Phase 1b Study of PV-10 and anti-PD-1 in Advanced Cutaneous Melanoma

Abstract No. 9559
2019 ASCO Annual Meeting
Chicago, IL 31 May – 4 June 2019

SS Agarwala [1], MI Ross [2], JS Zager [3], K Shirai [4], R Essner [5], BM Smithers [6], V Atkinson [6] and EA Wachter [7]

[1] St Luke's University Hospital and Health Network, Easton, PA; [2] MD Anderson Cancer Center, Houston, TX; [3] Moffitt Cancer Center, Tampa, FL; [4] Dartmouth-Hitchcock Medical Center, Lebanon, NH; [5] John Wayne Cancer Center, Santa Monica, CA; [6] Princess Alexandra Hospital, Brisbane, QLD AUS; [7] Provectus Biopharmaceuticals, Inc., Knoxville, TN.

Copies of this poster obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without written permission of the authors.

Background

PV-10 (rose bengal disodium) is a small molecule oncolytic immunotherapy in development for solid tumors, where intralesional injection can yield immunogenic cell death and tumor-specific reactivity in circulating T cells [1-4]. It has been administered as a single agent to over 300 cutaneous melanoma patients (pts) in Phase 1-3 testing and under expanded access [5-9]. PV-10 is also under investigation for percutaneous administration to hepatic tumors (e.g., hepatocellular carcinoma, metastatic uveal melanoma, and metastatic neuroendocrine tumors) [10-12].

Primary Oncolysis

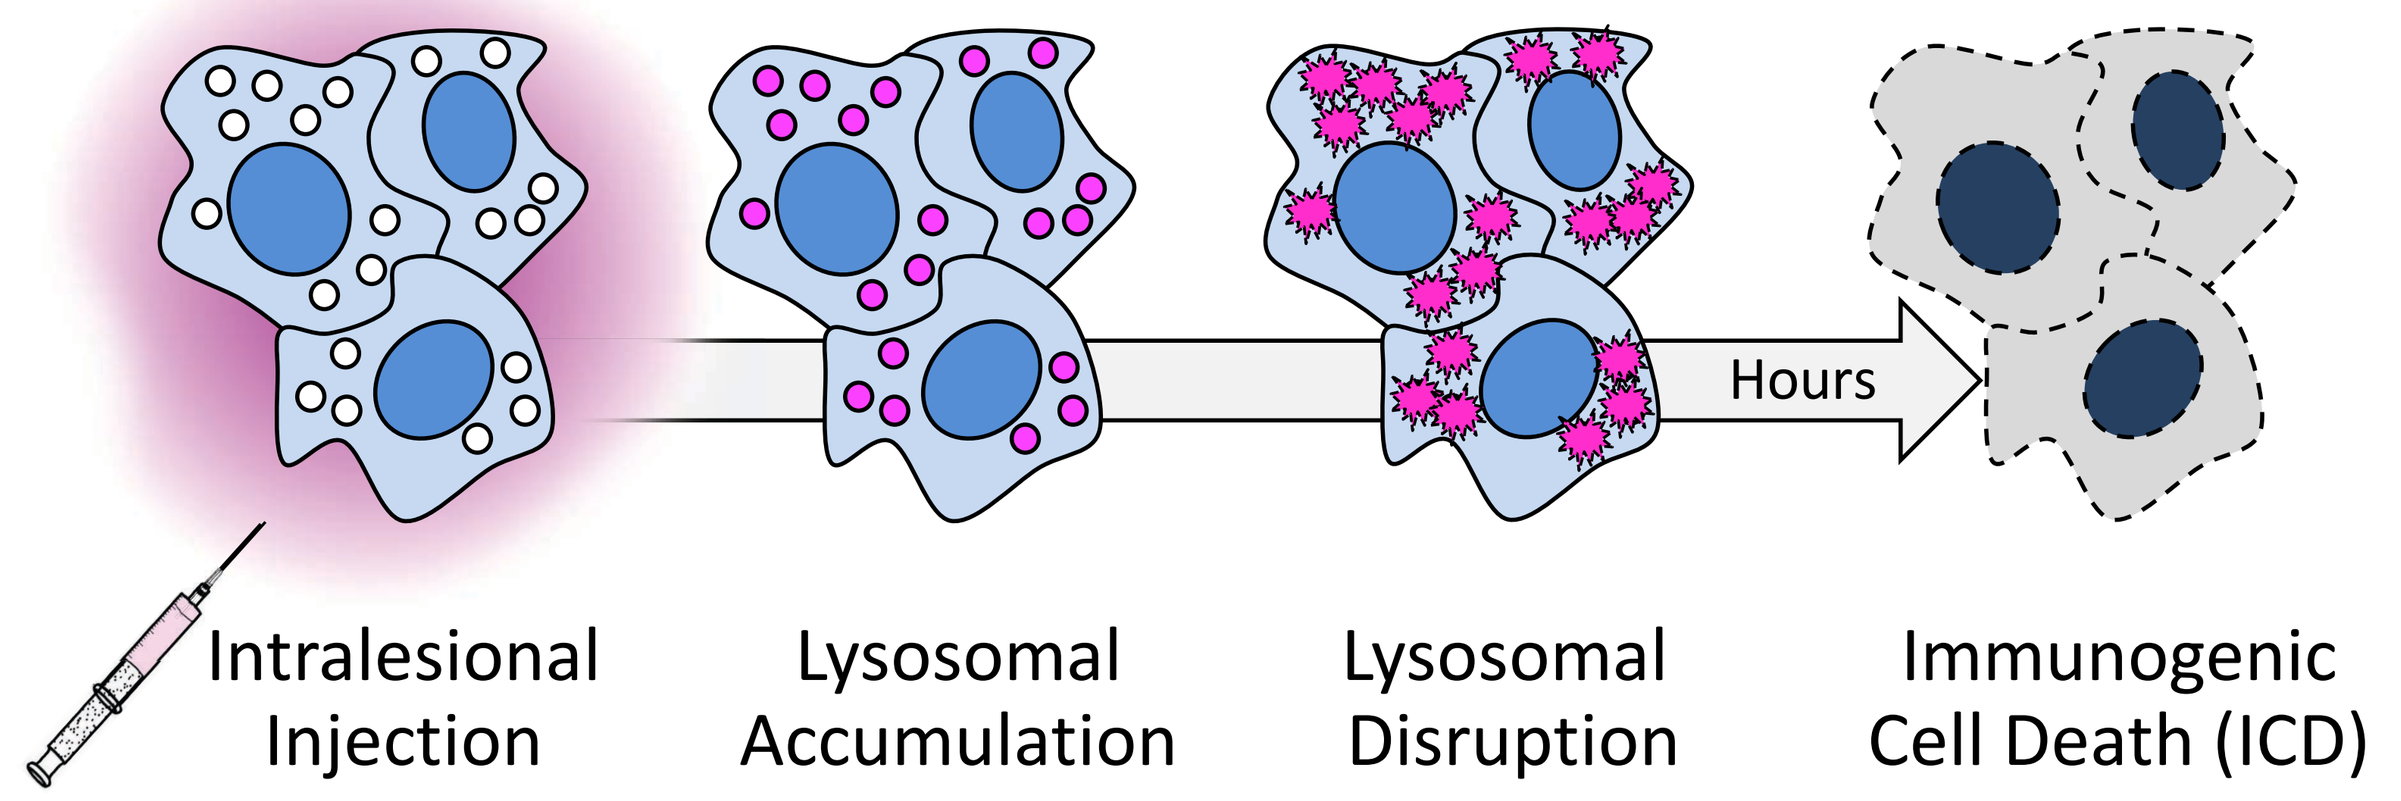
Hours
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Immunogenic Cell Death (ICD)

Secondary Adaptive Immunity

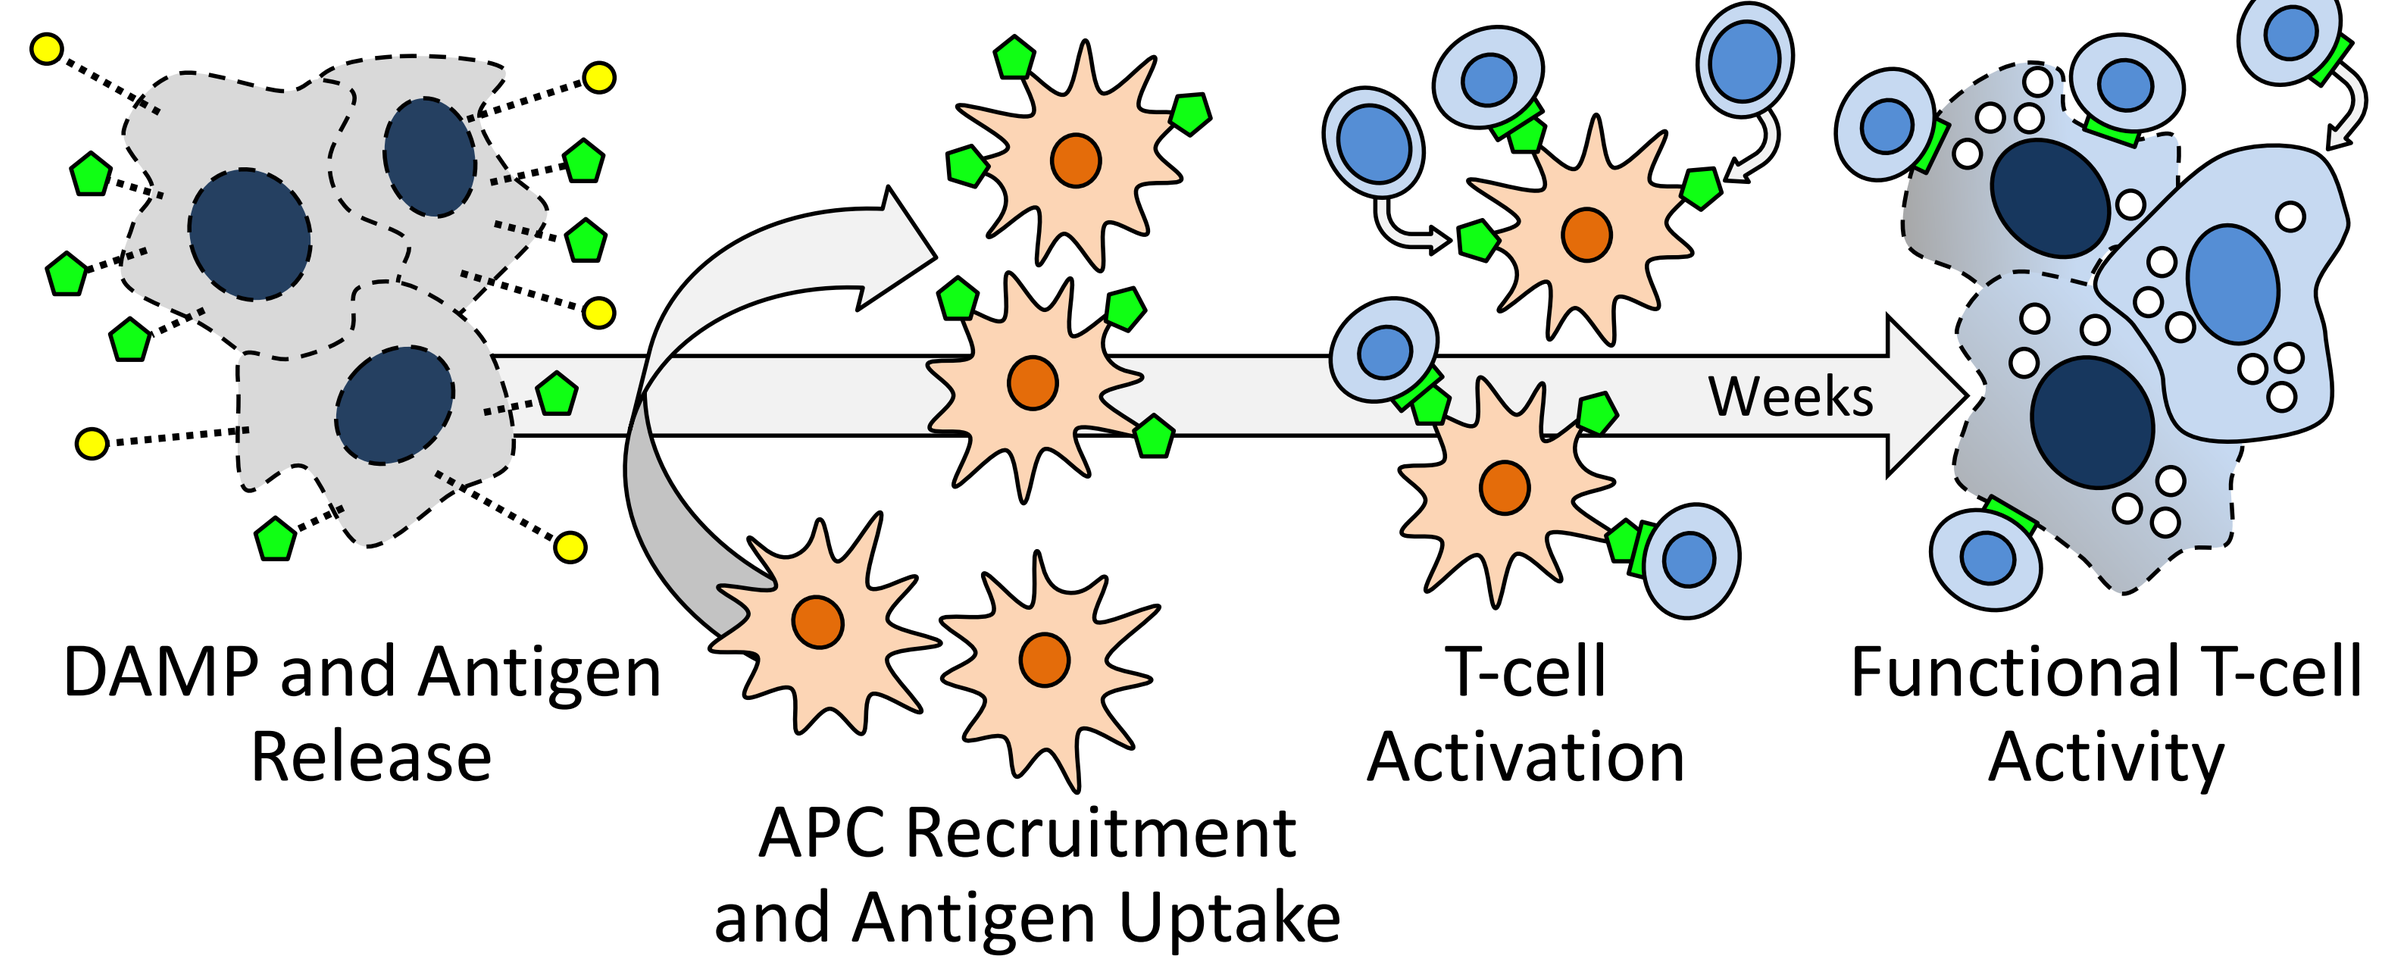
Weeks
DAMP and Antigen Release
APC Recruitment and Antigen Uptake
T-cell Activation
Functional T-cell Activity

