Generation of an Antitumor Response and Immunity Using a Small Molecule Drug (PV-10)

Abstract No. 1452582

Society for Immunotherapy of Cancer (SITC) Annual Meeting
26 – 28 October 2012

C. Dees, S. Blair, J. Harkins, T.C. Scott and E.A. Wachter

Provectus Pharmaceuticals, Inc., Knoxville, TN USA

For additional information:

Craig Dees (dees@pvct.com)

Abstract (Updated)

Generation of an Antitumor Response and Immunity using a Small Molecule Drug (PV-10)

PV-10 (10% Rose Bengal in 0.9% saline) has been used to chemoablate a wide variety of tumors in human clinical trials and in animal patients when delivered by intralesional (IL) injection [1,2]. PV-10 exhibits direct anti-tumoral activity targeted only on tumor tissue while sparing normal tissue. Additionally, PV-10 has stimulated the resolution of remote untreated tumors by an apparent immune-mediated antitumor response, including cutaneous and lung metastases. Therefore, we investigated the mechanism by which this remote "bystander effect" is produced using immunocompetent and immunodeficient mice. Tumor models examined included: hepatocellular carcinoma (HCC), melanoma, pancreatic and colon adenocarcinomas. PV-10 was found to chemoablate all tumors tested with no significant side effects. No response was achieved in control tumors injected with vehicle only. Durable immunity was produced to the cell line that was ablated, however tumors of different origin could be established in treated mice. Immunity to establishment of a tumor could be transferred using spleen cells from mice whose tumors had been previously ablated. Untreated tumors in the opposite flank of mice completely regressed or were reduced in size in immunocompetent mice (i.e., 8/8 untreated HCC tumors), but no remote tumors were observed to resolve in nude mice when a second tumor was treated by PV-10 chemoablation. In conclusion, resolution of untreated tumors is dependent on an immune mechanism that appears to involve immune cell mediation. We hypothesize that production of a vaccine-like immune response using a small molecule drug is possible and requires: 1) an intralesional route of injection that generates rapid, durable tumor destruction via autolysis [3]; 2) rapid clearance of drug from normal tissue; and 3) antitumor effects targeted only to tumor tissue. PV-10 appears to meet each of these requirements. Co-administration of PV-10 immuno-chemoablation with other systemic therapy can yield potent synergy in uninjected tumors.

Recent Reports of Immunologic Activity

Murine Model: C57BL/6 females with B16-F10 murine melanoma

Tumor Induction: Solitary subcutaneous (SQ) tumors were induced in one flank by injection of tumor cells (1 x 10^5 cells per tumor). In some animals, lung metastases were produced by synchronous injection of tumor cells into the tail vein (2.5 x 10^5 cells) while in another cohort, a second SQ tumor was implanted in the opposite flank (1 x 10^5 cells).

Treatment: 50 µL of PV-10 or phosphate buffered saline (PBS, control) was administered via IL injection to one SQ tumor 7 days after tumor implantation.

Follow up: 14 days post PV-10 or PBS injection, lungs were harvested and metastases counted. In some animals, splenocytes were assessed to measure the production of interferon-γ.

	Number of Lung Metastases
Controls	>250
	>250
	>250
	>250
	>250
PV-10	3
	0
	1
	>250
	>250

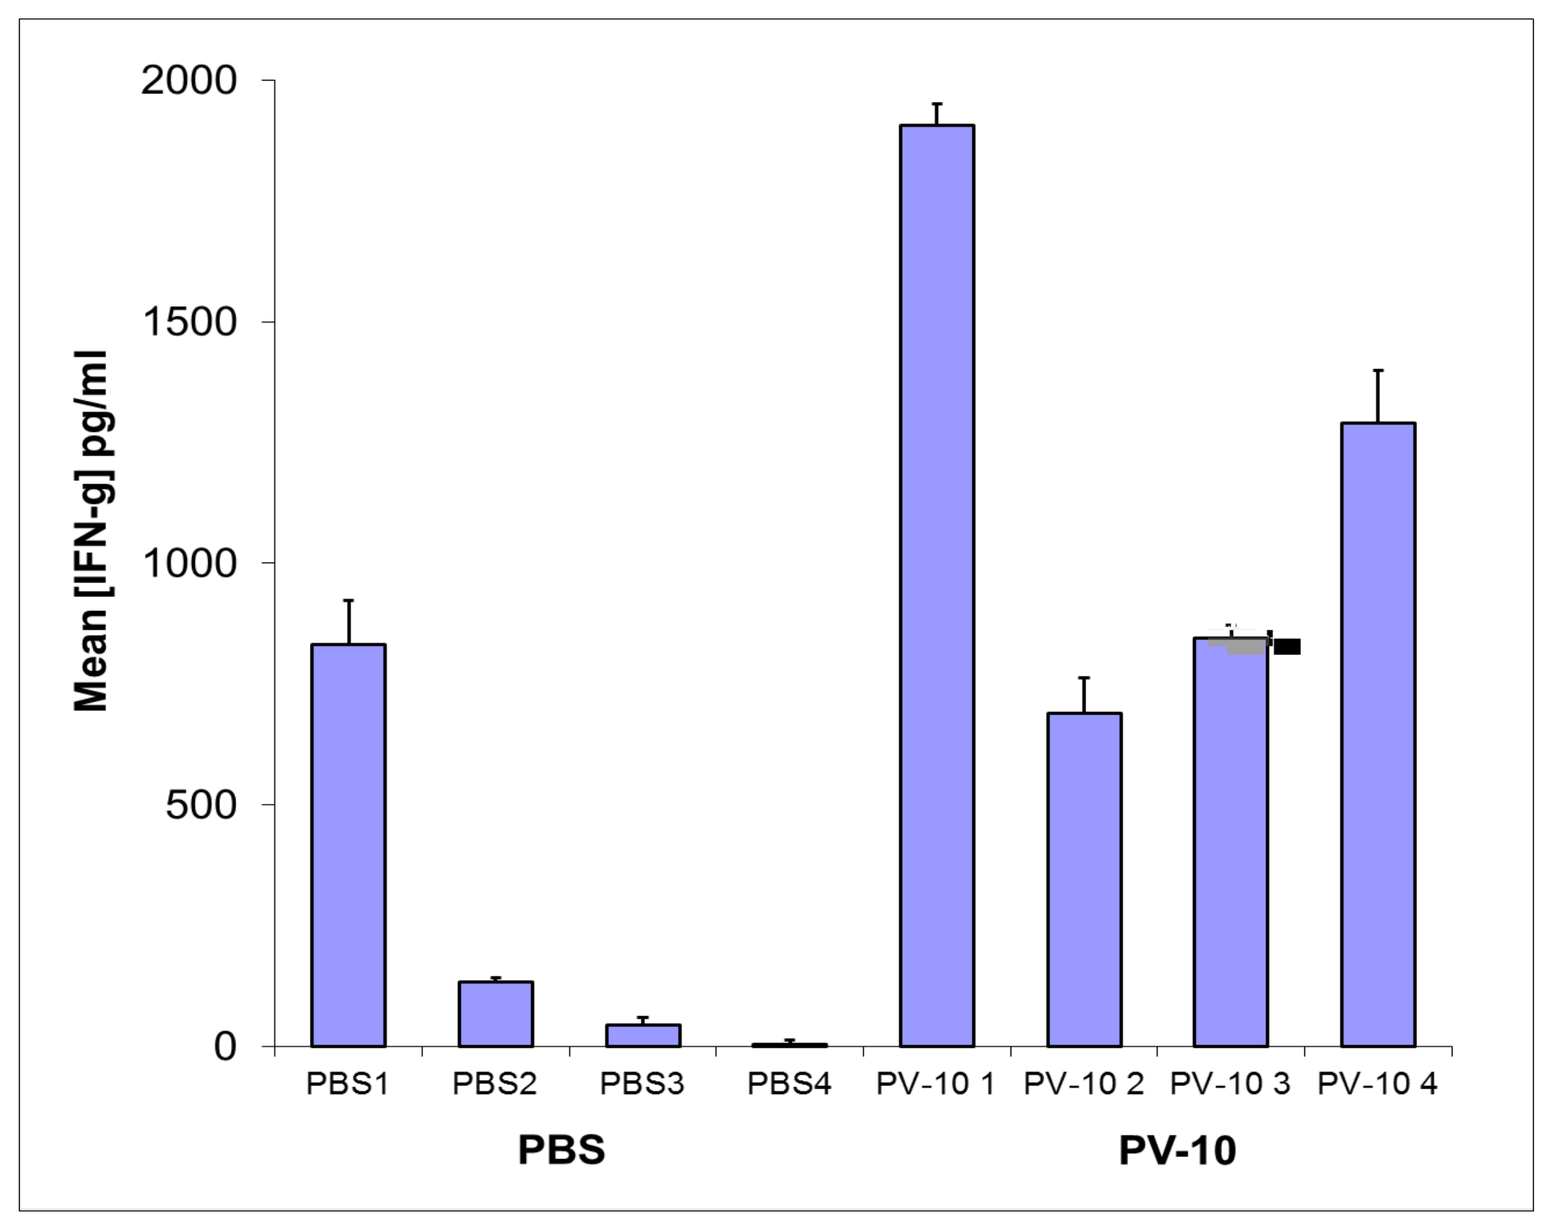
2000
1500
1000
500
0
Mean [IFN-g] pg/ml
PBS1
PBS2
PBS3
PBS4
PV-10 1
PV-10 2
PV-10 3
PV-10 4
PBS
PV-10

