Interim Results of a Phase 1b/2 Study of PV-10 and PD-1 Blockade in Advanced Melanoma

S Agarwala [1], M Ross [2], J Zager [3], K Shirai [4], R Essner [5], BM Smithers [6], V Atkinson [6], D Sarson [7] and E Wachter [8]

[1] St Luke's University Hospital and Health Network, Easton, PA USA; [2] MD Anderson Cancer Center, Houston, TX USA; [3] Moffitt Cancer Center, Tampa, FL USA; [4] Dartmouth-Hitchcock Medical Center, Lebanon, NH USA; [5] Cedars-Sinai Medical Center, Los Angeles, CA USA; [6] Princess Alexandra Hospital, Brisbane, QLD AUS; [7] Provectus Biopharmaceuticals Australia Pty Ltd, Sydney, NSW AUS and [8] Provectus Biopharmaceuticals, Inc., Knoxville, TN USA

2018 SMR Congress – 15th International Congress of the Society for Melanoma Research
Manchester, England – 24-27 October 2018

For additional information:
info@pvct.com

Updated Abstract and Background

PV-10 (rose bengal disodium) is a small molecule oncolytic immunotherapy in development for solid tumors, where intralesional injection can yield immunogenic cell death and tumor-specific reactivity in circulating T cells [1-4]. It has been administered as a single agent to 130 cutaneous melanoma patients in Phase 1 and 2 and 180 patients in expanded access, and is in Phase 3 for locally advanced cutaneous melanoma (Stage IIIB–IV M1a, **NCT02288897**) [5-8].

Study PV-10-MM-1201 (**NCT02557321**) is a Phase 1b/2 study of PV-10 in combination with anti-PD-1 (pembrolizumab) for patients with advanced melanoma (Stage IIIC-IV M1c). Patients must have at least 1 injectable lesion and be candidates for pembrolizumab. In Phase 1b patients receive combination treatment q3w for 5 cycles then pembrolizumab alone for up to 24 months; the primary endpoint is safety and tolerability with objective response rate (ORR) and progression free survival as key secondary endpoints (assessed via RECIST 1.1 after 5 cycles then q12w).

Full accrual for Phase 1b was reached in April 2018, with an intent-to-treat (ITT) population of 20 Stage IV and 3 Stage IIIC/IIID patients (median age 70 years, range 28-90) receiving at least 1 dose of PV-10 and pembrolizumab. All Treatment-Emergent Adverse Events (TEAEs) were consistent with established patterns for both drugs, with no significant overlap of AEs or unexpected toxicities; Grade 1-2 AEs attributed to the combination were observed in 6 patients. Most patients had extensive uninjected tumor burden, and complete response (CR) was observed in non-injected visceral disease, including lung and liver metastases. Interim efficacy data were positive, with 65% ORR and 9% CR by RECIST.

Acceptable safety and tolerability of the combination were observed with no unexpected safety issues. Response follow-up is ongoing for a substantial fraction of the ITT population. An Expansion Cohort is being opened to assess patients with prior checkpoint inhibition.

1. Wachter et al., Proceedings of SPIE 2002; 4620: 143. **2.** Liu et al., Oncotarget 2016; 7: 37893. **3.** Qin et al., Cell Death and Disease 2017; 8: e2584. **4.** Liu et al., PLoS ONE 2018; 13: e0196033. **5.** Thompson et al., Melanoma Research 2008; 18: 405. **6.** Thompson et al., Annals Surg Oncol 2015; 22: 2135. **7.** Lippey et al., J Surg Oncol 2016; 114: 380. **8.** Foote et al., J Surg Oncol 2017; 115: 891.