1. Wachter et al., Proceedings of SPIE 2002; 4620: 143. **2.** Liu et al., Oncotarget 2016; 7: 37893. **3.** Qin et al., Cell Death and Disease 2017; 8: e2584. **4.** Liu et al., PLoS ONE 2018; 13: e0196033. **5.** Thompson et al., Melanoma Res 2008; 18: 405. **6.** Thompson et al., Annals Surg Oncol 2015; 22: 2135. **7.** Lippey et al., J Surg Oncol 2016; 114: 380. **8.** Foote et al., J Surg Oncol 2017; 115: 891. **9.** Read et al., J Surg Oncol 2018; 117: 579. **10.** Goldfarb et al., CIO 2017. **11.** Patel et al., ISOO Biennial Conference 2019. **12.** Price et al., ASCO 2019 (abstract 4102).

Methods

Study **PV-10-MM-1201** (**NCT02557321**) is a Phase 1b/2 study of PV-10 in combination with anti-PD-1 (pembrolizumab) for patients with advanced cutaneous melanoma (Stage IIIB-IV M1c). Patients must have at least 1 injectable lesion and be candidates for pembrolizumab. In Phase 1b pts receive combination treatment q3w for 5 cycles then pembrolizumab alone for up to 24 months; the primary endpoint is safety and tolerability with objective response rate (ORR) and progression free survival (PFS) key secondary endpoints (by RECIST 1.1 after 5 cycles then q12w).

Phase 1b includes 3 cohorts:

- Main Cohort of up to 24 checkpoint-naïve patients
- Expansion Cohort 1 (EC1) of up to 24 checkpoint-refractory patients
- Expansion Cohort 2 (EC2) of up to 24 Stage III in-transit or satellite patients

Full accrual of the Main Cohort of Phase 1b was reached in April 2018, with EC1 and EC2 currently open to accrual.

Study Participants

Subject Characteristics
(Phase 1b Main Cohort ITT Population, N = 23)

ID / Age / Gender	Stage	CUT/SQ Lesions [a]	Location(s) of Target Lesions	Site(s) of Non-Target Lesions
0111 / 68 / F	IIIC	8	Scalp (x2)	Scalp
0501 / 82 / M	IIIC	15	LUE (x2)	LNs
0110 / 90 / F [b]	IIID	5	Scalp (x2)	Scalp
0106 / 78 / M	M1a	1	RLE (x2)	LLE
0108 / 82 / F	M1a	2	LUE (x2)	LUE
0109 / 60 / M	M1a	1	Chest (x2), Axillary LN	Axillary LN and Skin
0202 / 52 /M	M1a	1	Chest Wall	Chest Wall and Axillary LN
0204 / 28 / M	M1a	TNC	Jaw, LUE	Scalp, Face, Neck, Torso, LUE, Axillary LN
0205 / 50 / F [c]	M1a	3	Chest Wall	Back
0207 / 44 / M	M1a	1	Infraclavicular (SQ)	Subcutaneous
0209 / 66 / M	M1a	2	Back, LUE	Spinal Muscle and LN, Back
0102 / 47 / M [d]	M1b	4	Scalp	Lung
0104 / 79 / M [d]	M1b	3	LLE	Bilateral Lung
0107 / 68 / M	M1b	1	Flank (SQ)	Lung
0203 / 76 / M	M1b	2	RLE (x2)	Lower Extremity and Lung
0206 / 73 / M	M1b	1	RUE	Lung
0401 / 70 / M [e]	M1b	1	Shoulder, Lung, Liver	Liver and Lung
0502 / 47 / M	M1b	2	Head, Lung	Lung and Hilar LN
0101 / 81 / M	M1c	3	SQ (x2), Axillary LN	Liver and Lung
0105 / 69 / M	M1c	4	RUE, R Axilla, Liver (x2)	Bone, Liver and Lung
0112 / 80 / M	M1c	1	Scalp, Liver (x2)	Liver
0402 / 79 / M [f]	M1c	2	Axilla, Flank, Lung, Liver, Peritoneum	Peritoneum, Retroaortic LNs and Lung
0403 / 78 / M	M1c	TNC	Shoulder (x2), Clavicle, Bladder (x2)	Bladder and Bone

Abbreviations: CI, checkpoint inhibition; CUT, cutaneous; LLE, left lower extremity; LN, lymph node; LUE, left upper extremity; ITT, intent-to-treat; N, number; RLU, right lower extremity; RUE, right upper extremity; SQ, subcutaneous; TNC, too numerous to count.

[a] Subjects had a median of 2.0 injectable lesions (range 1 – 15); Subjects 0204 and 0403 excluded from calculation (TNC).
[b] Subject 0110 previously received nivolumab (12 months ending 2 months prior to enrollment – enrolled under waiver for prior CI).
[c] Subject 0205 previously treated with dabrafenib and trametinib.
[d] Subjects 0102 and 0104 previously treated with pegylated interferon alfa-2b.
[e] Subject 0401 previous treated with doxorubicin and olaratumab.
[f] Subject 0402 previously treated with adjuvant vaccine; and with ipilimumab (ending 3 years prior to enrollment – enrolled under waiver for prior CI).

Safety

Treatment-Emergent Adverse Events (TEAEs) were consistent with established patterns for each drug, with 6 Grade 1-2 TEAEs attributed to the combination in 6 subjects.

Treatment-Emergent Adverse Events (TEAEs)
Occurring in >1 Subject, or Any Grade 3 or Higher
(Phase 1b Main Cohort ITT Population, N = 23)

	TEAEs Related to PV-10		TEAEs Related to Pembrolizumab		TEAEs Related to Combination	
	All	≥ G3	All	≥ G3	All	≥ G3
GENERAL DISORDERS AND ADMINISTRATION SITE CONDITIONS						
Injection site pain	18	*1*	0	0	0	0
Injection site discharge	7	0	0	0	0	0
Injection site oedema	6	0	0	0	0	0
Injection site photosensitivity reaction	5	0	0	0	0	0
Injection site discolouration	4	0	0	0	0	0
Injection site ulcer	3	0	0	0	0	0
Injection site haemorrhage	2	0	0	0	0	0
Injection site pruritus	2	0	0	0	2	0
Injection site erythema	2	0	0	0	1	0
Fatigue	0	0	10	0	1	0
Influenza like illness	0	0	1	0	1	0
Pyrexia	0	0	1	0	1	0
INJURY, POISONING AND PROCEDURAL COMPLICATIONS						
Eschar	2	0	0	0	0	0
SKIN AND SUBCUTANEOUS TISSUE DISORDERS						
Pruritus	0	0	4	*1*	0	0
Rash	0	0	4	0	0	0
Rash maculo-papular	0	0	3	0	0	0
METABOLISM AND NUTRITION DISORDERS						
Hypothyroidism	0	0	7	0	0	0
Hyperthyroidism	0	0	4	0	0	0
Hyperglycaemia	0	0	3	*1*	0	0
IMMUNE SYSTEM DISORDERS						
Myasthenia gravis	0	0	1	*1*	0	0
Pemphigoid	0	0	1	*1*	0	0
Psoriasis	0	0	1	*1*	0	0
GASTROINTESTINAL DISORDERS						
Diarrhoea	0	0	4	0	0	0
Dry mouth	0	0	2	0	0	0
MUSCULOSKELETAL AND CONNECTIVE TISSUE DISORDERS						
Arthralgia	0	0	6	0	0	0
Neck pain	0	0	2	0	0	0
RESPIRATORY, THORACIC AND MEDIASTINAL DISORDERS						
Dyspnoea	0	0	3	0	0	0
INVESTIGATIONS						
Alanine aminotransferase increased	0	0	2	0	0	0
Aspartate aminotransferase increased	0	0	2	0	0	0
Lymphocyte count decreased	0	0	2	*1*	0	0

AEs coded using MedDRA v21.1 for system organ class (SOC) and preferred term (PT).
Subjects with more than one occurrence of the same AE are counted once based on maximum severity.
All AEs deemed at least possibly related to PV-10 were Grade 1 or 2 except for a single occurrence of Grade 3 injection site pain.
All Grade 3 or higher AEs deemed at least possibly related to pembrolizumab were Grade 3 except for a single occurrence of exacerbation of myasthenia gravis (Grade 5).
All AEs deemed at least possibly related to the combination were Grade 1 except for a single occurrence of Grade 2 increase in TSH.