Toomey, Kodumudi, Martin, Mackay, Sarnaik and Pilon-Thomas, Ann Surg Oncol 2012; 19: S125

Conclusion: Ablation of SQ tumor with PV-10 led to significant reduction in synchronous lung metastases vs control (IL treatment with PBS). Following PV-10 treatment, splenocytes produced interferon-γ in response to B16-F10 challenge; minimal response was observed in control animals (IL treatment with PBS).

Additional data expected to be reported by Pilon-Thomas et al. in 2013

Statistical Methods

Tumor Area: Tumor areas (length x width) were analyzed using an Analysis of Covariance (ANCOVA) equal slopes model, with pre-treatment Study Day 0 values used as the covariate, to adjust for baseline differences. Normality was assessed by various plots of residuals and by the Shapiro-Wilk test, while homogenous group variance was assessed using Levene's test at Day 0 (assessments at the 1% level of significance and visual inspection of residual plots). Consistent violations of either assumption across time points would have resulted in an appropriate data transformation (e.g. log, rank, etc.) to correct the problem.

Survival Time and Time to Tumor Progression: Survival and tumor progression times (total tumor burden, targeted tumor and bystander tumor, using WHO criteria to define tumor progression) were analyzed using the Kaplan-Meier product-limit log-rank test.

Group Comparisons: Pairwise group differences for all statistical analyses were assessed using the Tukey-Kramer multiple comparisons adjustment, which is based on Tukey's Studentized Range Test. All statistical tests of pairwise treatment differences were conducted two-sided, at the 5% level of significance.

Statistical analyses performed by **Arbor Analytics, LLC**.

Arbor Analytics

Hepatocellular Carcinoma (Bilateral Model)

Murine Model: C57BL/6 females with Hepa 1-6 murine hepatocellular carcinoma

Tumor Induction: Tumors were induced in each flank by SQ injection of tumor cells (2 x 10^5 cells per tumor). After 12 days, mice that manifested bilateral flank tumors were randomized into one of 4 treatment groups (PV-10 alone, N = 4 mice; systemic 5-FU alone, N = 4; PV-10 + 5-FU combination, N = 5; or saline alone, N = 5) stratified by tumor size.

Treatment: A single IL injection of 50-100 µL of PV-10 (dose based on tumor volume) or 100 µL 0.9% saline control was administered to the larger of the two lesions on Study Day 0 (day of treatment). In the chemotherapy and combination therapy groups, 2 cycles of 30 mg/kg 5-Fluorouracil @ 50 mg/mL were administered intraperitoneally 3 times a week for 2 weeks starting on Study Day 0 and Day 39 [4].

Follow up: Each animal's tumor burden was evaluated pre-dosing on Study Day 0 and at intervals of 2-5 days thereafter. Tumors were measured with calipers to the nearest mm. Scabs and lesions that were no longer palpable were recorded as "0".

Outcome Assessment: Time to Tumor Progression was evaluated using modified WHO criteria for progression (mice were considered progressed when tumor area increased ≥ 25% and > 1 mm in any dimension). The tumors were evaluated separately and as total tumor burden (arithmetic sum). ANCOVA assessment was performed on injected (Target) and uninjected (Bystander) Lesions. P-values vs. control.

