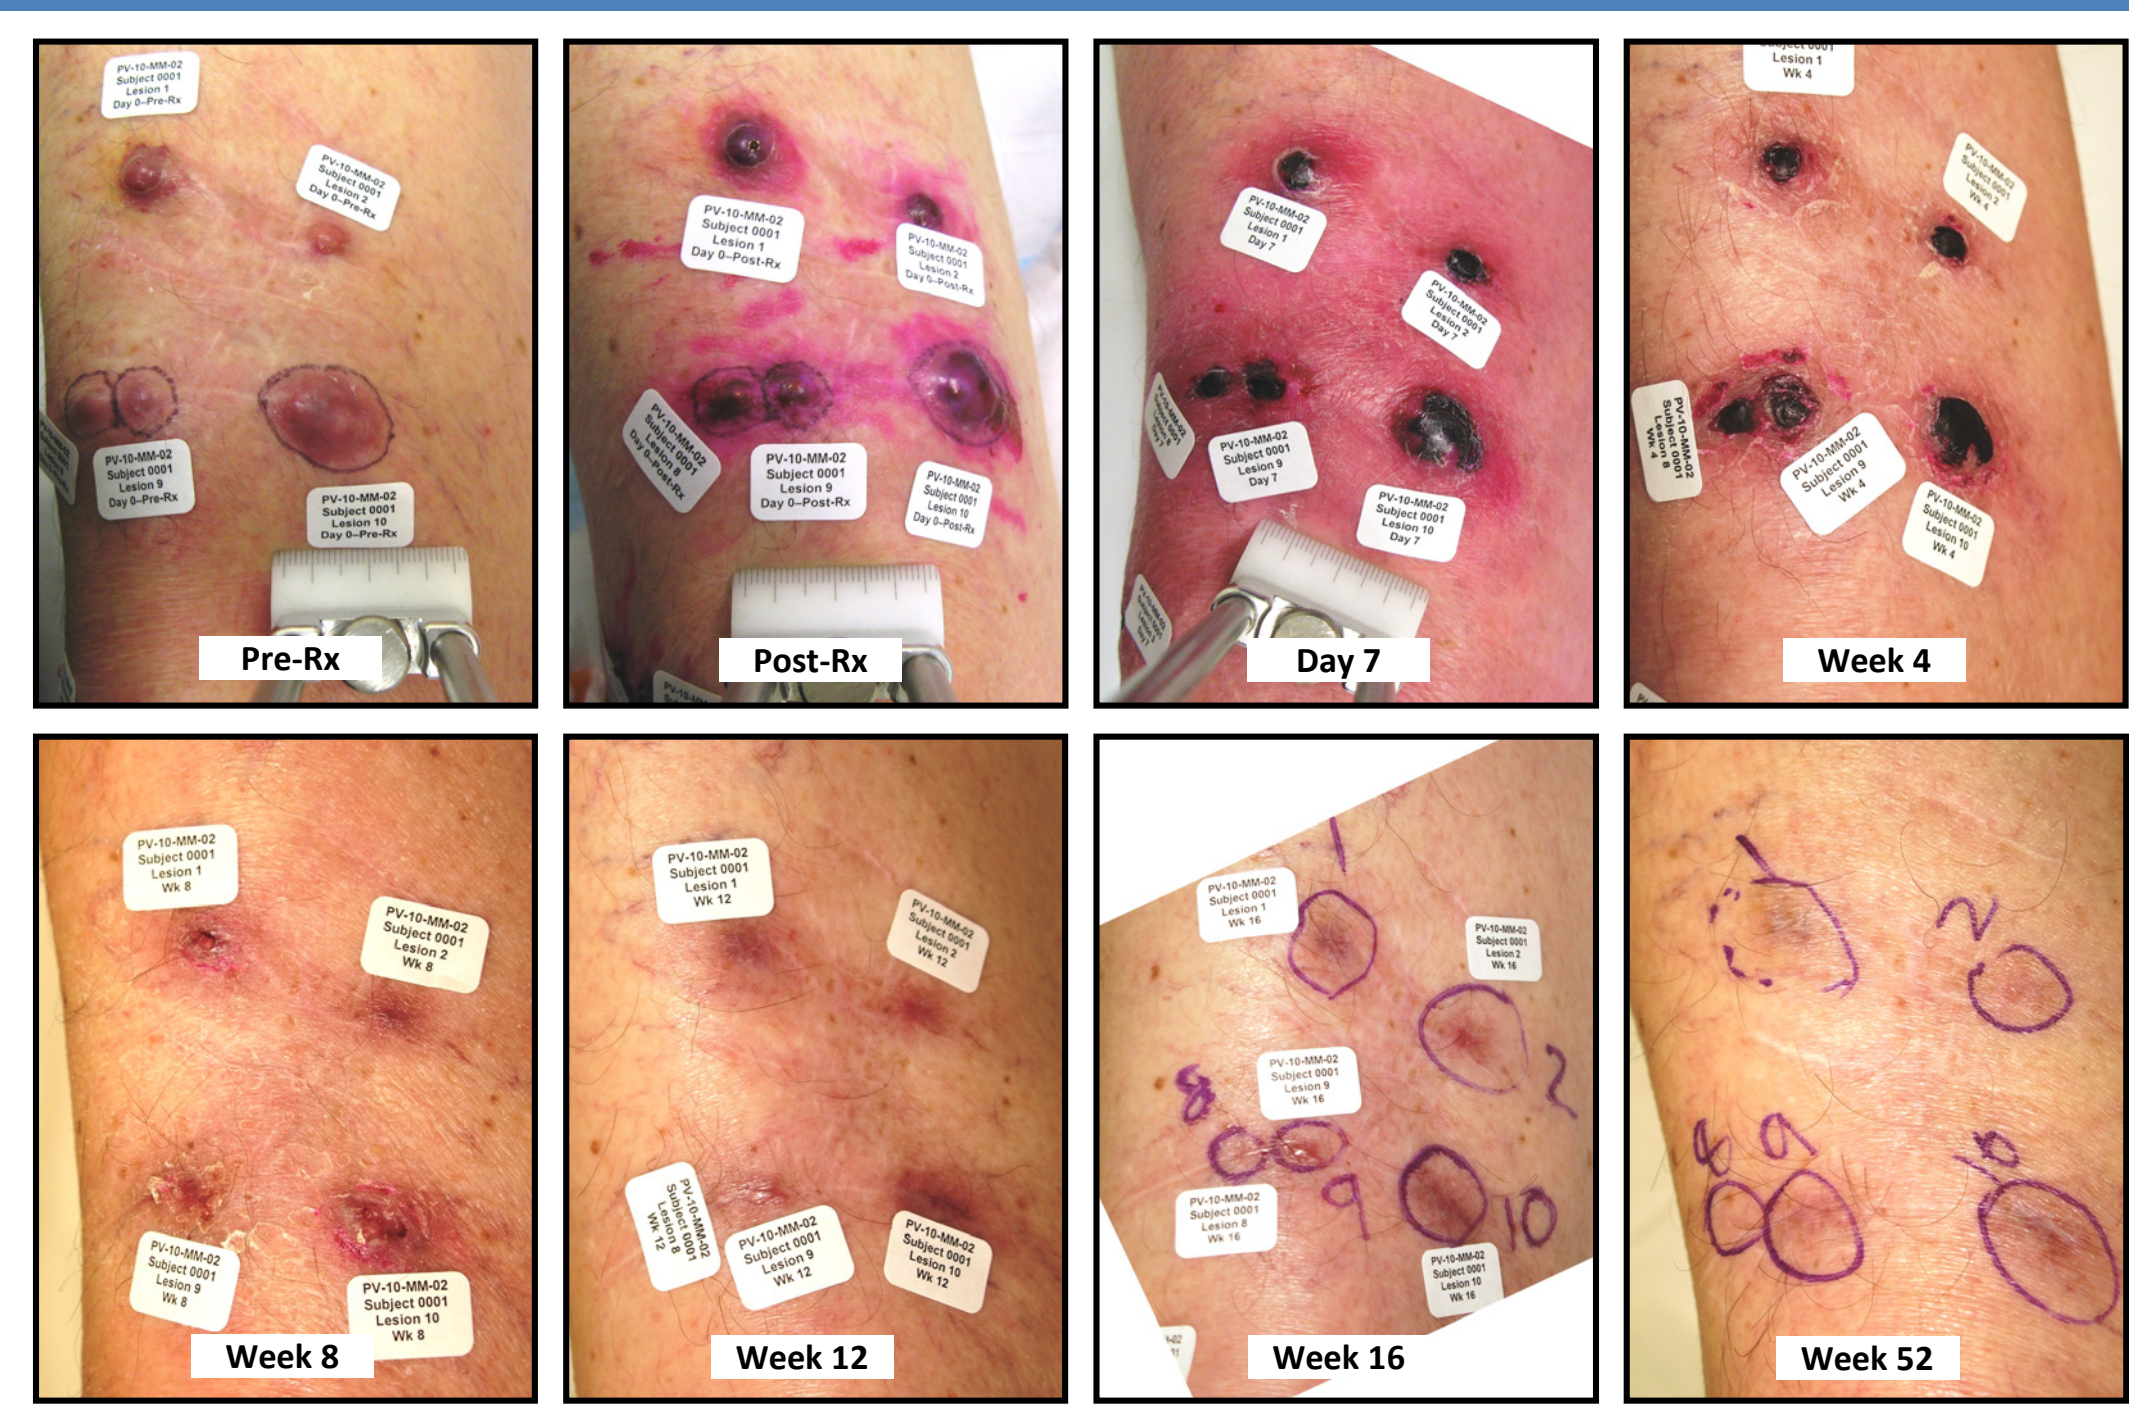
Pre-Rx
Post-Rx
Day 7
Week 4
Week 8
Week 12
Week 16
Week 52