Study Treatment and Objective Response

PV-10 Dose Exposure (Main Cohort)

- PV-10 injection was limited to cutaneous and subcutaneous lesions only (nodal and visceral lesions were not injected)
- 85 lesions were injected among 23 subjects in 1–5 injection cycles

PV-10 Dose Exposure (Phase 1b Main Cohort ITT Population, N = 23)	Median	Mean	Range
Lesions Injected	2.0	3.1	1 – 21
Total Injections	5.0	11.6	1 – 82
Injection Cycles	4.0	3.7	1 – 5
Dose per Injection Cycle	1.9 mL	4.8 mL	0.15 – 15 mL

Response

- Response assessed by RECIST 1.1
- Target Lesions assigned at screening to represent each subject's disease burden
- Most subjects had substantial uninjected disease burden
- Robust response was observed after minimal intervention
 - 65% of subjects achieved an objective response (26% after ≤ 3 treatment cycles)
 - 30% of injected Target Lesions achieved CR after a single injection
 - 53% of injected Target Lesions achieved CR after 3 or fewer injections

RECIST Response
(Target Lesions and Overall per RECIST)

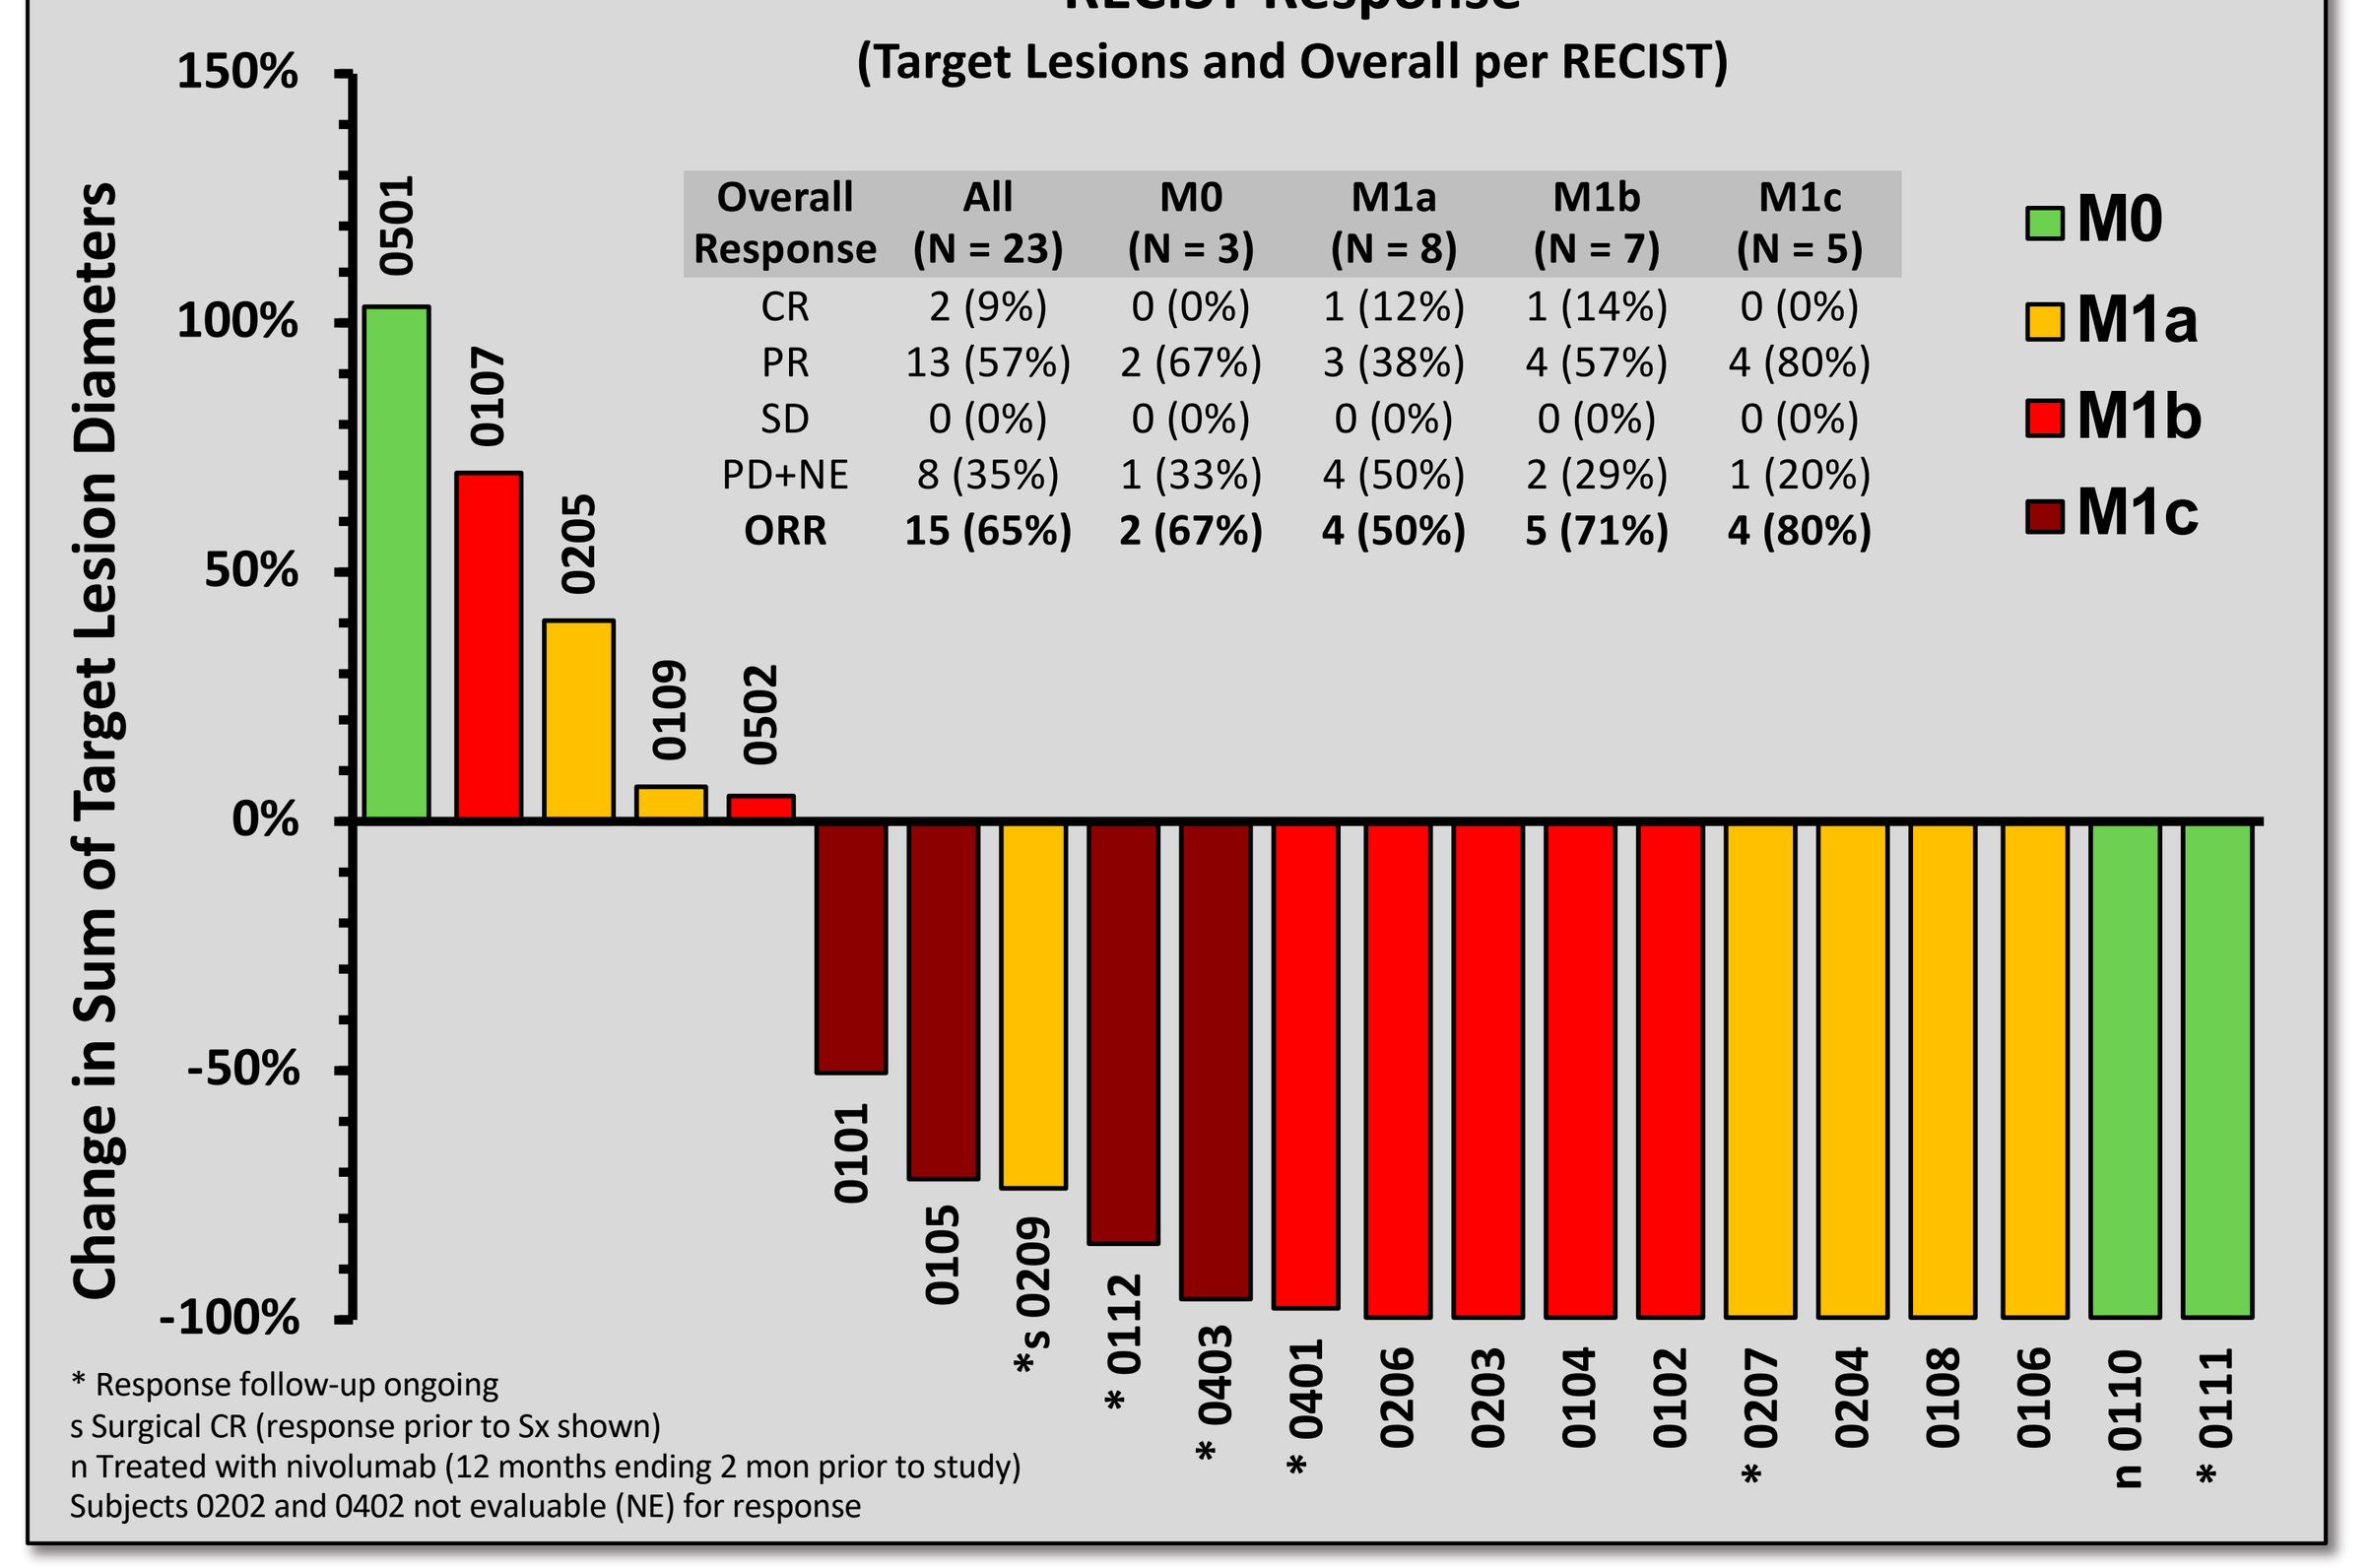
Change in Sum of Target Lesion Diameters
150%
100%
50%
0%
-50%
-100%
0501
0107
0205
0109
0502
0101
0105
*s 0209
* 0112
* 0403
* 0401
0206
0203
0104
0102
* 0207
0204
0108
0106
n 0110
* 0111
M0
M1a
M1b
M1c