Time to Progression: Injected (Target) Tumors

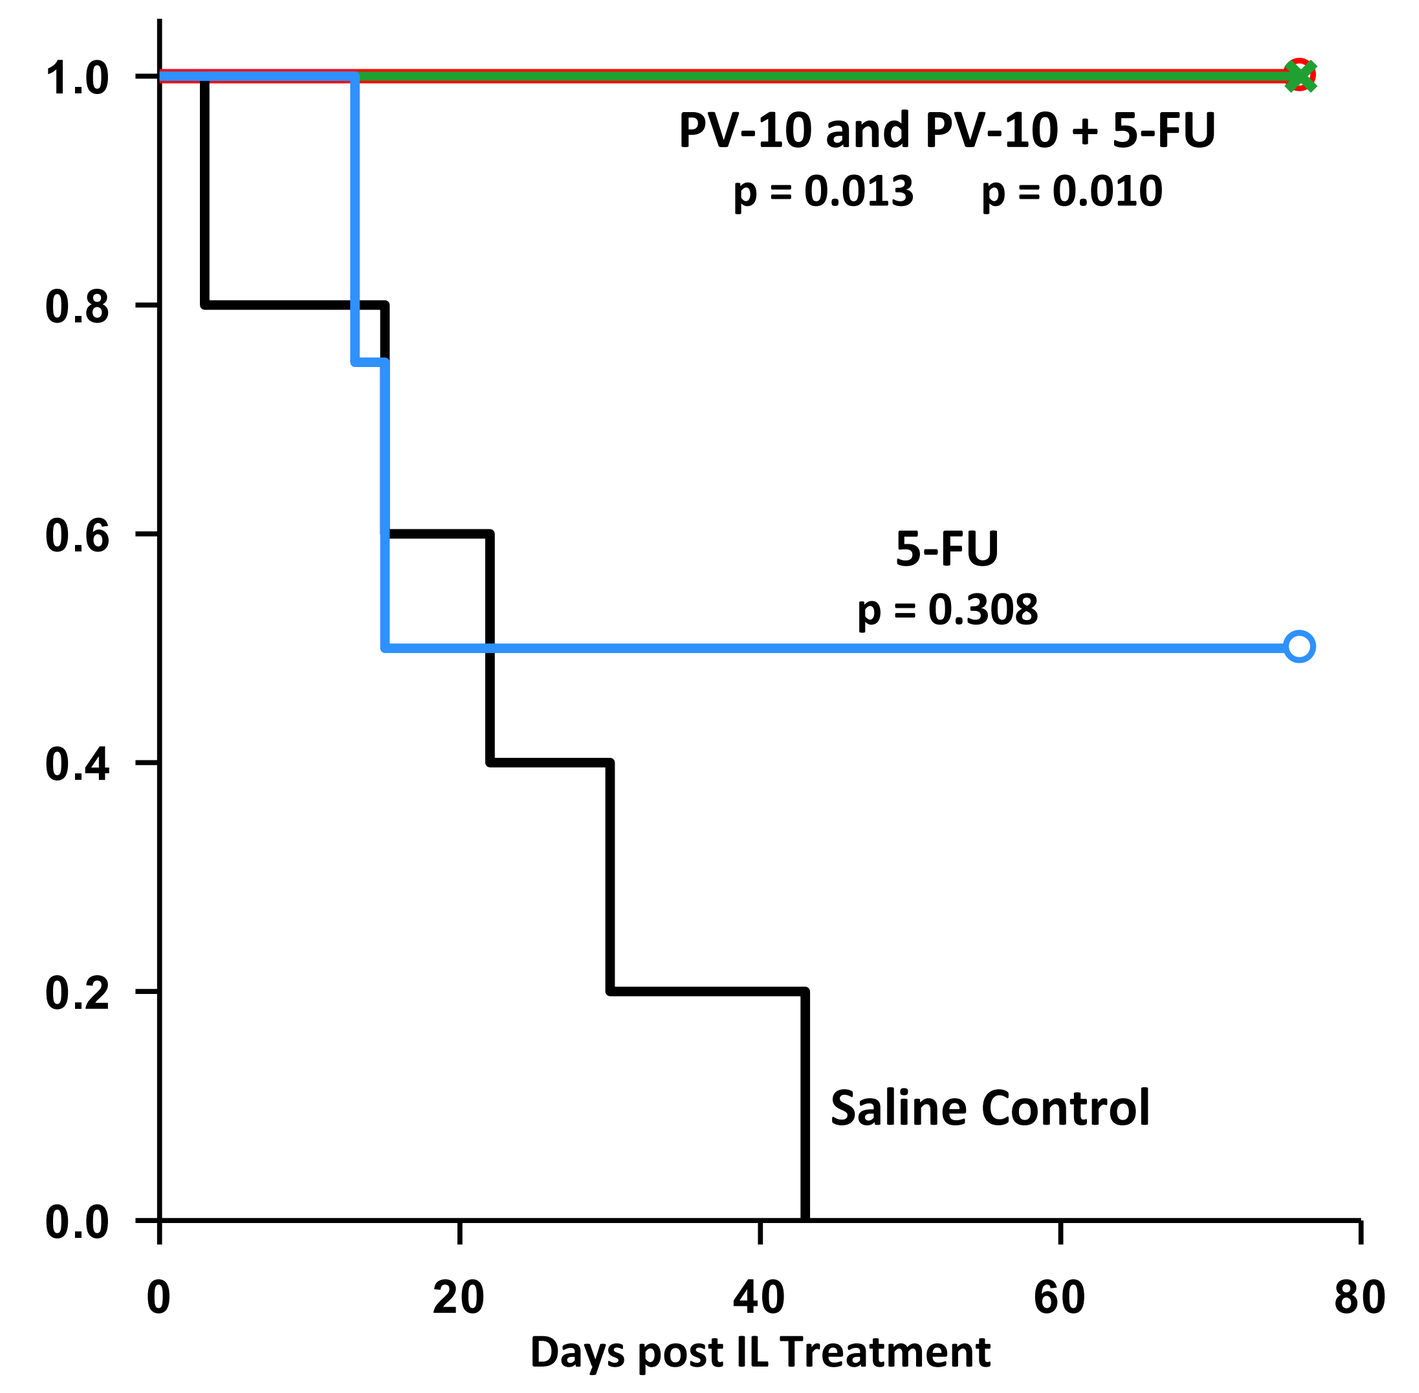
PV-10 and PV-10 + 5-FU
p = 0.013 p = 0.010
5-FU
p = 0.308
Saline Control
Days post IL Treatment

Time to Progression: Uninjected Tumors

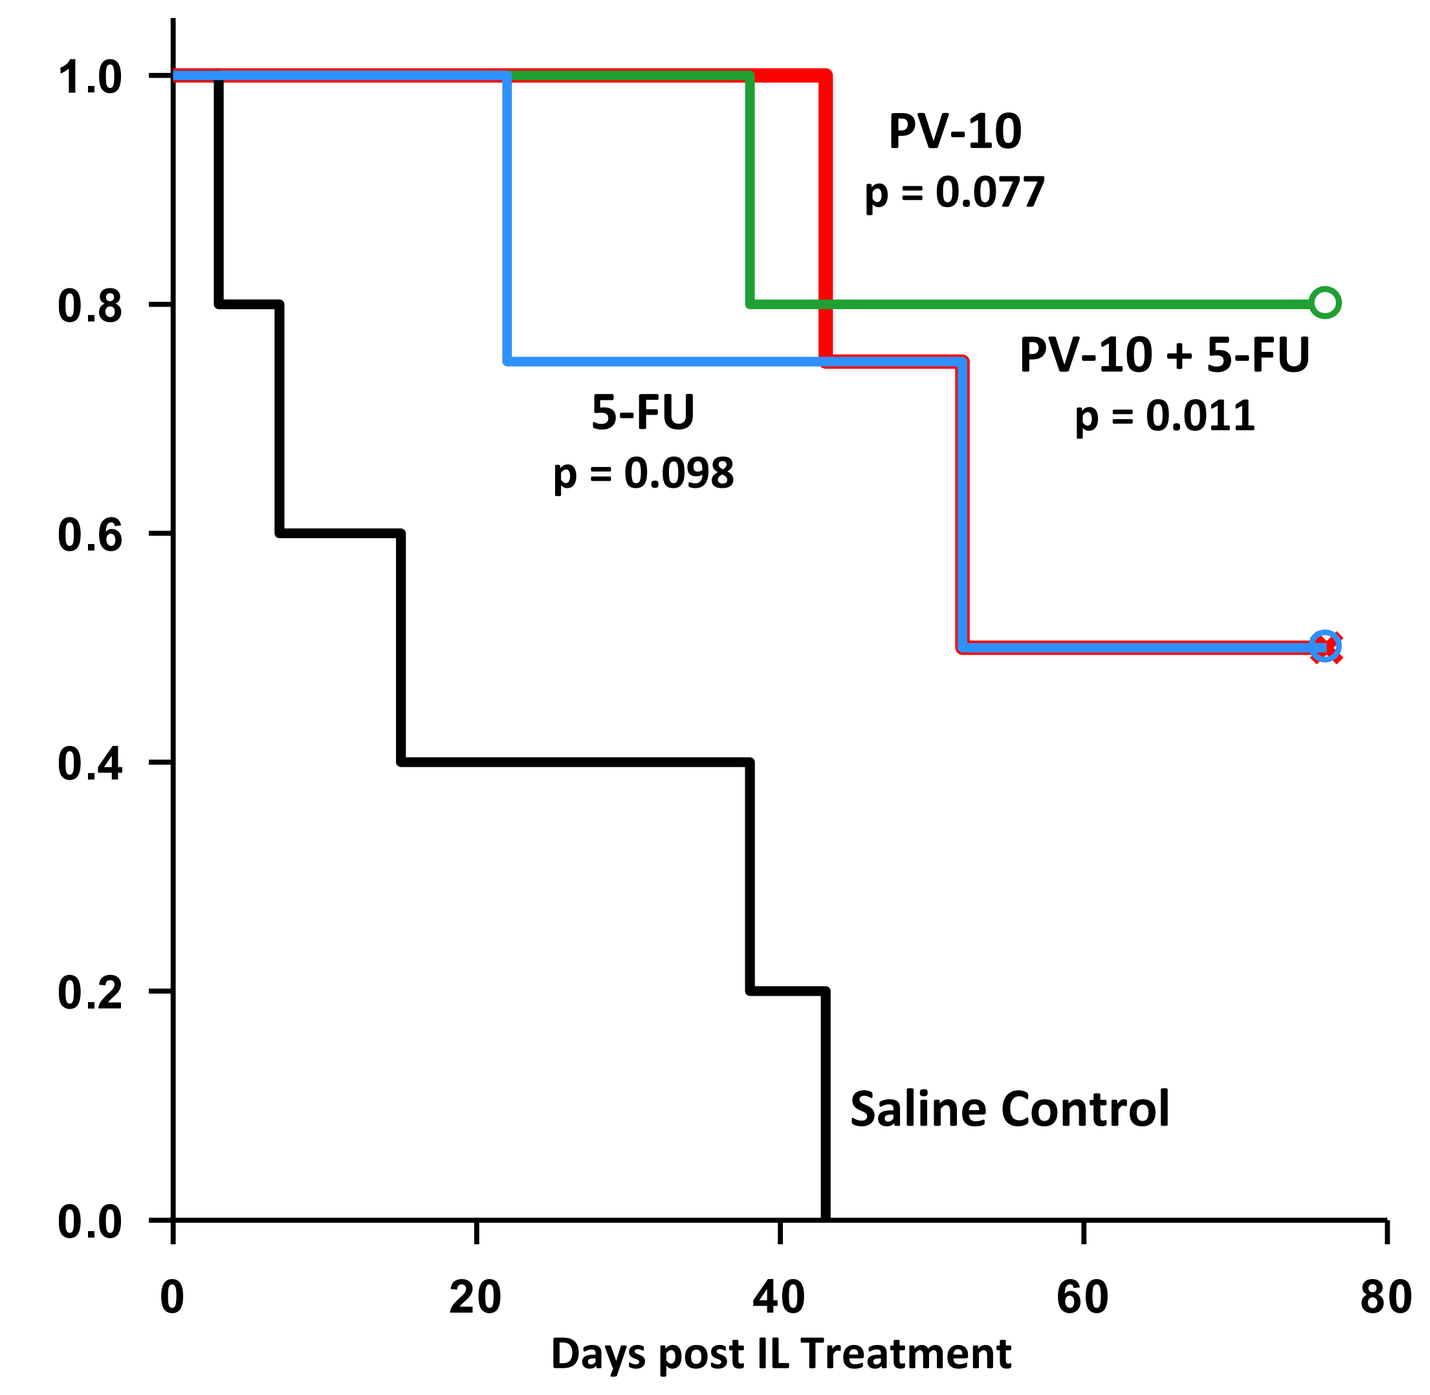
PV-10
p = 0.077
PV-10 + 5-FU
p = 0.011
5-FU
p = 0.098
Saline Control
Days post IL Treatment