Small Molecule Oncolytic Immunotherapy

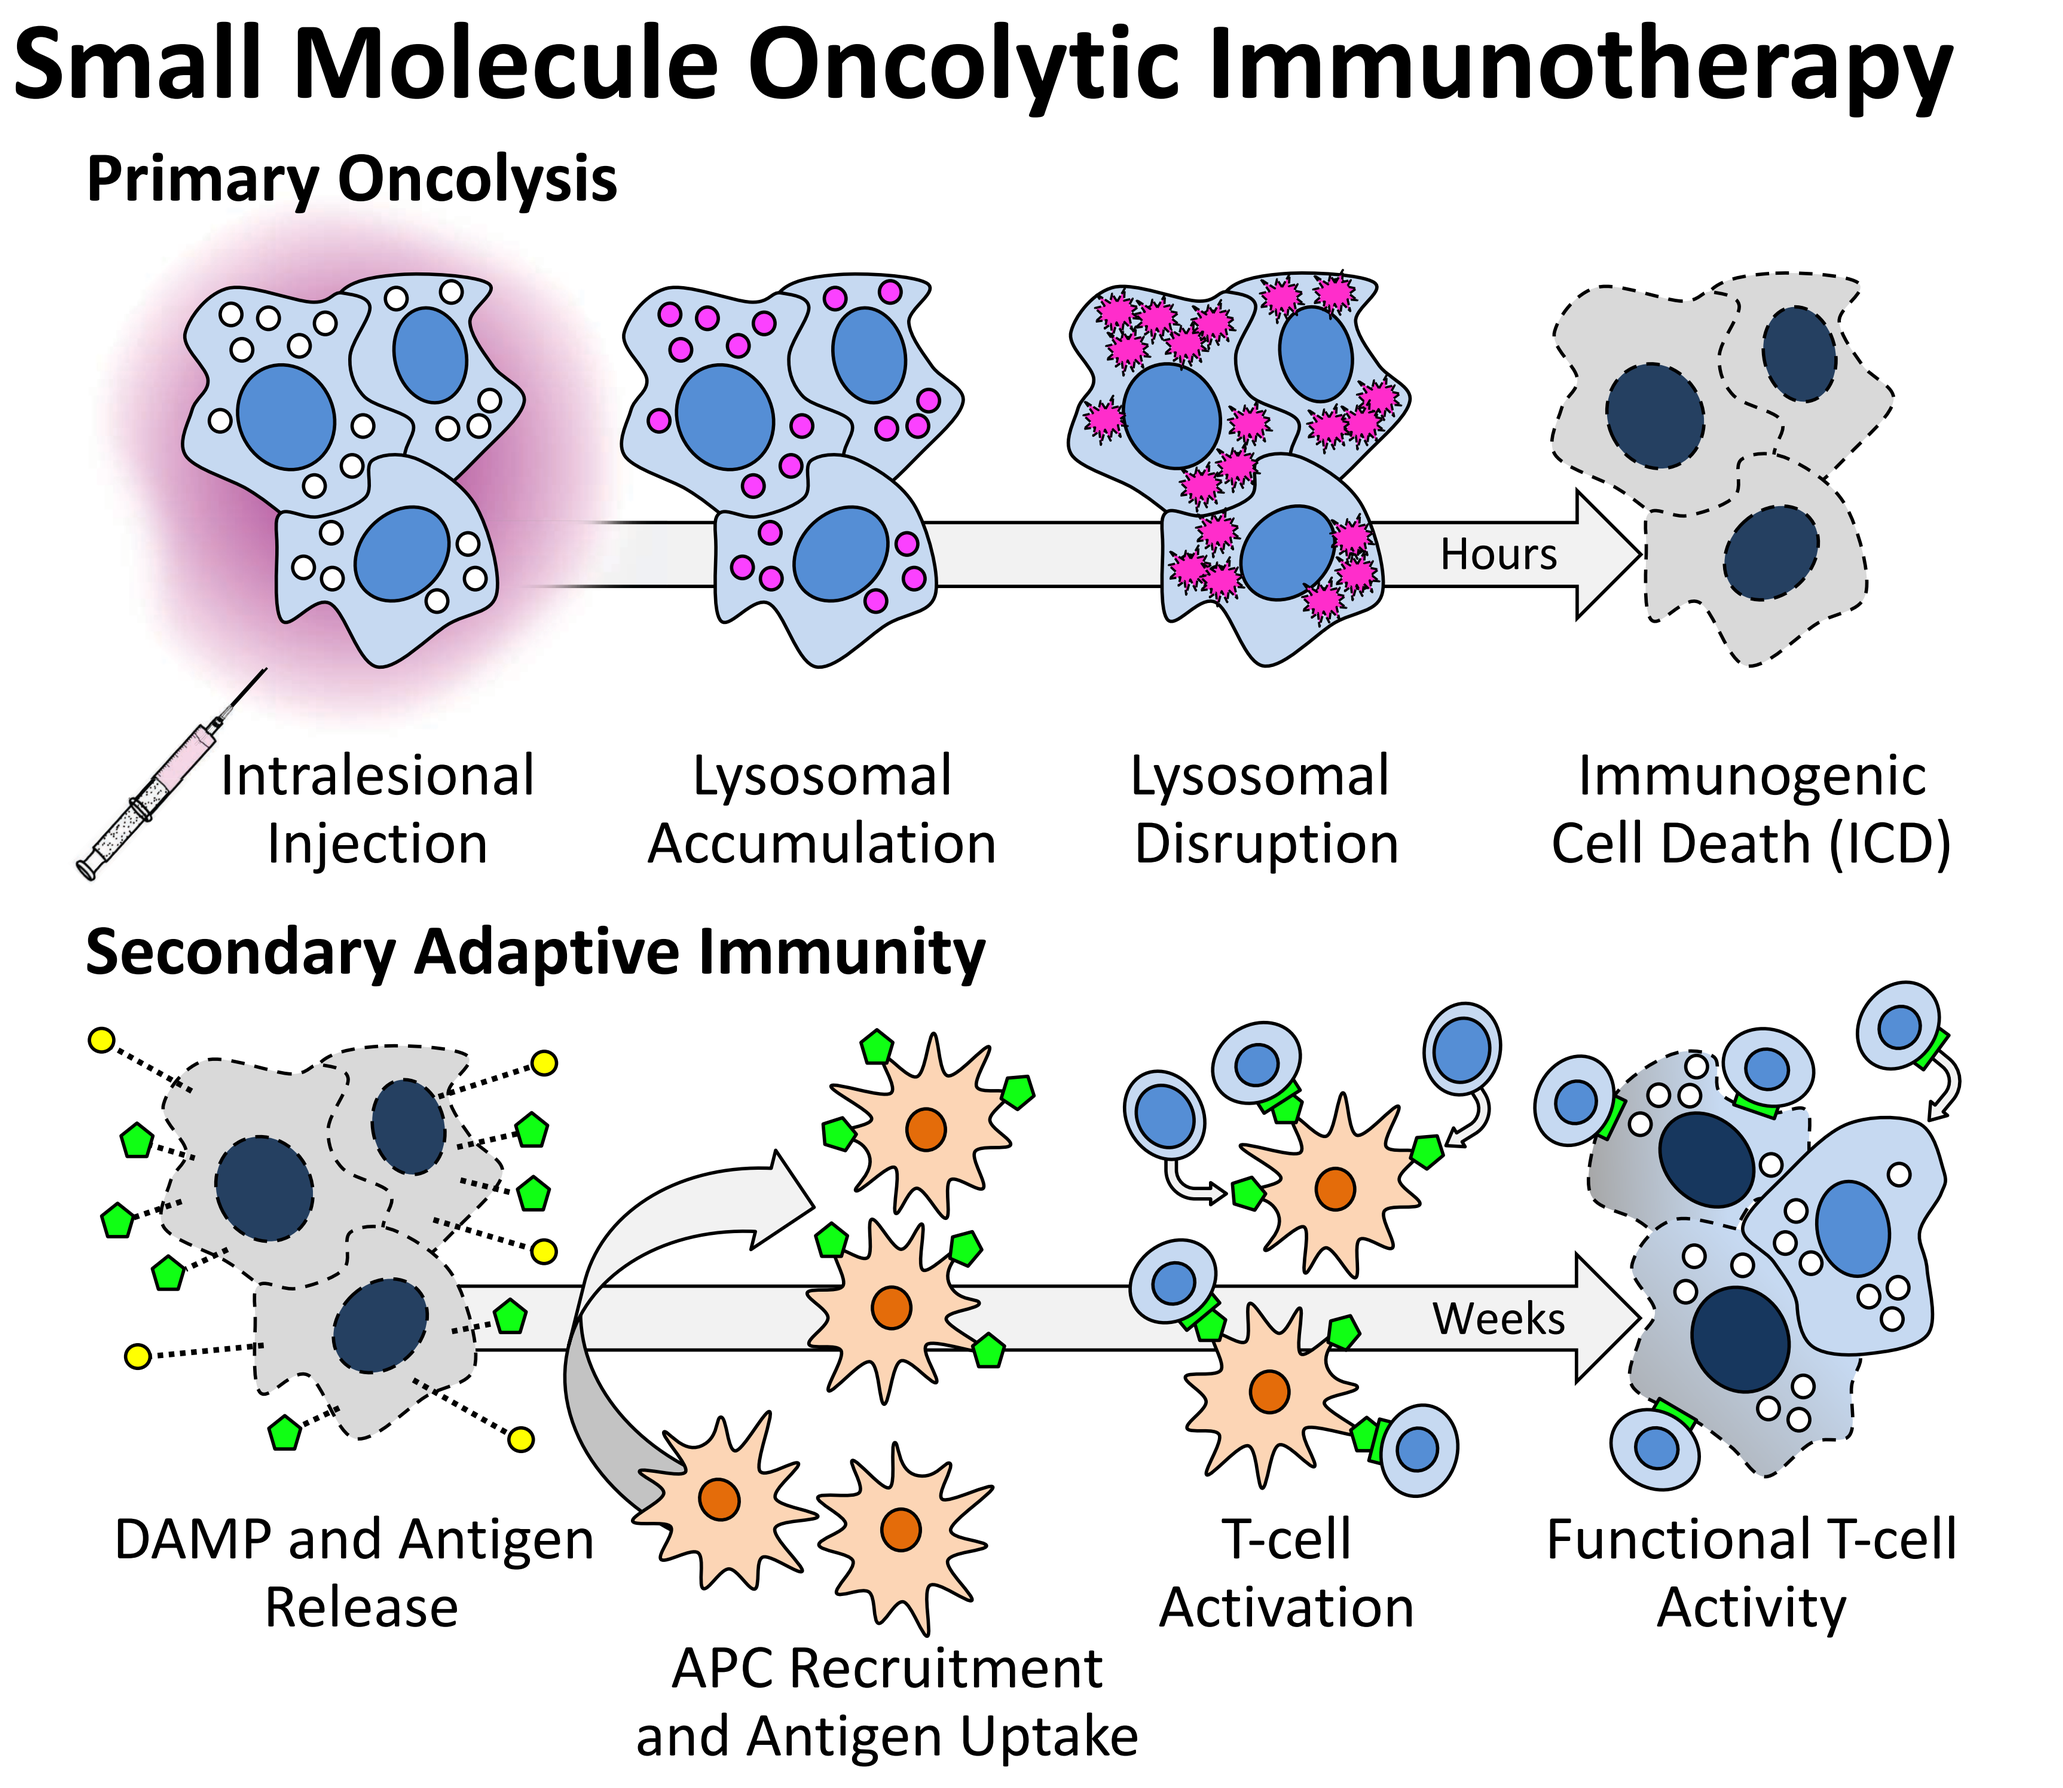

Primary Oncolysis
Hours
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Immunogenic Cell Death (ICD)
Secondary Adaptive Immunity
Weeks
DAMP and Antigen Release
APC Recruitment and Antigen Uptake
T-cell Activation
Functional T-cell Activity

- Functional T cell Activity in Peripheral Blood of Melanoma Patients [2]
- Immunologic Priming of PV-10 Complementary to Checkpoint Inhibition in Murine Melanoma Models [4]

Study Overview and Interim Data

Phase 1b Study Participants

Study Population (Phase 1b Main Cohort ITT Population, N = 23)

Gender	Male Female	19 (83%) 4 (17%)
Age	Median (Range)	70.0 (28 – 90)
Race	Black or African American White	1 (4%) 22 (96%)