Overall Response	All (N = 23)	M0 (N = 3)	M1a (N = 8)	M1b (N = 7)	M1c (N = 5)
CR	2 (9%)	0 (0%)	1 (12%)	1 (14%)	0 (0%)
PR	13 (57%)	2 (67%)	3 (38%)	4 (57%)	4 (80%)
SD	0 (0%)	0 (0%)	0 (0%)	0 (0%)	0 (0%)
PD+NE	8 (35%)	1 (33%)	4 (50%)	2 (29%)	1 (20%)
ORR	**15 (65%)**	**2 (67%)**	**4 (50%)**	**5 (71%)**	**4 (80%)**

* Response follow-up ongoing
s Surgical CR (response prior to Sx shown)
n Treated with nivolumab (12 months ending 2 mon prior to study)
Subjects 0202 and 0402 not evaluable (NE) for response

Injected Target Lesion Response
(N = 30 Injected Target Lesions in 23 Subjects)

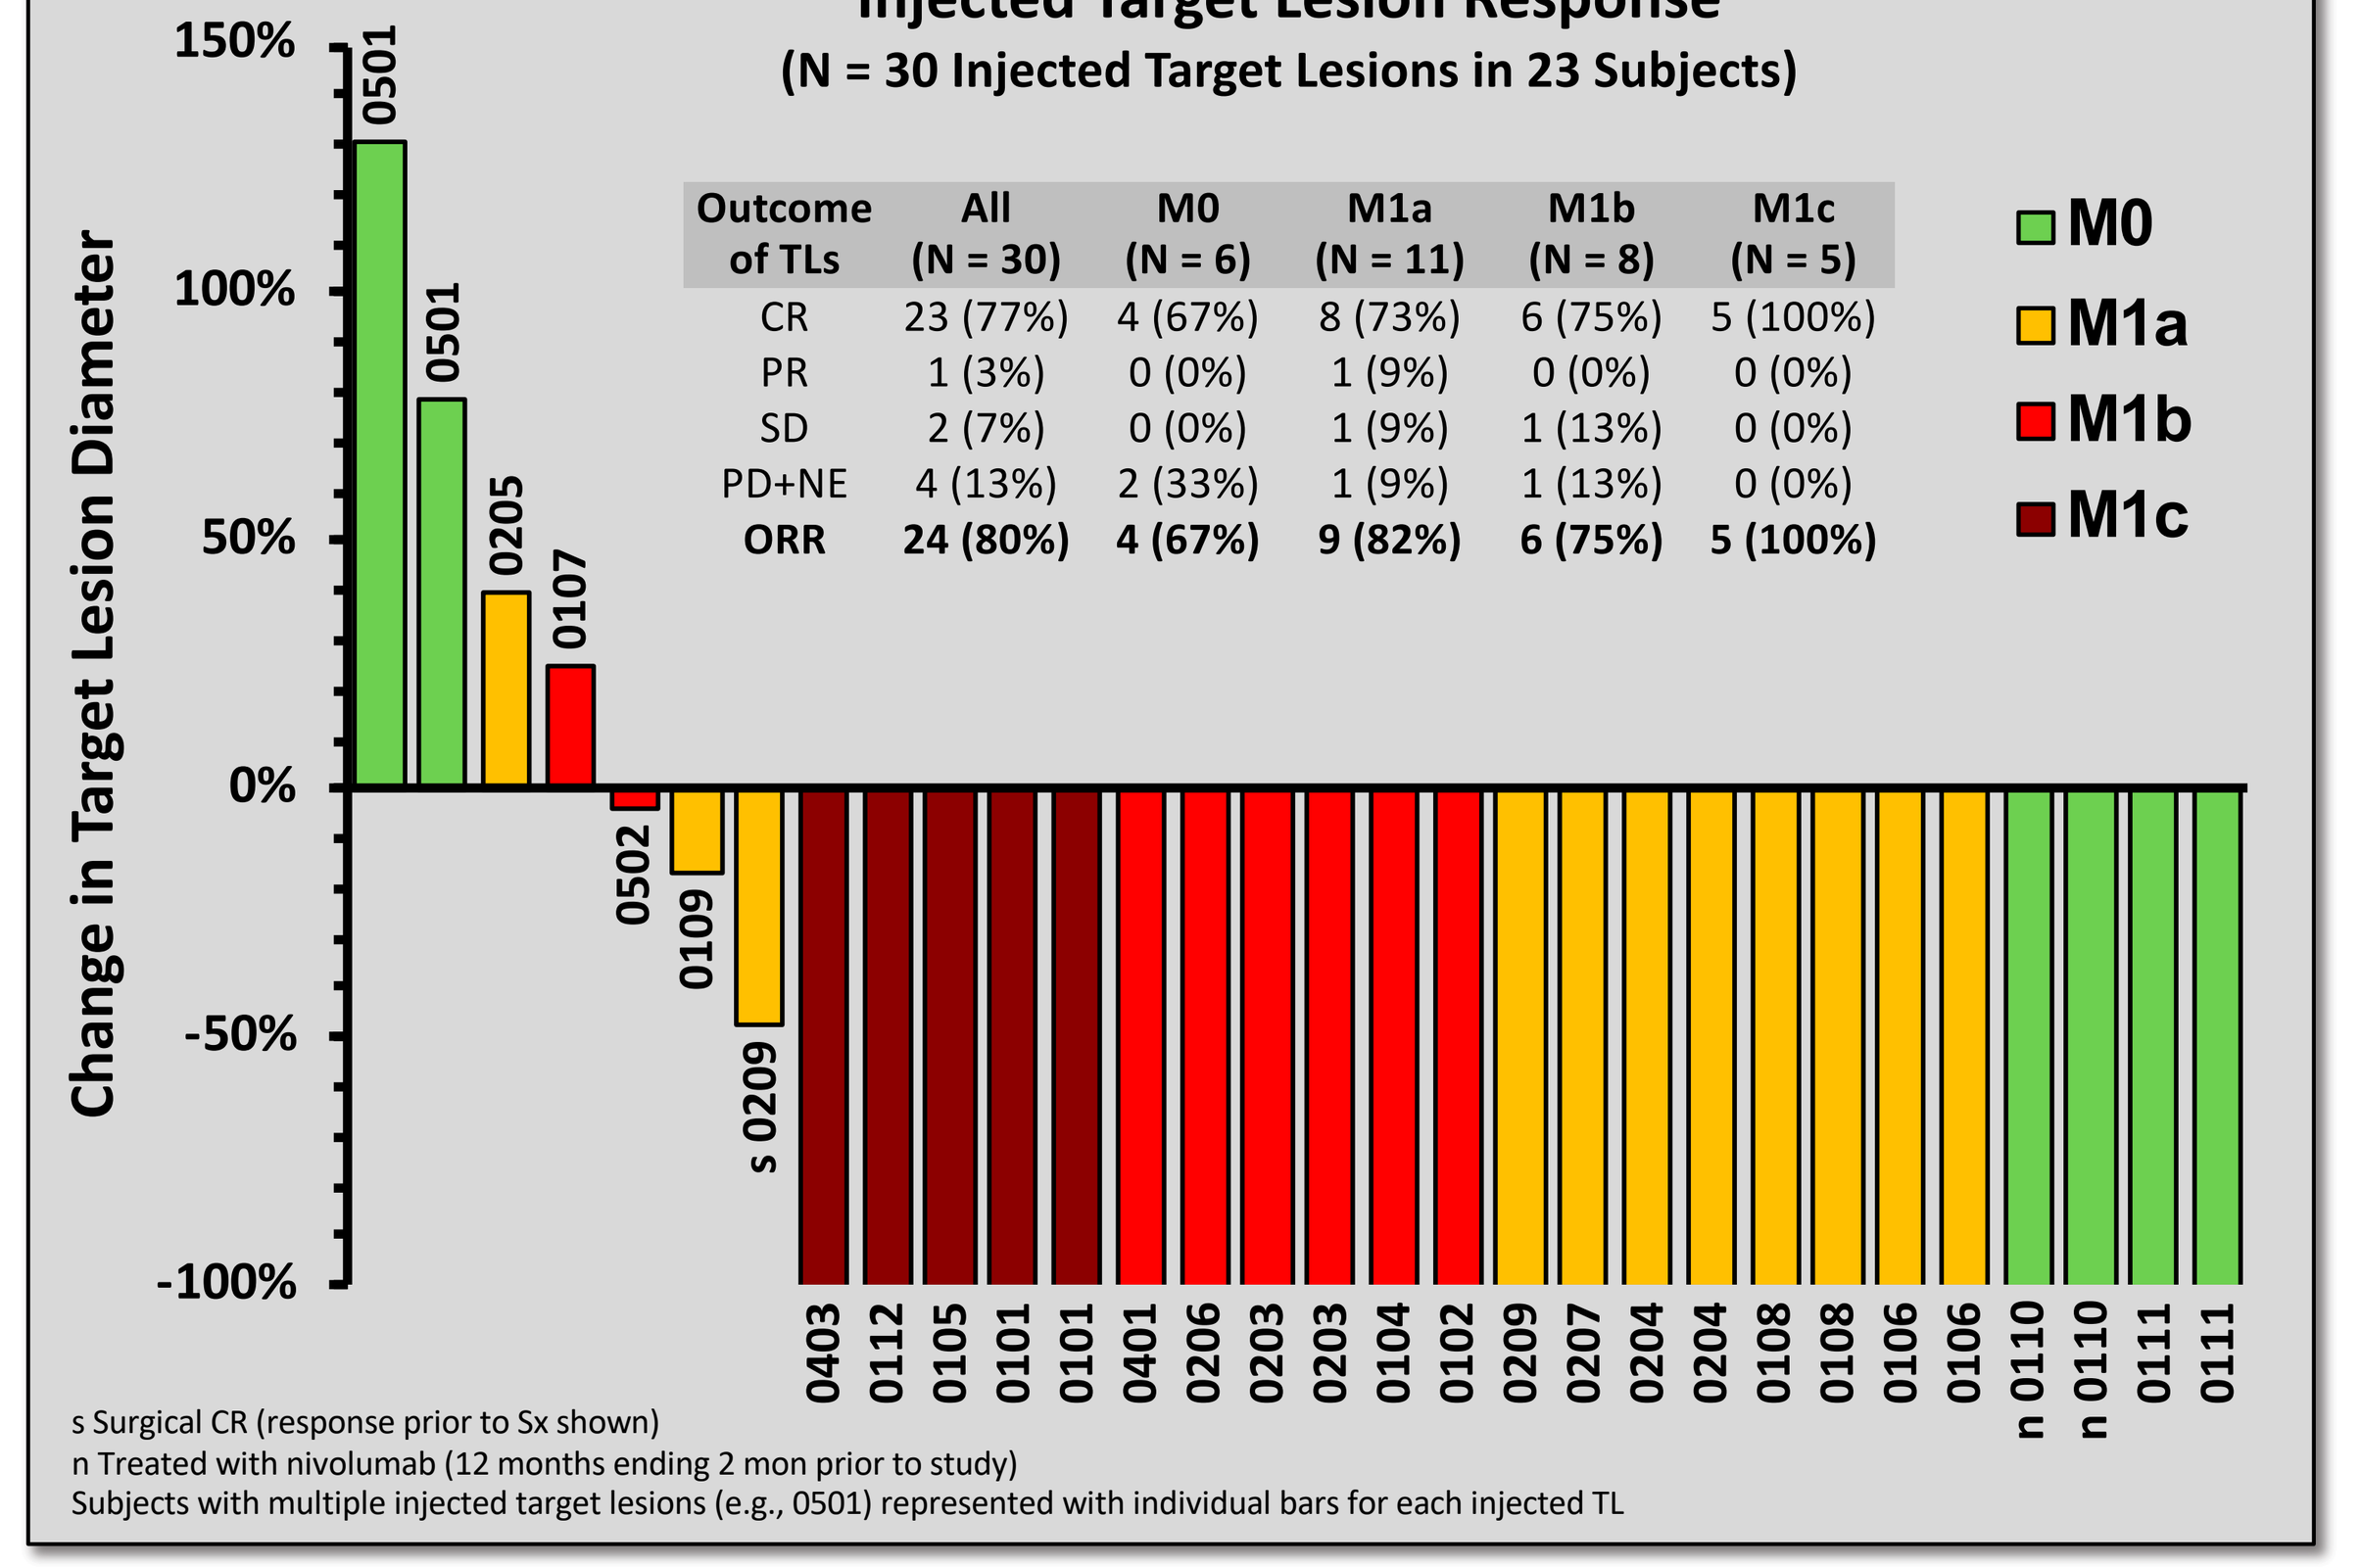
Change in Target Lesion Diameter
150%
100%
50%
0%
-50%
-100%
0501
0501
0205
0107
0502
0109
s 0209
0403
0112
0105
0101
0101
0401
0206
0203
0203
0104
0102
0209
0207
0204
0204
0108
0108
0106
0106
n 0110
n 0110
0111
0111
M0
M1a
M1b
M1c