Time to Progression: Total Tumor Burden

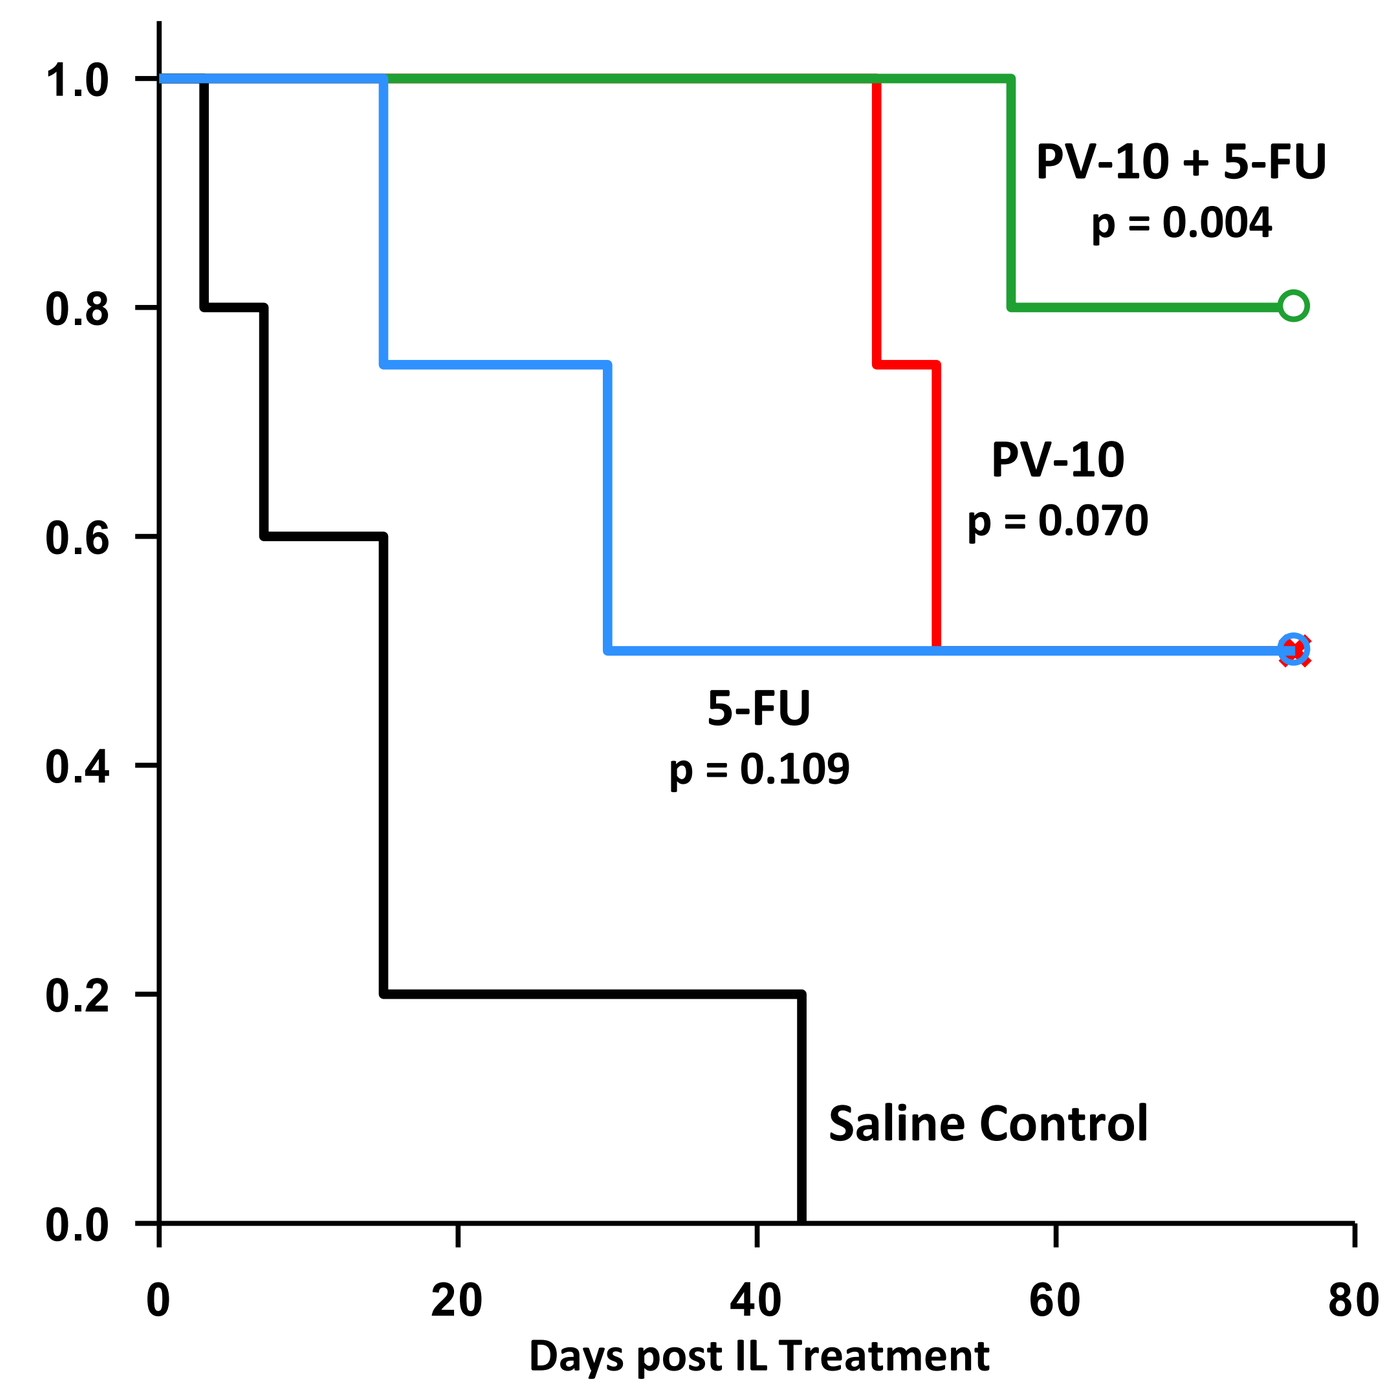
PV-10 + 5-FU
p = 0.004
PV-10
p = 0.070
5-FU
p = 0.109
Saline Control
Days post IL Treatment