Male, age 78, Stage III (M1a in transit), onset 1992, multiple Sx of mets since 2005.
Single treatment with 2.0 mL PV-10 to 10 lesions; 1 untreated bystander lesion.

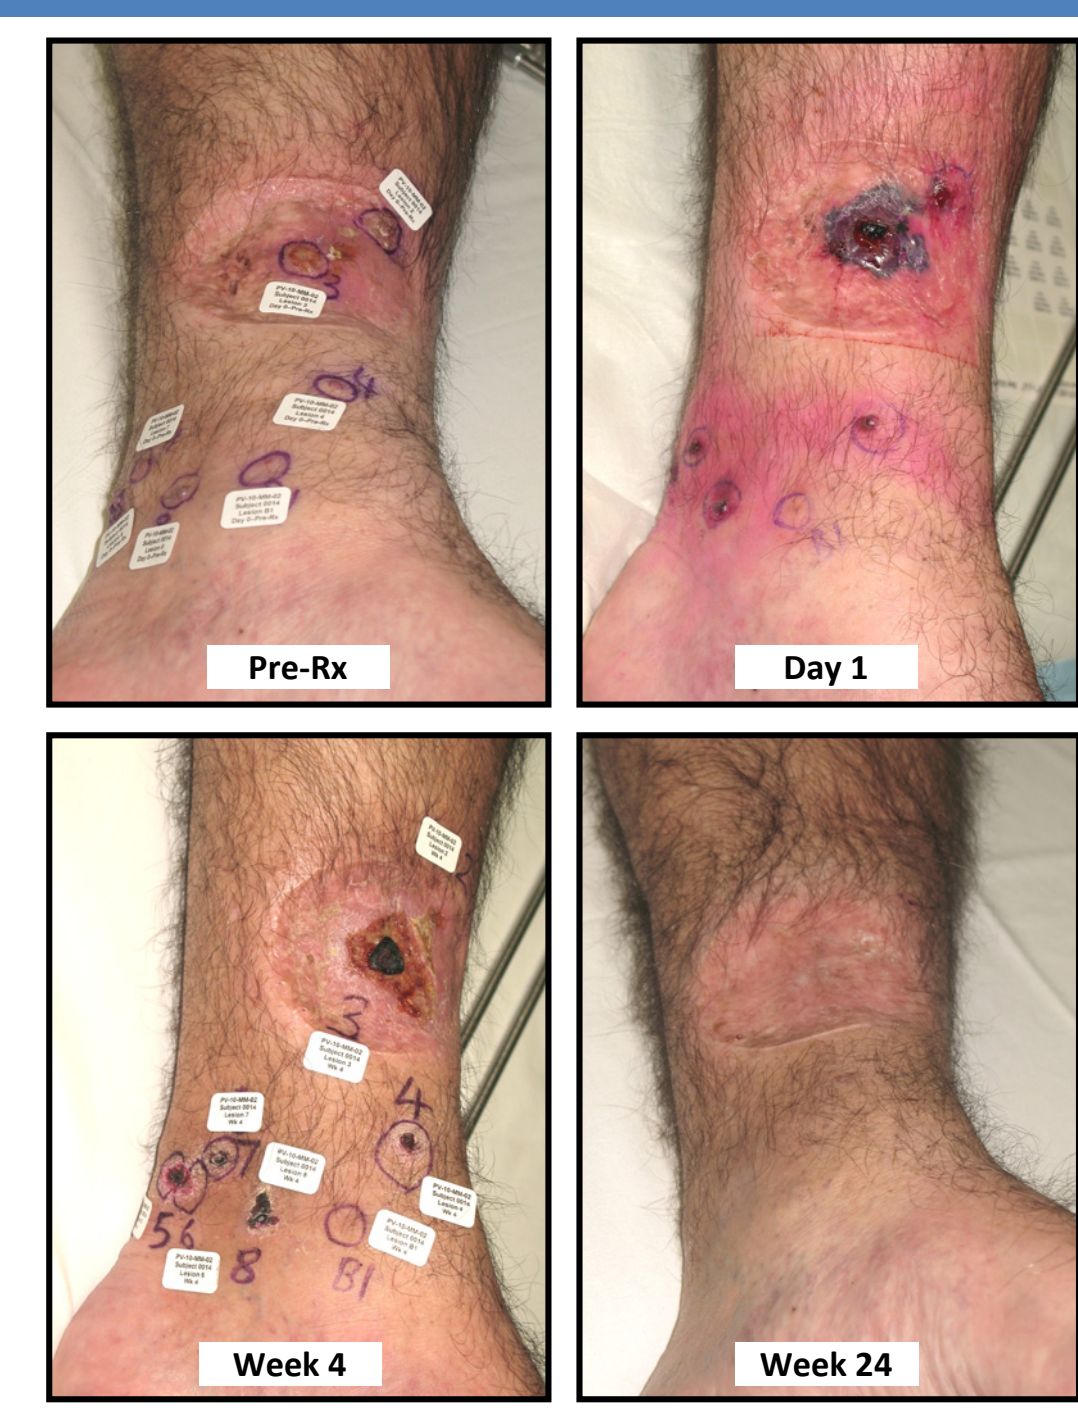
Pre-Rx
Day 1
Week 4
Week 24

Male, 48, Stage III (M0 in transit) since 2008, Sx of 1^O and mets.
Single treatment with 1.3 mL PV-10 to 10 lesions.

PHASE 2 INTERIM EFFICACY

Objective Response of Study Lesions
All Subjects (N=40)

Best Response (RECIST, N = 40 Subjects through Week 24)	Target Lesions N		Bystander Lesions N	
N (Subjects)	40		40	
CR	12	30%	5	24%
PR	12	30%	5	24%
SD	6	15%	3	14%
PD	10	25%	8	38%
ND	--	--	19	(48%)
CR + PR	24	60%	10	48%
CR + PR + SD (*Locoregional Disease Control*)	30	75%	13	62%

Objective Response of Bystander Lesions
Grouped According to Objective Response of Target Lesions
All Subjects (N=40)

Bystander Lesion Response	Subjects with POSITIVE Objective Response of Target Lesions		Subjects with NEGATIVE Objective Response of Target Lesions		χ^2	Fisher Exact
N (Subjects)	24		16			
CR	5	38%	0	0%		
PR	4	31%	1	12%		
SD	0	0%	3	38%		
PD	4	31%	4	50%		
ND	11	(46%)	8	(50%)		
CR + PR	9	69%	1	12%	χ^2 = 4.318	P = 0.024
SD + PD	4	31%	7	88%	1 df	
					P = 0.038	