Outcome of TLs	All (N = 30)	M0 (N = 6)	M1a (N = 11)	M1b (N = 8)	M1c (N = 5)
CR	23 (77%)	4 (67%)	8 (73%)	6 (75%)	5 (100%)
PR	1 (3%)	0 (0%)	1 (9%)	0 (0%)	0 (0%)
SD	2 (7%)	0 (0%)	1 (9%)	1 (13%)	0 (0%)
PD+NE	4 (13%)	2 (33%)	1 (9%)	1 (13%)	0 (0%)
ORR	**24 (80%)**	**4 (67%)**	**9 (82%)**	**6 (75%)**	**5 (100%)**

s Surgical CR (response prior to Sx shown)
n Treated with nivolumab (12 months ending 2 mon prior to study)
Subjects with multiple injected target lesions (e.g., 0501) represented with individual bars for each injected TL

Treatment Pattern and Response Characteristics

Change in Target Lesion Tumor Burden by RECIST 1.1

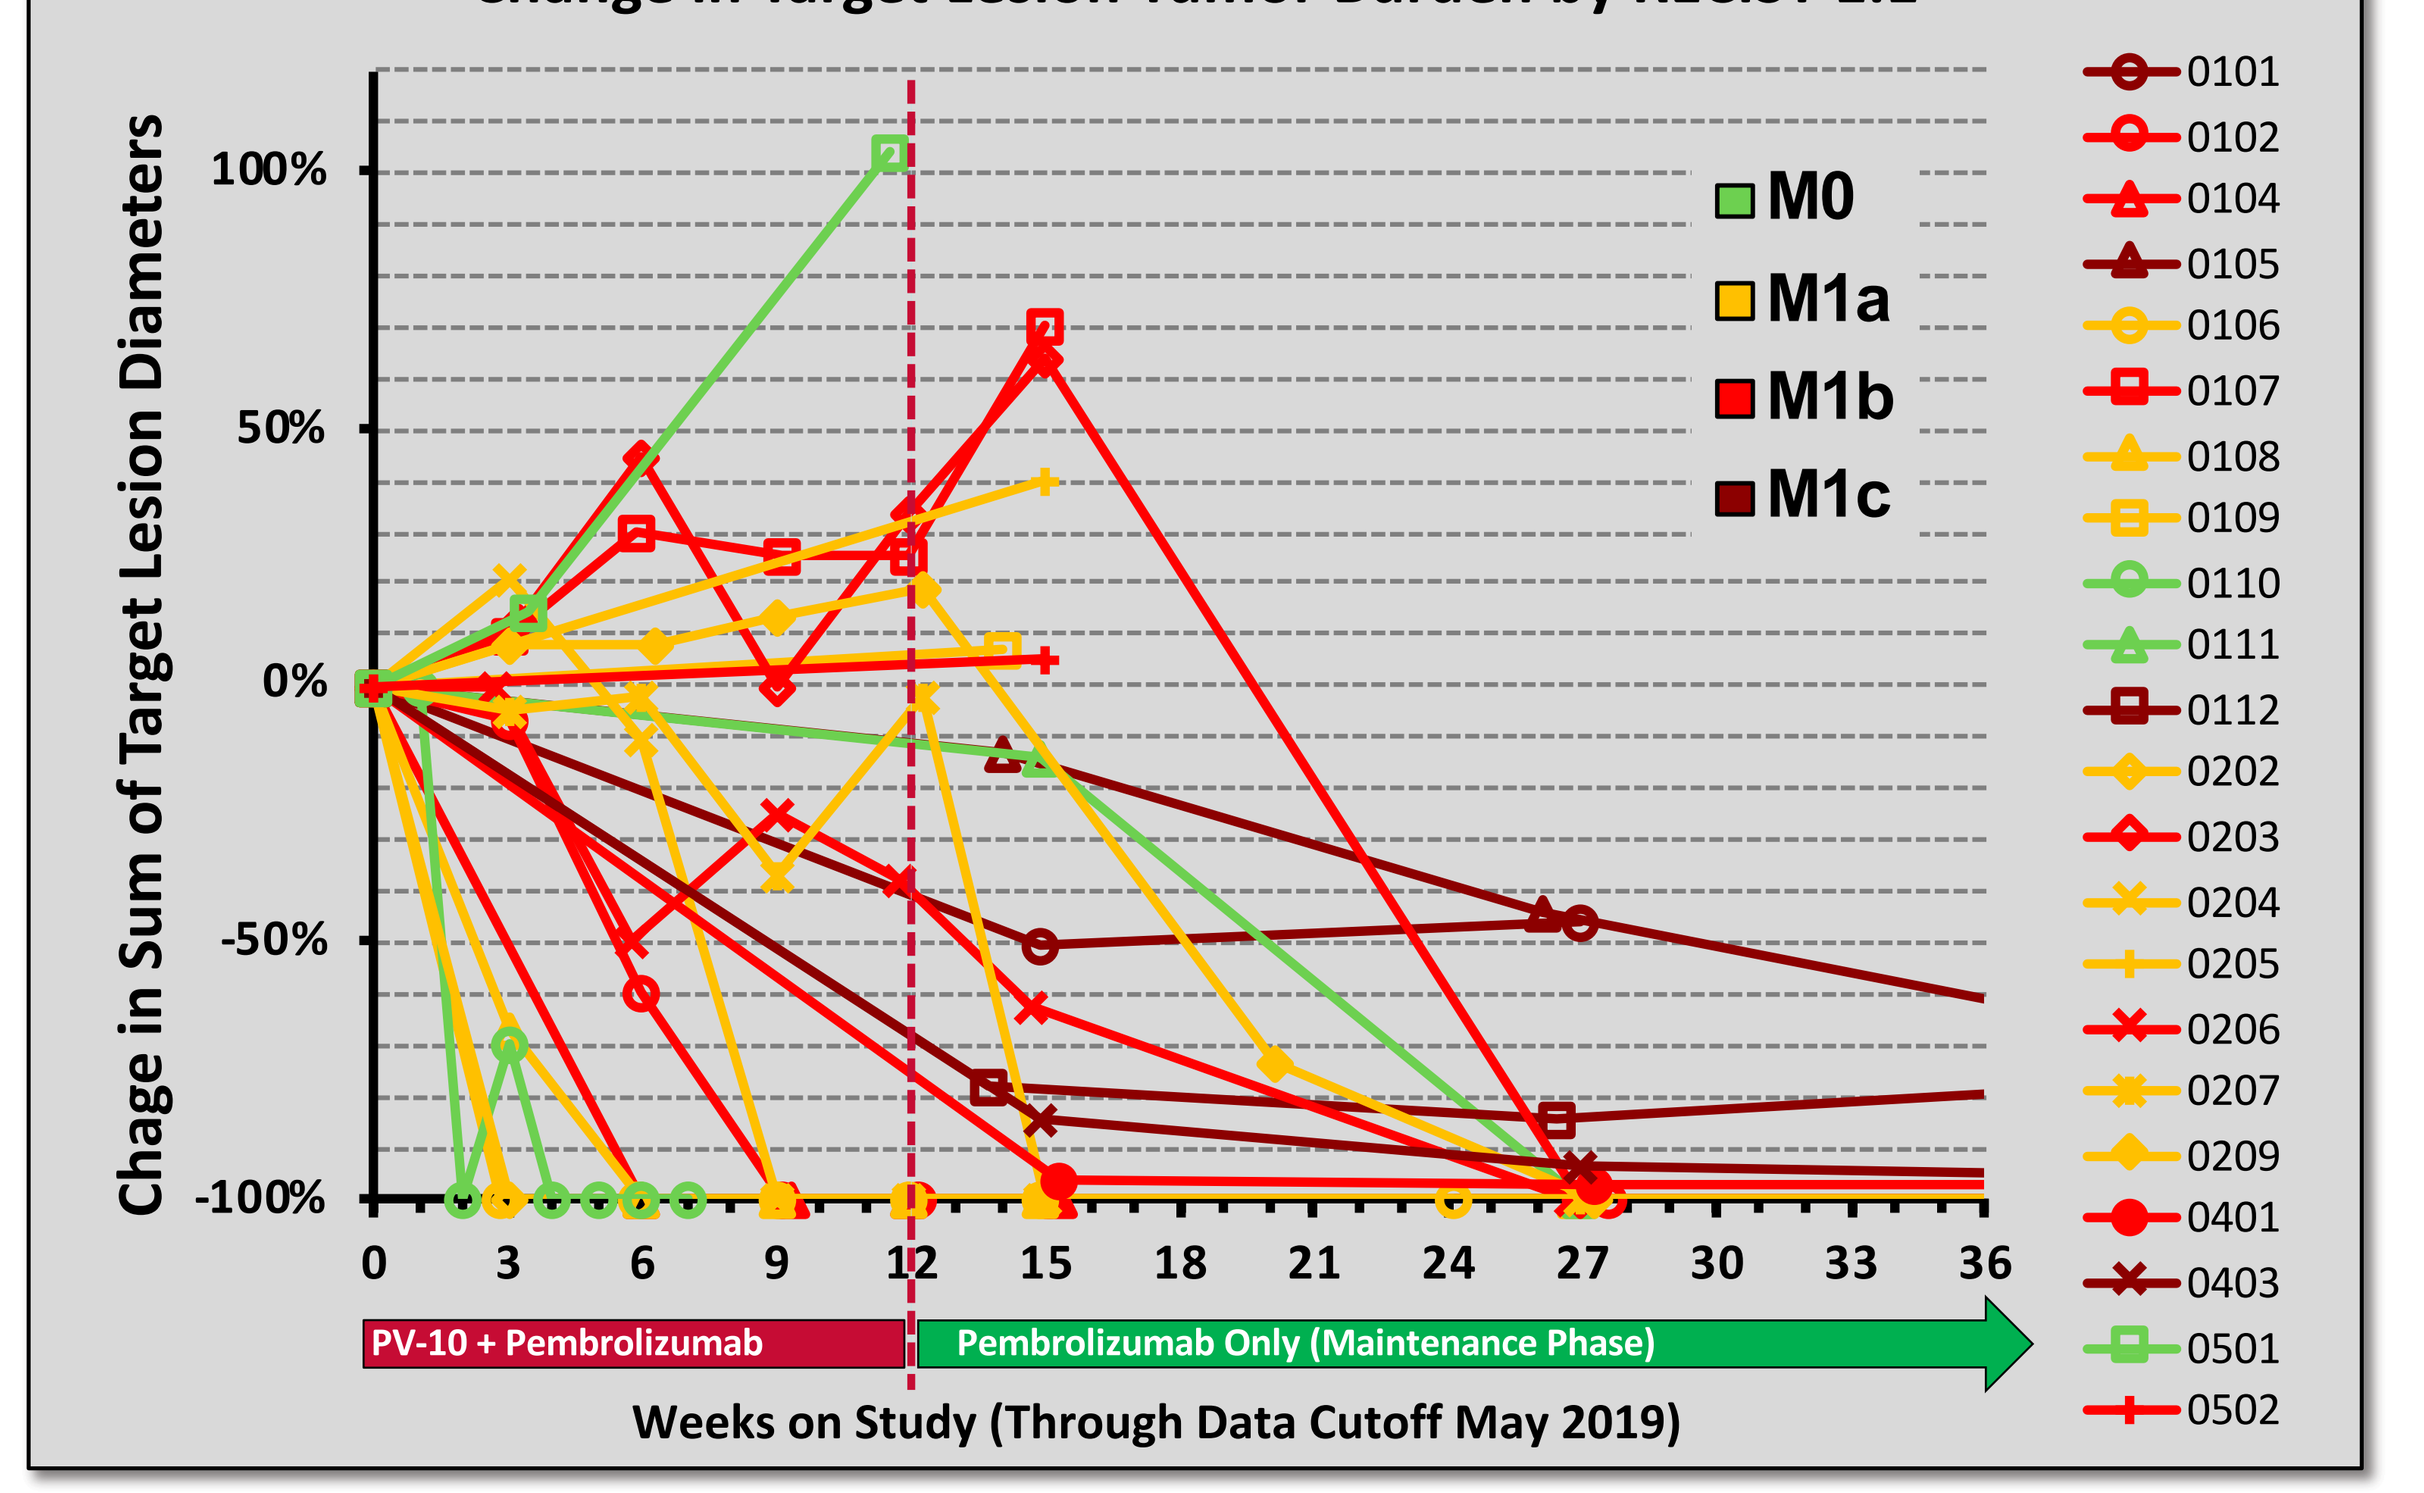
Chage in Sum of Target Lesion Diameters
100%
50%
0%
-50%
-100%
0
3
6
9
12
15
18
21
24
27
30
33
36
PV-10 + Pembrolizumab
Pembrolizumab Only (Maintenance Phase)
Weeks on Study (Through Data Cutoff May 2019)
M0
M1a
M1b
M1c
0101
0102
0104
0105
0106
0107
0108
0109
0110
0111
0112
0202
0203
0204
0205
0206
0207
0209
0401
0403
0501
0502