Tumor Growth: Injected (Target) Tumors

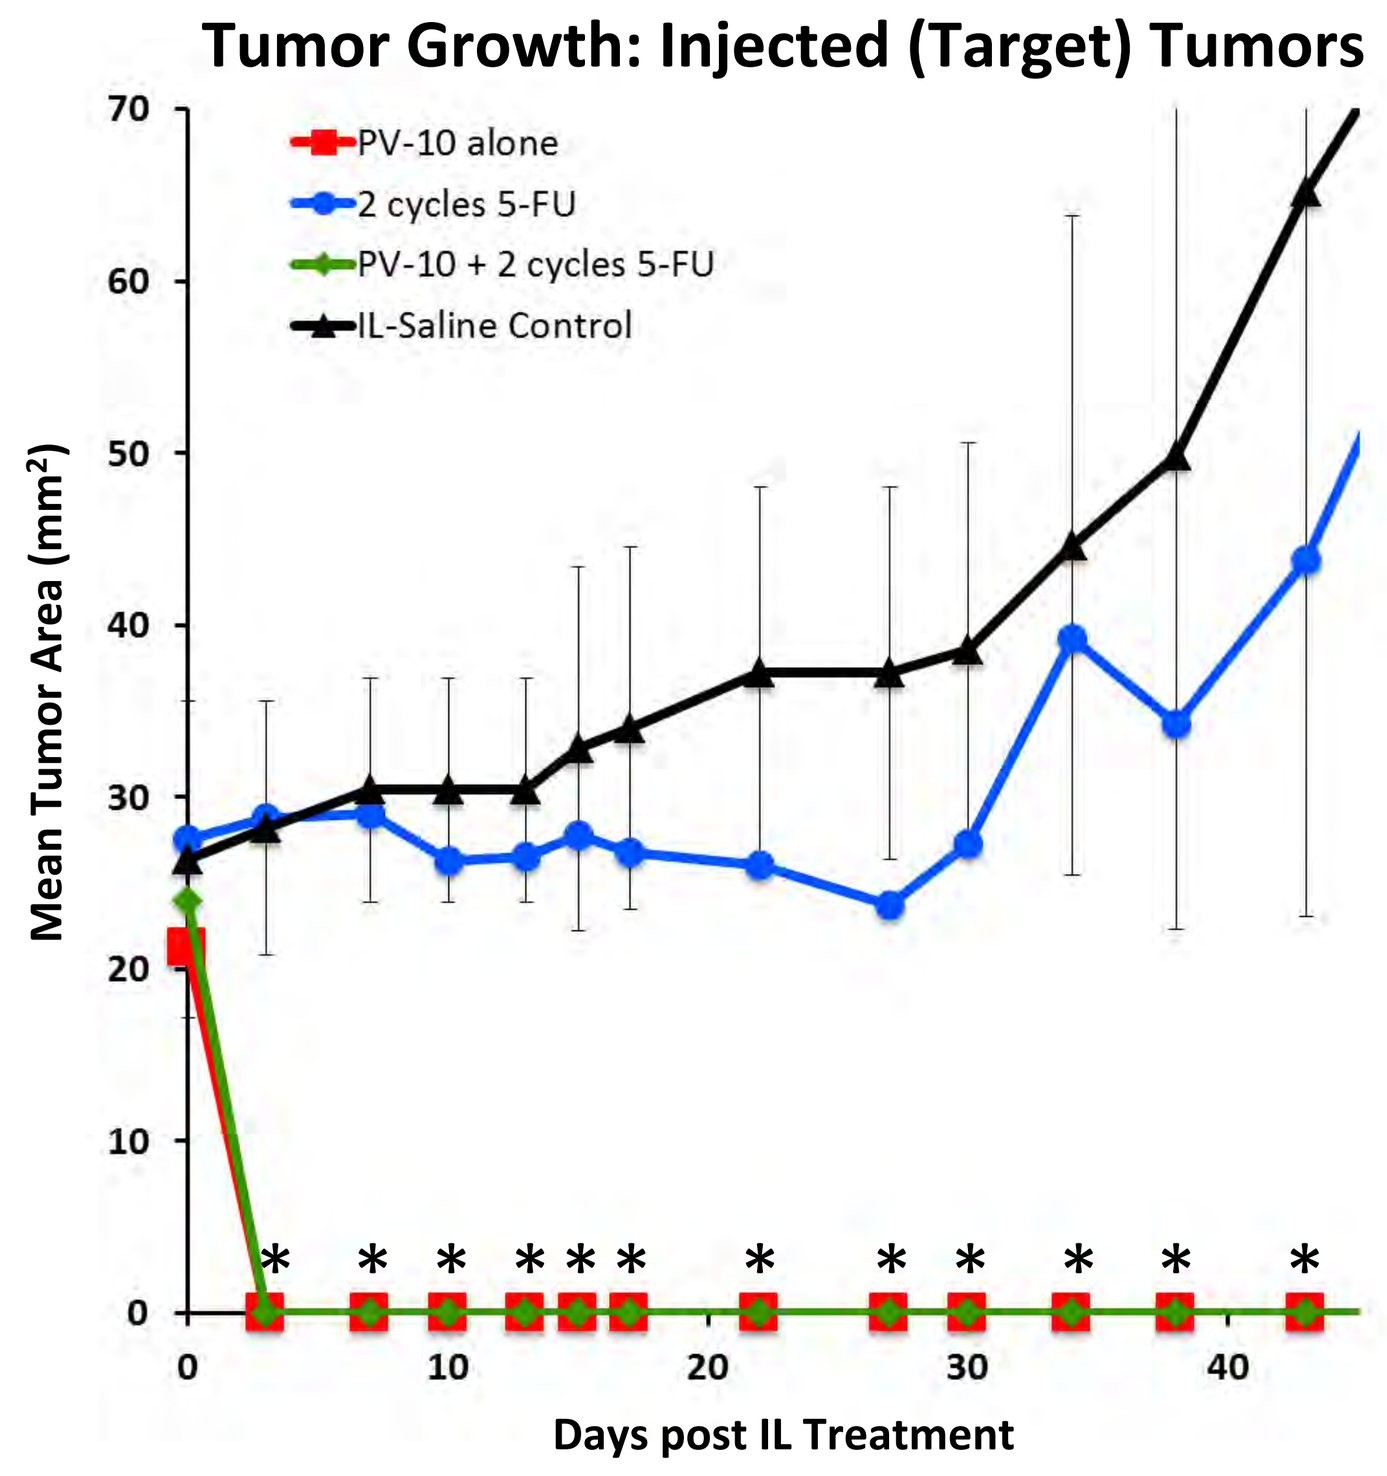
PV-10 alone
2 cycles 5-FU
PV-10 + 2 cycles 5-FU
IL-Saline Control
Mean Tumor Area (mm²)
Days post IL Treatment

*P-values significant for PV-10 group and PV-10 + 2 cycles 5-FU vs. Saline control at Days 3-43 (p < 0.039); standard deviations shown for IL-Saline group.