Response of each subject's bystander lesions (overall subject response) as a function of the subject's objective response of target lesions (POSITIVE Objective Response = CR + PR subjects; NEGATIVE Objective Response = SD + PD subjects). Statistical significance of response rates tested using the Chi-Square and Fisher Exact tests. Nineteen subjects had no designated bystander lesion (or no assessable lesion) to assess (ND) and were censored.

Objective response may be a good predictor of long-term outcome

- ***Phase 1 subjects achieving CR or PR of their injected lesions exhibited longer overall and disease specific survival than those achieving SD or PD***
 - **Overall Survival (OS)**
 CR + PR Cohort: Median = 42.1 months Mean = 35.2 months
 SD + PD Cohort: Median = 12.3 months Mean = 19.8 months
 - **Disease Specific Survival (DSS)**
 CR + PR Cohort: Median = 44.1 months Mean = 38.0 months
 SD + PD Cohort: Median = 14.6 months Mean = 20.5 months
 - **Survival data current as of 6 May 2009**
- ***Survival data (PFS and OS) currently being collected for Phase 2 subjects***

CONCLUSIONS

PV-10 is well tolerated, eliciting a robust response in a majority of patients

- The safety and efficacy profile compare favorably with existing and emerging therapies
- Suitable for repeat treatment of patients with partially-responsive lesions or new lesions to maximize OR and long-term outcome
- Potential for locoregional control of metastatic disease

Locoregional treatment may yield systemic benefit via the bystander effect

Phase 2 interim results (N=40) are consistent with phase 1 results (N=20)