Treatment Pattern and Duration of Response

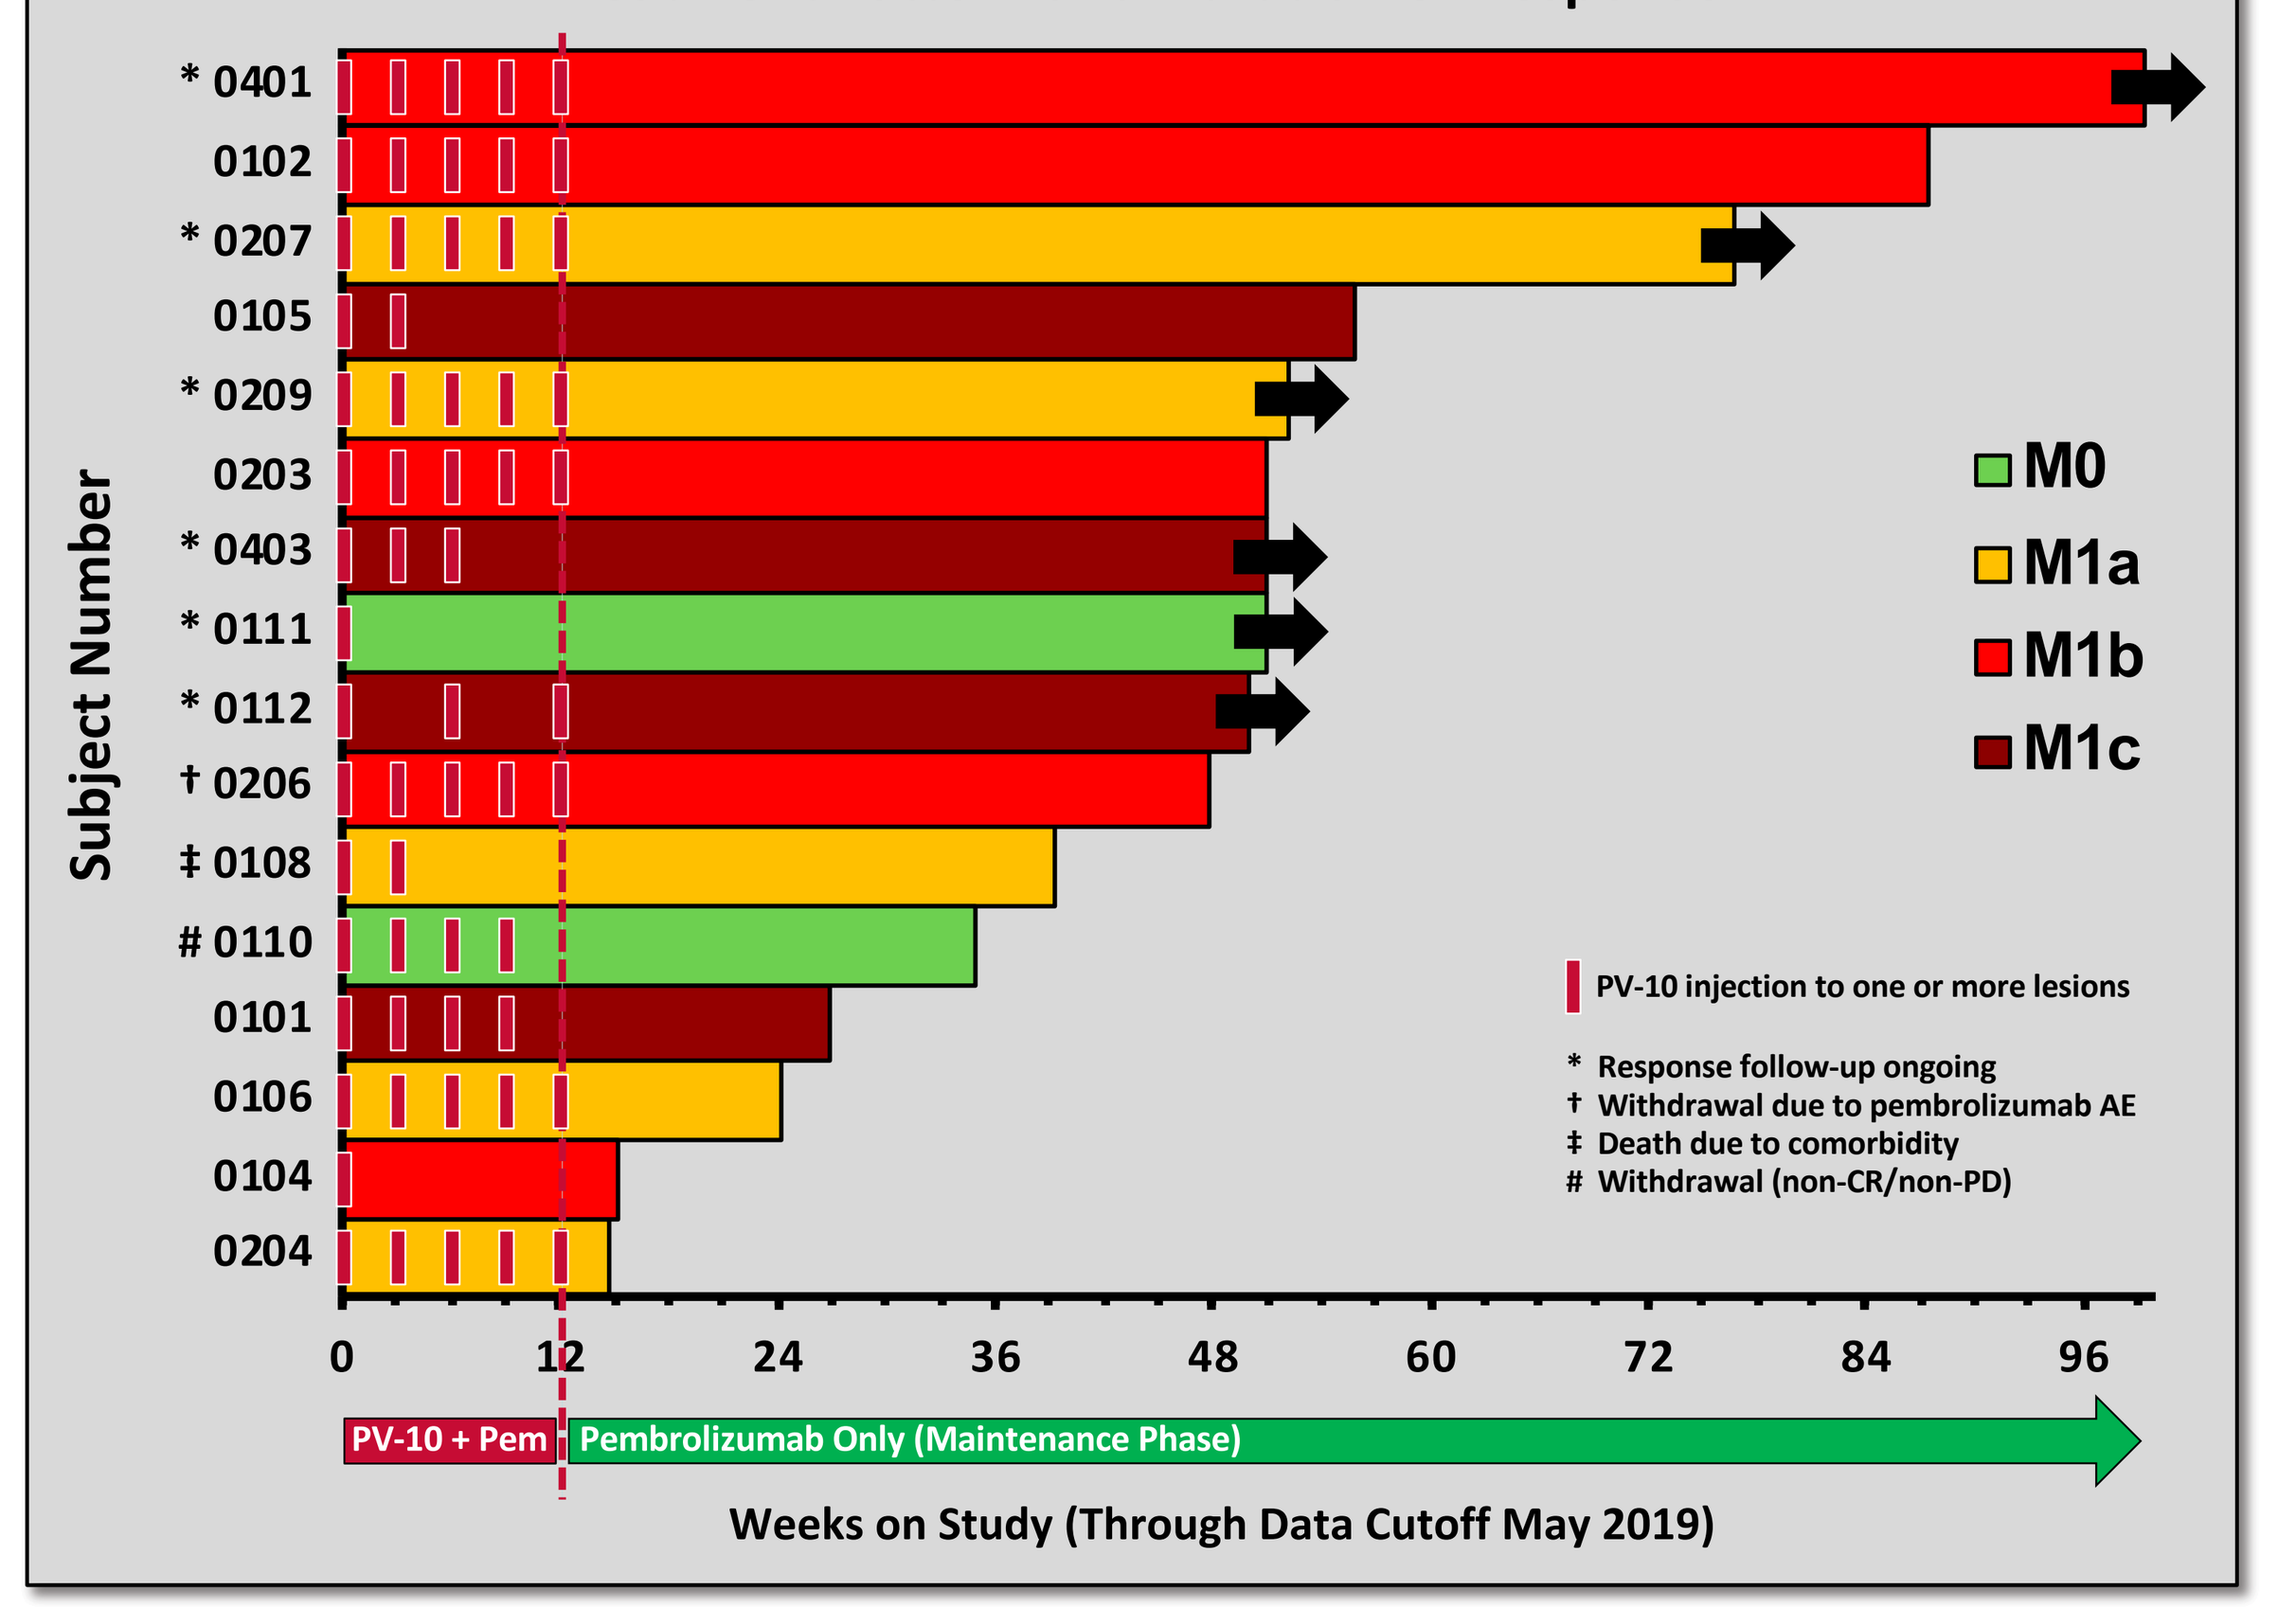
Subject Number
* 0401
0102
* 0207
0105
* 0209
0203
* 0403
* 0111
* 0112
† 0206
‡ 0108
0110
0101
0106
0104
0204
0
12
24
36
48
60
72
84
96
PV-10 + Pem
Pembrolizumab Only (Maintenance Phase)
Weeks on Study (Through Data Cutoff May 2019)
M0
M1a
M1b
M1c
PV-10 injection to one or more lesions
* Response follow-up ongoing
† Withdrawal due to pembrolizumab AE
‡ Death due to comorbidity
Withdrawal (non-CR/non-PD)