Tumor Growth: Uninjected Tumors

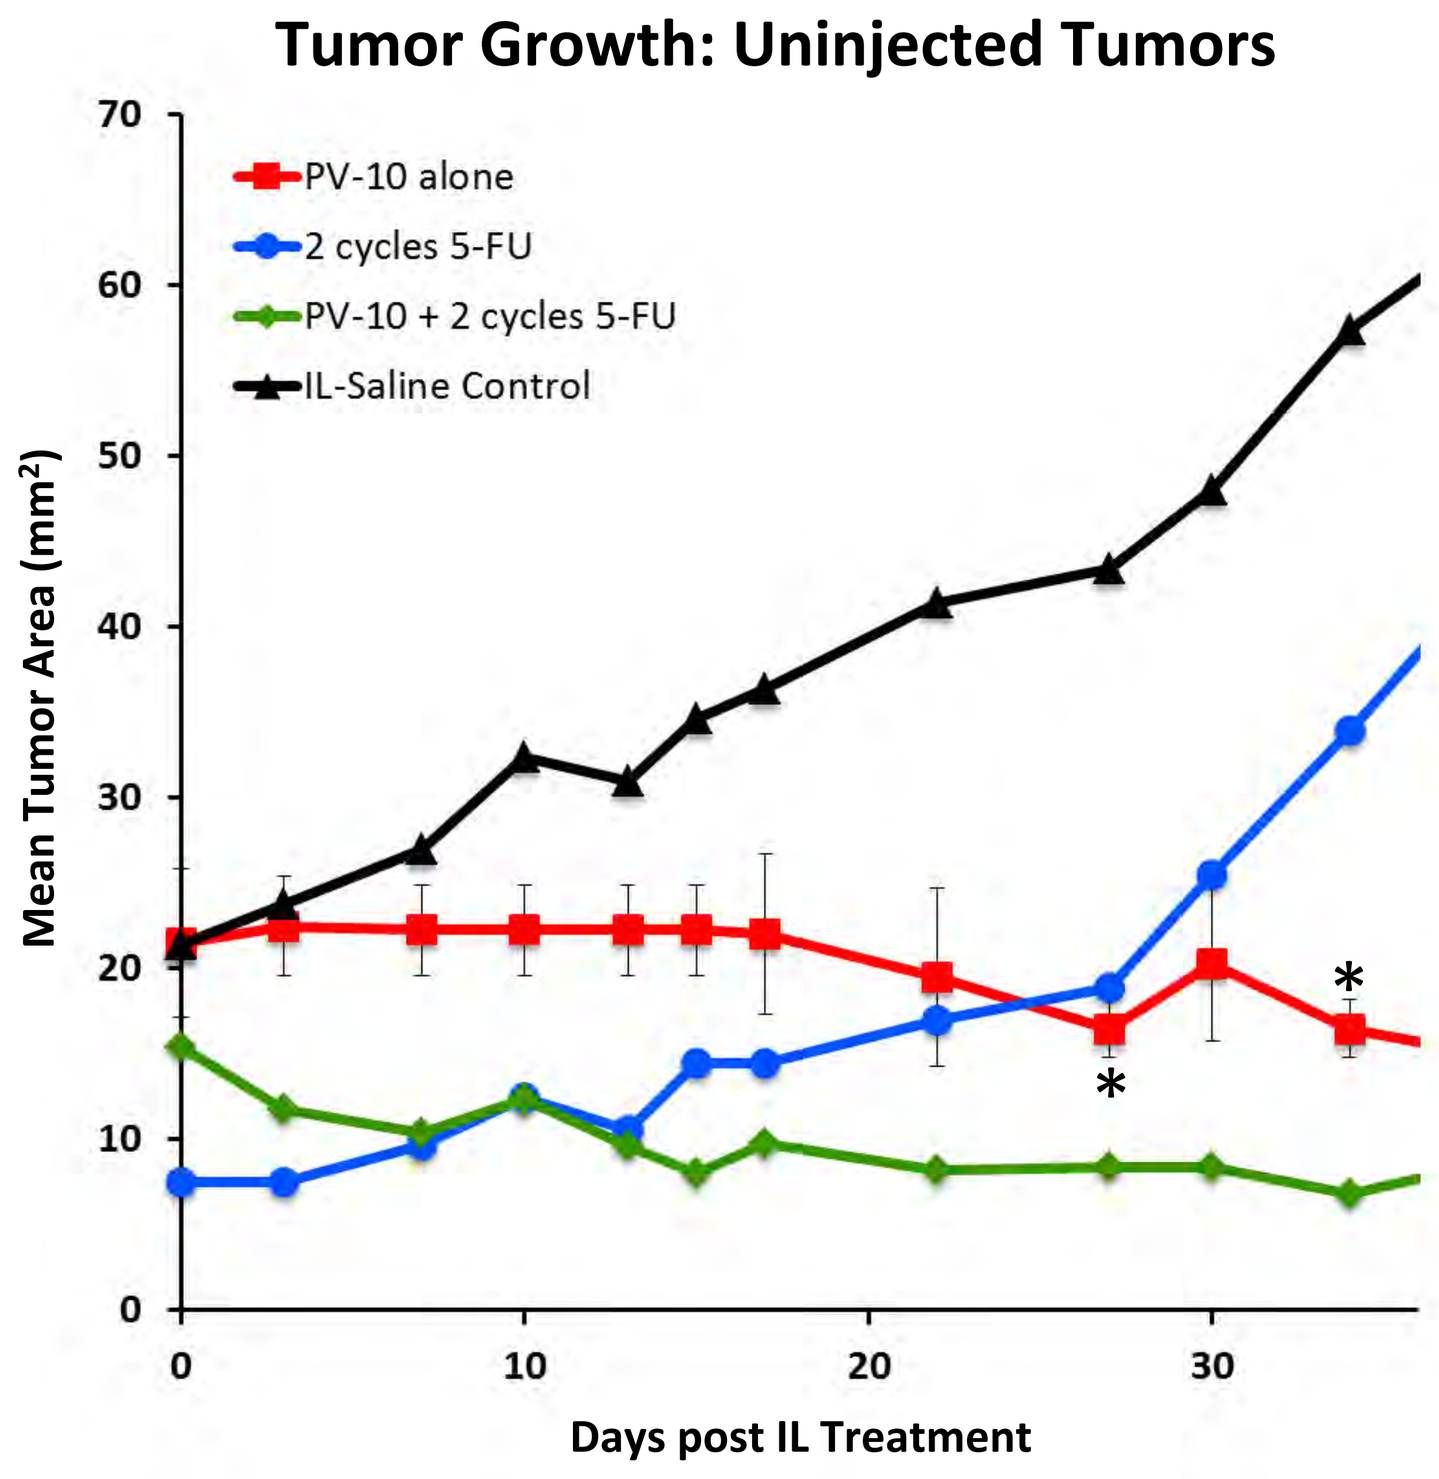
PV-10 alone
2 cycles 5-FU
PV-10 + 2 cycles 5-FU
IL-Saline Control
Mean Tumor Area (mm²)
Days post IL Treatment

*P-values significant for PV-10 group vs. Saline control at Days 27 (p = 0.044) and 34 (p = 0.029); standard deviations shown for PV-10 treatment group.