AJCC Disease Stage	IIIC/IIID IV M1a IV M1b IV M1c	3 (13%) 8 (35%) 6 (26%) 6 (26%)
Number of Cutaneous / Subcutaneous Lesions	Median (Range)	2.0 (1 – 15) [a]
Prior Therapy	Checkpoint Inhibition Other Immunotherapy Targeted Therapy Chemotherapy Radiotherapy Surgery	2 (9%) [b] 3 (13%) [c] 1 (4%) [d] 1 (4%) [e] 2 (9%) 23 (100%)
BRAF Mutation Status	Mutant Wild-Type Not Reported	7 (30%) 9 (39%) 7 (30%)

[a] Subjects 0204 and 0403 with baseline disease burden too numerous to count (TNC) are excluded from calculation.
[b] Subjects 0110 and 0402 enrolled under waiver for prior treatment with nivolumab and ipilimumab, respectively.
[c] Subjects 0102 and 0104 previously treated with pegylated interferon alfa-2b; subject 0402 previously treated with adjuvant vaccine.
[d] Subject 0205 previously treated with dabrafenib and trametinib.
[e] Subject 0401 previous treated with doxorubicin and olaratumab.

- ITT population is all subjects receiving at least one dose of PV-10 and pembrolizumab
- All subjects had measurable Target Lesions assigned per RECIST
- PV-10 injection to cutaneous and subcutaneous lesions (not to nodal or visceral lesions)
- Phase 1b Main Cohort eligibility restricted to checkpoint inhibitor-naïve patients
- Predominantly Stage IV subjects with high median age
- Subjects had substantial systemic disease burden in addition to their injectable lesions

Interim Efficacy Results

Subject Summary (Phase 1b Main Cohort ITT Population, N = 23)