Progression-Free Survival (PFS) – Preliminary Results

Study	N	Stage IV Pts	CR	PR	ORR	PFS (months)
PV-10 + Pembrolizumab (current study)	23	87%	9%	57%	65%	12.3 [a]
Talimogene Laherparepvec + Pembrolizumab [b]	21	62%	24%	33%	57%	>12
SD-101 + Pembrolizumab (single injection/cycle) [c]	39	77%	3%	36%	39%	4.2
SD-101 + Pembrolizumab (four injections/cycle) [c]	37	59%	17%	53%	70%	NR
Pembrolizumab Single Agent [d]	556	97%	6%	28%	34%	4.1-5.5
Nivolumab Single-Agent [e]	210	NR	8%	32%	40%	5.1
Nivolumab + Ipilimumab [f]	95	89%	22%	37%	59%	>12

[a] Preliminary estimate through data cutoff May 2019. [b] Long et al., ASCO 2016 (abstract 9568). [c] Ribas et al., ASCO 2018 (abstract 9513). [d] Robert et al., NEJM 2015; 372: 2521. [e] Robert et al., NEJM 2015; 372: 320. [f] Postow et al., NEJM 2015; 372: 2006.

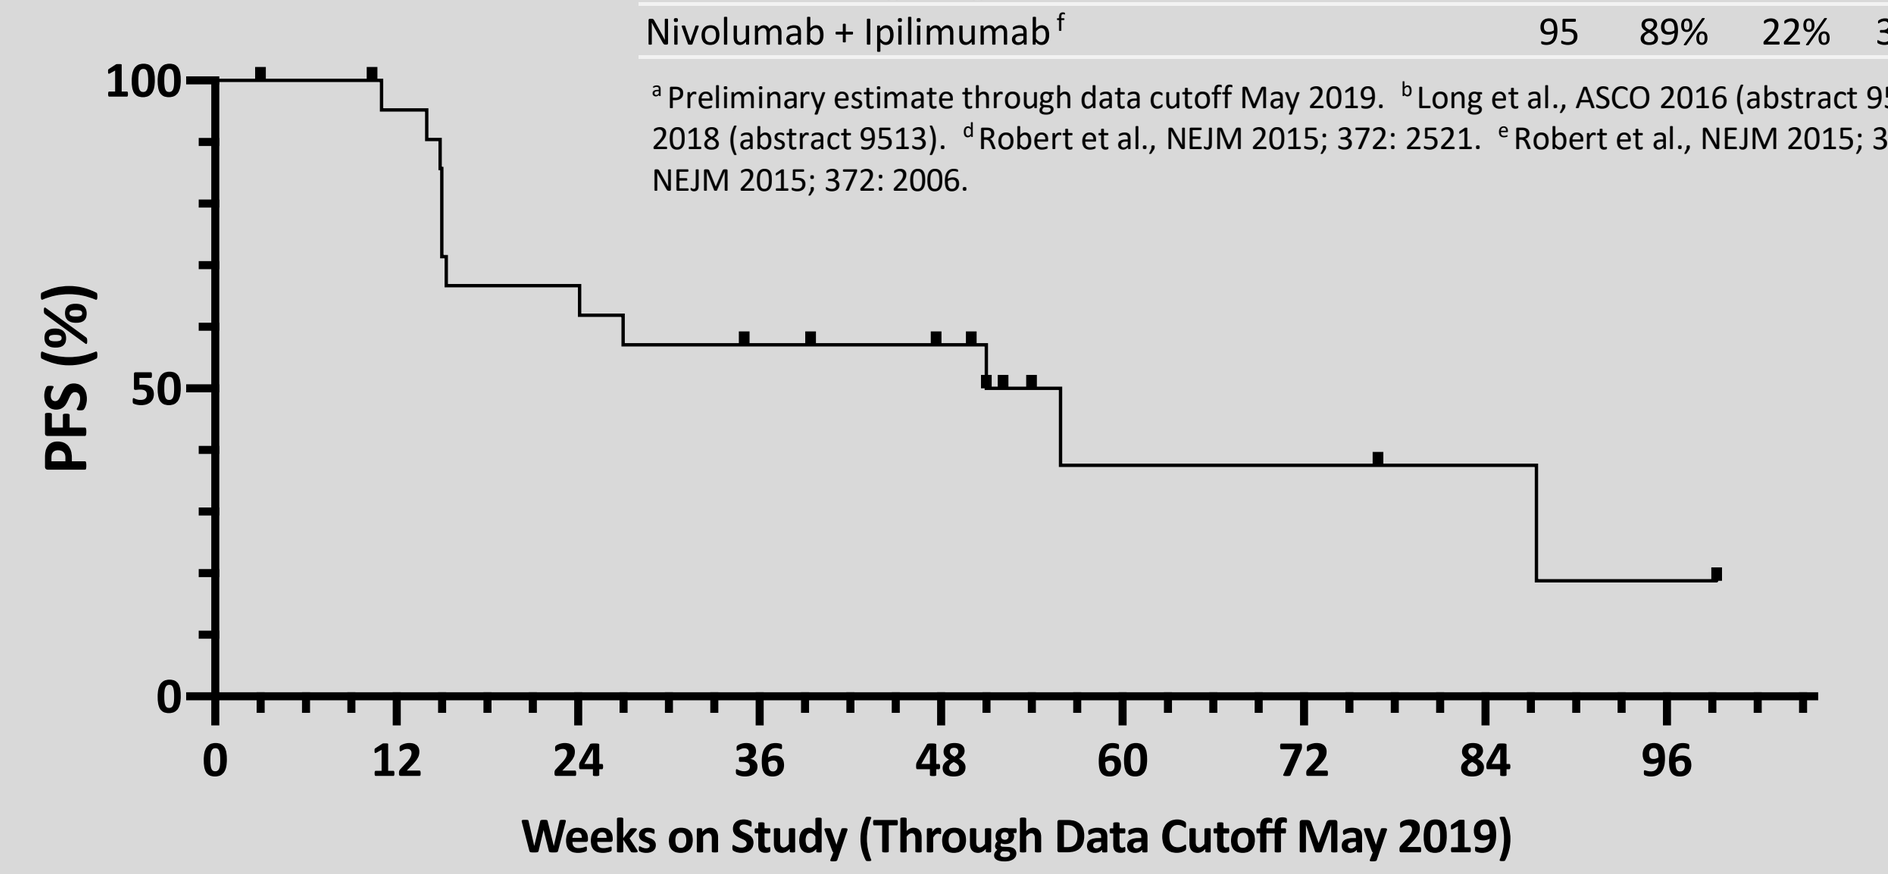
PFS (%)
100
50
0
0
12
24
36
48
60
72
84
96
Weeks on Study (Through Data Cutoff May 2019)

Progression

Disease Progression in Main Cohort

- PV-10 injection was limited to five cycles during the first 12 weeks of study
- Most progressions occurred after completion of PV-10, often at injectable sites

Pattern of Progression
(Phase 1b Main Cohort ITT Population, N = 23)

Subject	Stage	BOR	PFS (weeks)	Site of Progression: Existing Lesions [a]	Site of Progression: Local Recurrence [b]	Site of Progression: New Lesions [c]
0501	IIIC	PD	11.0			LN, SQ
0109	M1a	SD	14.0	LN	Y	LN
0204	M1a	CR	14.9			Lung, Liver, Bone
0107	M1b	PD	15.0	Lung		LN, Adrenal
0205	M1a	SD	15.0			LN, SQ
0502	M1b	SD	15.0			Liver
0104	M1b	CR	15.3			LN, Lung
0106	M1a	CR	24.1			CUT
0101	M1c	PR	27.0	LN, SQ		Lung
0203	M1b	CR	51.0			CNS
0105	M1c	PR	54.1	SQ, Liver		
0102	M1b	CR	87.4			LN

Abbreviations: BOR, best overall response; CNS, central nervous system; CUT, cutaneous; LN, lymph node; SQ, subcutaneous; Y, yes.
[a] Progression occurred in one or more lesions present at baseline.
[b] Progression occurred at location previously treated with PV-10.
[c] Progression occurred in one or more lesions not present at baseline.