Conclusions:

- PV-10 yields rapid, durable reduction in tumor burden due to immediate autolysis of injected tumors
- Regression of uninjected tumors is consistent with an immunologic response to the ablated tumor
- Combination of PV-10 with systemic chemotherapy can yield synergistic benefit in untreated tumors supportive of ongoing clinical work [2]

Melanoma (Lung Metastasis Model)

Murine Model: 6 C57BL/6 females with B16-F10 murine melanoma

Tumor Induction: Solitary SQ tumors were induced in the flank by injection of tumor cells (2 x 10^5 cells per tumor). Lung metastases were produced by synchronous injection of tumor cells into the tail vein (2.5 x 10^5 cells)

Treatment: 9 days after tumor implantation, a single IL injection of 50-100 µL of PV-10 was administered into the flank tumor (N = 3 mice). Controls were untreated (N = 3 mice).

Follow up: 21 days after tumor implantation, lungs were harvested and metastases were counted on the anterior and posterior sides using a 1 mm^2 grid. Each mm^2 of tumor was scored as 1 lung metastasis.

Intralesional PV-10

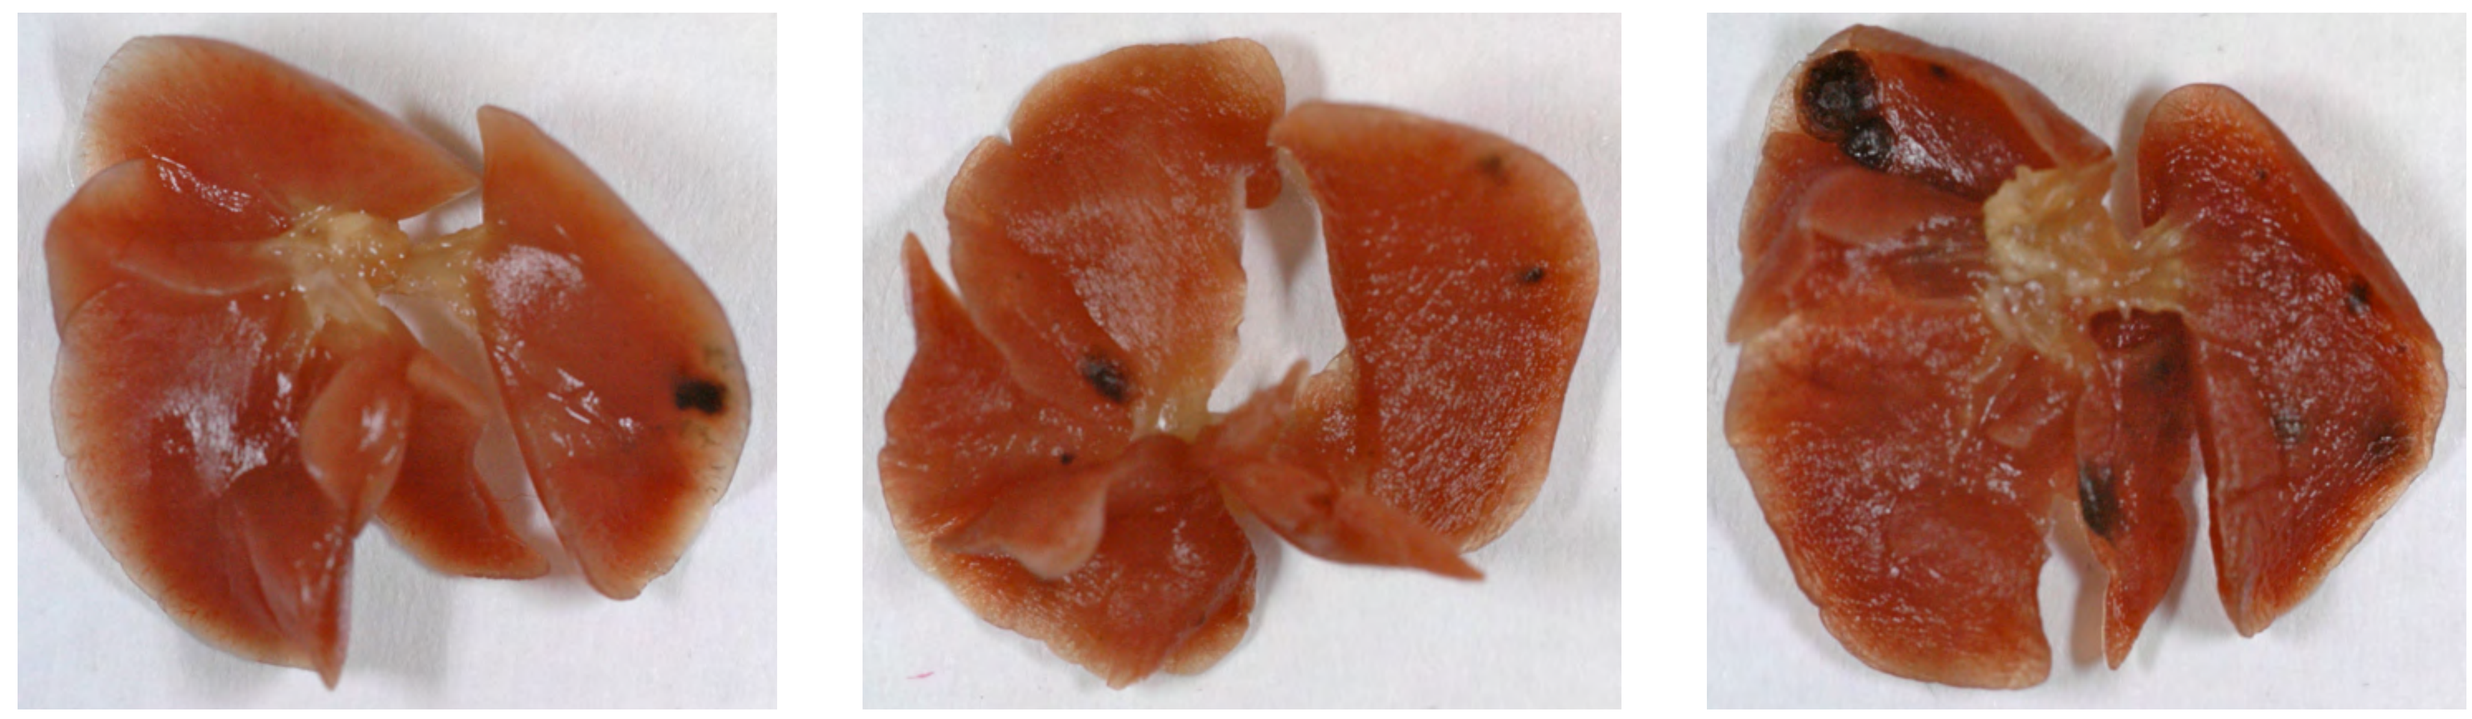

1 Lung Met — 3 Lung Mets — 7 Lung Mets

Control

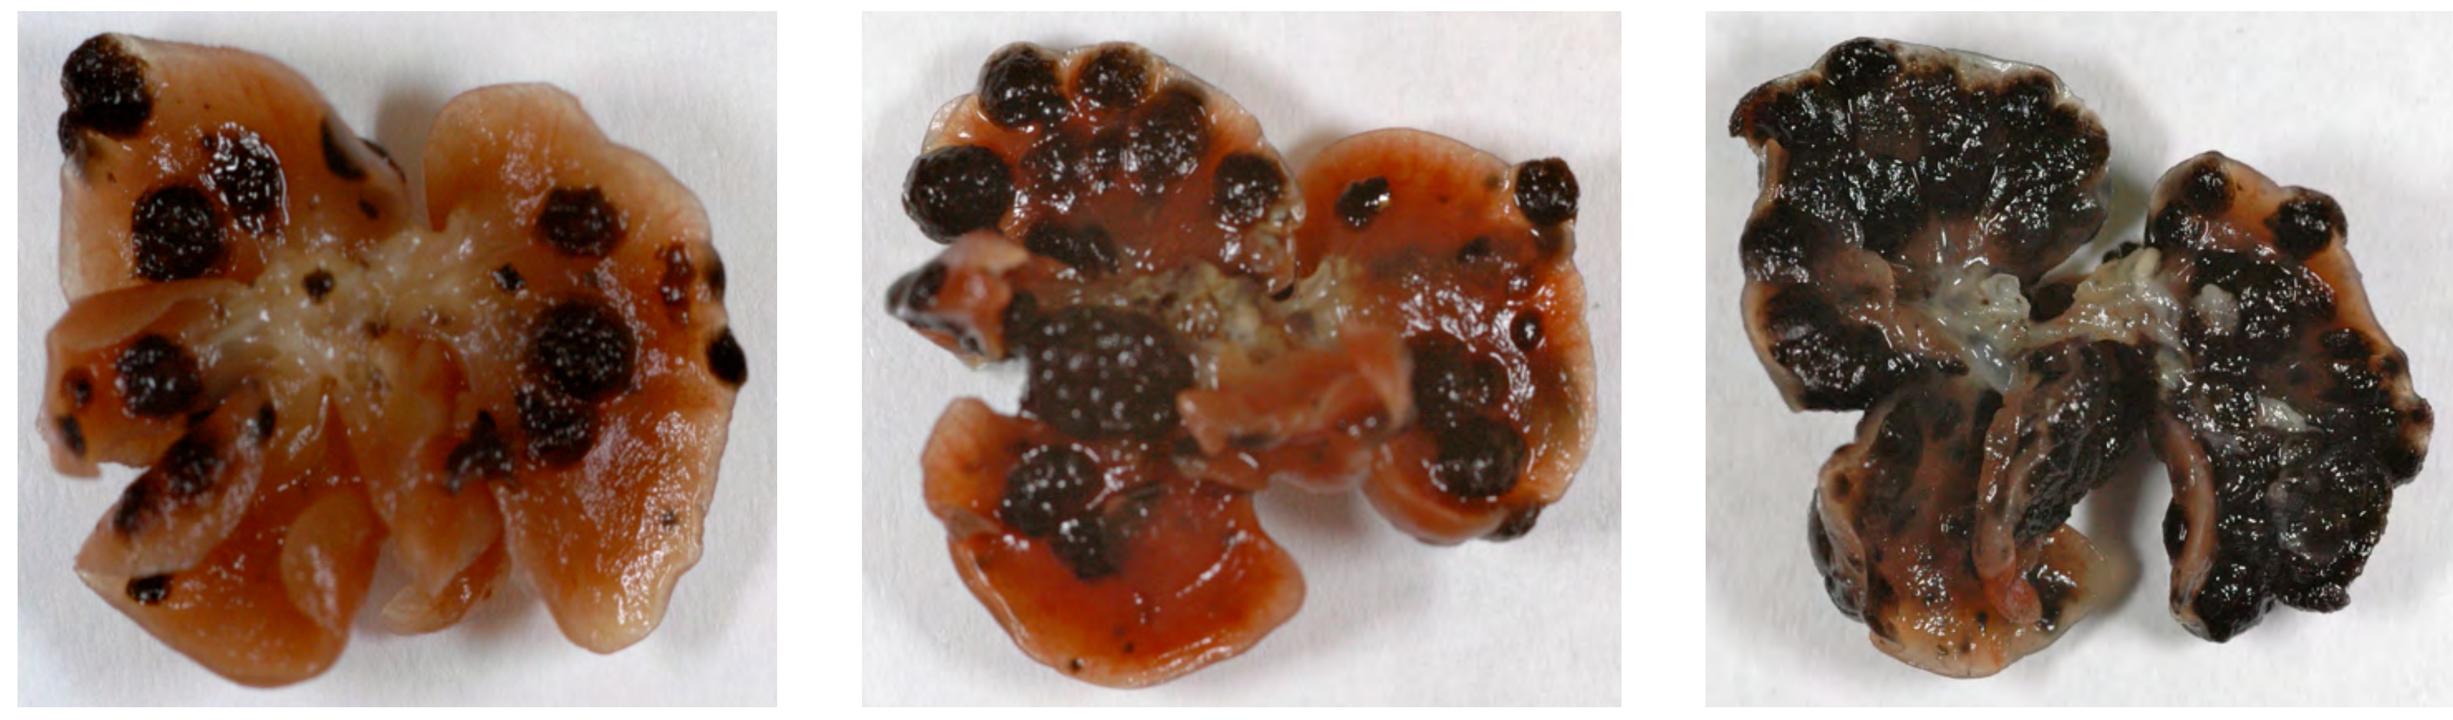

42 Lung Mets — > 100 Lung Mets — > 100 Lung Mets

Results: Mice that received IL PV-10 exhibited a dramatic reduction in number of synchronous lung metastases. Mice treated with PV-10 had a mean of 4 lung metastases (range 1 to 7). Control mice had a mean >81 (range 42 to > 100 lung metastases).

Conclusions:

- This model demonstrates the biological relevance of PV-10 immuno-chemoablation

Re-challenge and Adoptive Transfer Models

Murine Model: C57BL/6 and BALB/c (nu/nu) females (Hepa 1-6 and B16-F10)

Primary Tumor Induction: Solitary SQ tumors were induced in the flank by injection of Hepa 1-6 tumor cells (1 – 2 x 10^5 cells per tumor).

Treatment: A single IL injection of 50-100 µL of PV-10 was administered on Study Day 0.

Secondary Tumor Induction: Mice were re-challenged with either Hepa 1-6 or B16-F10 tumor cells in the flank opposite to the previous PV-10 ablation (SQ injection of 1–2 x 10^5 cells per tumor). Tumor burden was evaluated up to 17 days after re-challenge.