Peripheral Blood Mononuclear Cells (PBMCs)

Changes occurred in PBMCs upon initiation of combination therapy

- Activated T cell population increased during the treatment interval
- NK and NKT cell populations exhibited transient increases one week after PV-10 injection (Weeks 1 and 4)
- Cytotoxic T cell, helper T cell and total T cell populations were stable
- These changes are similar to those observed for single-agent PV-10 [2]

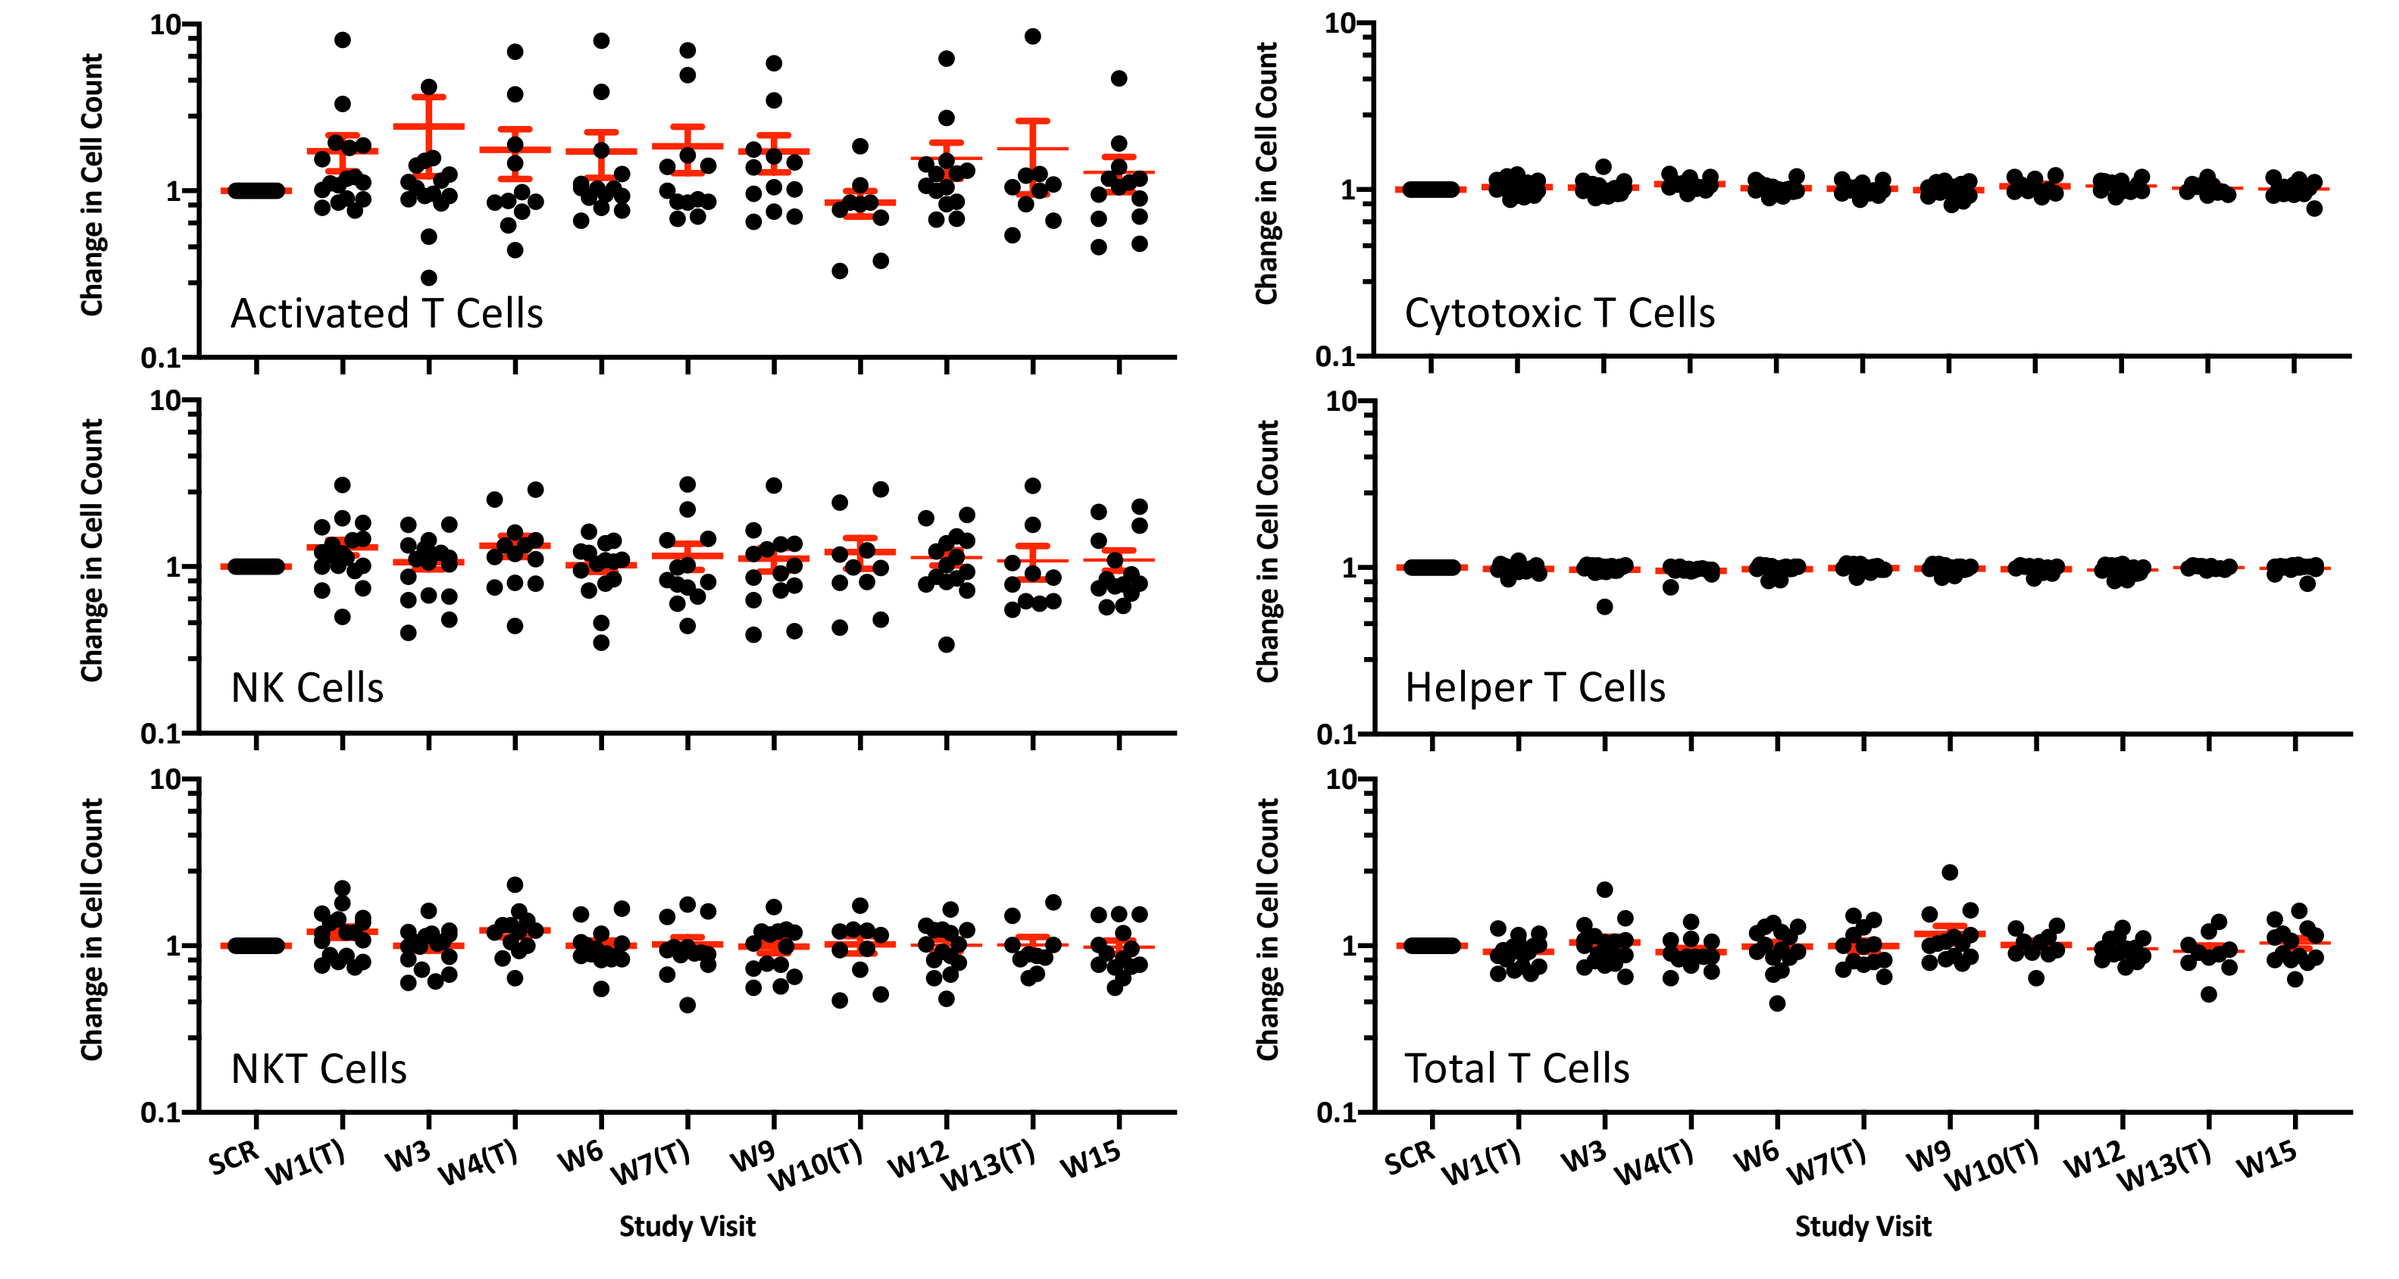
Change in Cell Count
10
1
0.1
Activated T Cells
Cytotoxic T Cells
NK Cells
Helper T Cells
NKT Cells
Total T Cells
SCR
W1(T)
W3
W4(T)
W6
W7(T)
W9
W10(T)
W12
W13(T)
W15
Study Visit

Conclusions

- Treatment-Emergent Adverse Events were consistent with established patterns for each drug, principally Grade 1-2 injection site reactions attributed to PV-10 and Grade 1-3 immune-mediated reactions attributed to pembrolizumab, with no significant overlap or unexpected toxicities
- Response rate (9% CR, 65% ORR) and durability of response (PFS estimated at 12.3 months) were superior to either therapy alone, and response was consistent across all disease stages in a patient population with substantial uninjected disease burden
- Extended treatment with PV-10 may further improve response rate and durability
- Two Phase 1b Expansion Cohorts (24 pts each) have been opened to patients refractory to prior checkpoint inhibition and with in-transit or satellite disease
- These data support initiation of the Phase 2 randomized controlled portion of the trial to further evaluate and demonstrate the benefit of the combination

Citation: J Clin Oncol 2019; 37 (suppl; abstr 9559)