Treatment	Initial Tumor (Hepa 1-6)	Tumor Re-challenge (Day)	Animals with Measureable Tumor on Re-challenge / Animals Re-challenged	
			Hepa 1-6 Re-challenge	B16-F10 Re-challenge
Control	Single flank	21	15 / 15	*
PV-10	Single flank	21	0 / 6	6 / 6
PV-10	Single flank	50	*	7 / 7
PV-10[1]	Bilateral flank	81	0 / 8	*

[1] PV-10 injected in single flank, contralateral tumor regressed prior to re-challenge
*Not done

Data Not Shown: Immunodeficient mice manifested all tumor types at re-challenge after PV-10 ablation.

Adoptive Transfer: Spleen cells harvested from immunocompetent mice 21 days after ablation of SQ tumors (Hepa 1-6) were administered via IP injection (1 x 10^6 primed cells) to 5 naïve immunocompetent mice. Control cells from mice bearing HCC tumors that had not been ablated were similarly transferred to 5 additional mice. Following transfer of spleen cells, recipient mice were challenged with 1 x 10^6 Hepa 1-6 cells and observed for 14 days.

Results: Mice receiving primed spleen cells failed to develop tumors on challenge (0 / 5 recipient mice developed SQ tumors at the challenge site). Mice receiving spleen cells from untreated donors exhibited no evidence of immunity (4 / 5 developed tumors on challenge).

Conclusions:

- Chemoablation with PV-10 results in the induction of tumor-specific immunity
- Immunodeficient mice exhibited no anti-tumor effect on re-challenge
- Adoptive transfer of immunity only occurred following chemoablation in donor mice
- Tumor-specific, long-term immunity and adoptive transfer implicate immune cell mediation
- This immuno-chemoablative response to PV-10 is tantamount to "in situ vaccination"

References

1. Agarwala, S.S. et al. Immuno-chemoablation of metastatic melanoma with intralesional rose bengal. ESMO 2012 Congress, Vienna.
2. A study to assess PV-10 chemoablation of cancer of the liver. Clinical trial NCT00986661.
3. Wachter et al. Functional imaging of photosensitizers using multiphoton microscopy. SPIE 2002; 4620: 143-7.
4. Yoo, B.K. et al. Identification of genes conferring resistance to 5-fluorouracil. Proc Natl Acad Sci USA, 2009; 106 (31): 12938-43.