Demographics and Dosing: ID / Age / Gender	Stage	CUT/SQ Lesions	Cycles of PV-10	Lesions Injected per Cycle	Target Lesion Response: N : Lesion Location(s)	BORR (% Change)	Non-Target Lesion Response: Lesion Location(s)	BORR	Overall Response: RECIST 1.1
0111 / 68 / F	IIIC	8	1	3.0	2 : Scalp (x2)	CR	Scalp	Non-CR/Non-PD	PR [a]
0110 / 90 / F	IIID	5	4	4.0	2 : Scalp (x2)	CR	Scalp	Non-CR/Non-PD	PR [a, b]
0501 / 82 / M	IIIC	15	3	3.7	2 : LUE (x2)	PD	LNs	Non-CR/Non-PD	PD
0207 / 44 / M	M1a	1	5	1.4	1 : Infraclavicular (SQ)	CR	Subcutaneous	CR	CR [a]
0106 / 78 / M	M1a	1	5	6.0	2 : RLE (x2)	CR	Left Lower Extremity	Non-CR/Non-PD	PR
0108 / 82 / F	M1a	2	2	1.5	2 : LUE (x2)	CR	Left Upper Extremity	Non-CR/Non-PD	PR
0204 / 28 / M	M1a	TNC	5	16.4	2 : Jaw, LUE	CR	Scalp, Face, Neck, Torso, LUE, Axillary LN	Non-CR/Non-PD	PR
0209 / 66 / M	M1a	2	5	2.0	2 : Back, LUE	SD	Spinal Muscle and LN, Back	Non-CR/Non-PD	SD [a]
0109 / 60 / M	M1a	1	5	1.0	3 : Chest (x2), Axillary LN	SD	Axillary LN and Skin	PD	PD
0205 / 50 / F	M1a	3	5	4.8	1 : Chest Wall	PD	Back	Non-CR/Non-PD	PD
0202 / 52 /M	M1a	1	1	1.0	1 : Chest Wall	NE	Chest Wall and Axillary LN	NE	NE [c]
0206 / 73 / M	M1b	1	5	1.0	2 : RUE	CR	Lung	CR	CR [d]
0102 / 47 / M	M1b	4	5	2.6	1 : Scalp	CR	Lung	Non-CR/Non-PD	PR
0104 / 79 / M	M1b	3	1	3.0	1 : LLE	CR	Bilateral Lung	Non-CR/Non-PD	PR
0203 / 76 / M	M1b	2	5	2.0	2 : RLE (x2)	CR	Lower Extremity and Lung	Non-CR/Non-PD	PR
0502 / 47 / M	M1b	2	4	1.0	2 : Head, Lung	SD	Lung and Hilar LN	PD	PD
0107 / 68 / M	M1b	1	5	1.0	1 : Flank (SQ)	PD	Lung	PD	PD
0112 / 80 / M	M1c	1	3	1.0	3 : Scalp, Liver (x2)	PR	Liver	Non-CR/Non-PD	PR [a]
0401 / 70 / M	M1c	1	5	1.0	3 : Shoulder, Lung, Liver	PR	Liver and Lung	Non-CR/Non-PD	PR [a]
0403 / 78 / M	M1c	TNC	3	2.3	4 : Shoulder (x2), Clavicle, Bladder	PR	Bladder and Bone	Non-CR/Non-PD	PR [a]
0101 / 81 / M	M1c	3	4	2.5	3 : SQ (x2), Axillary LN	PR	Liver and Lung	CR	PR
0105 / 69 / M	M1c	4	2	2.5	4 : RUE, R Axilla, Liver (x2)	PR	Bone, Liver and Lung	Non-CR/Non-PD	PR
0402 / 79 / M	M1c	2	2	2.0	5 : Axilla, Flank, Lung, Liver, Peritoneum	NE	Peritoneum, Retroaortic LNs and Lung	Non-CR/Non-PD	NE [e]

Abbreviations: CUT, cutaneous; LLE, left lower extremity; LN, lymph node; LUE, left upper extremity; ITT, intent-to-treat; N, number; NE, not evaluable; R, right; RLU, right lower extremity; RUE, right upper extremity; SQ, subcutaneous; TNC, too numerous to count.
[a] Response follow-up ongoing at data cutoff date.
[b] Subject 0110 previously received nivolumab (12 months ending 2 months prior to enrollment).
[c] Subject 0202 withdrawn after first cycle of PV-10 + pembrolizumab per investigator decision (due to Grade 1 injection site haemorrhage, injection site reaction and injection site discolouration).
[d] Subject 0206 developed Grade 3 pemphigoid at week 48 resulting in permanent discontinuation of pembrolizumab.
[e] Subject 0402 previously received ipilimumab (ending 3 years prior to enrollment); withdrawn after second cycle of PV-10 + pembrolizumab due to onset of myasthenia gravis.

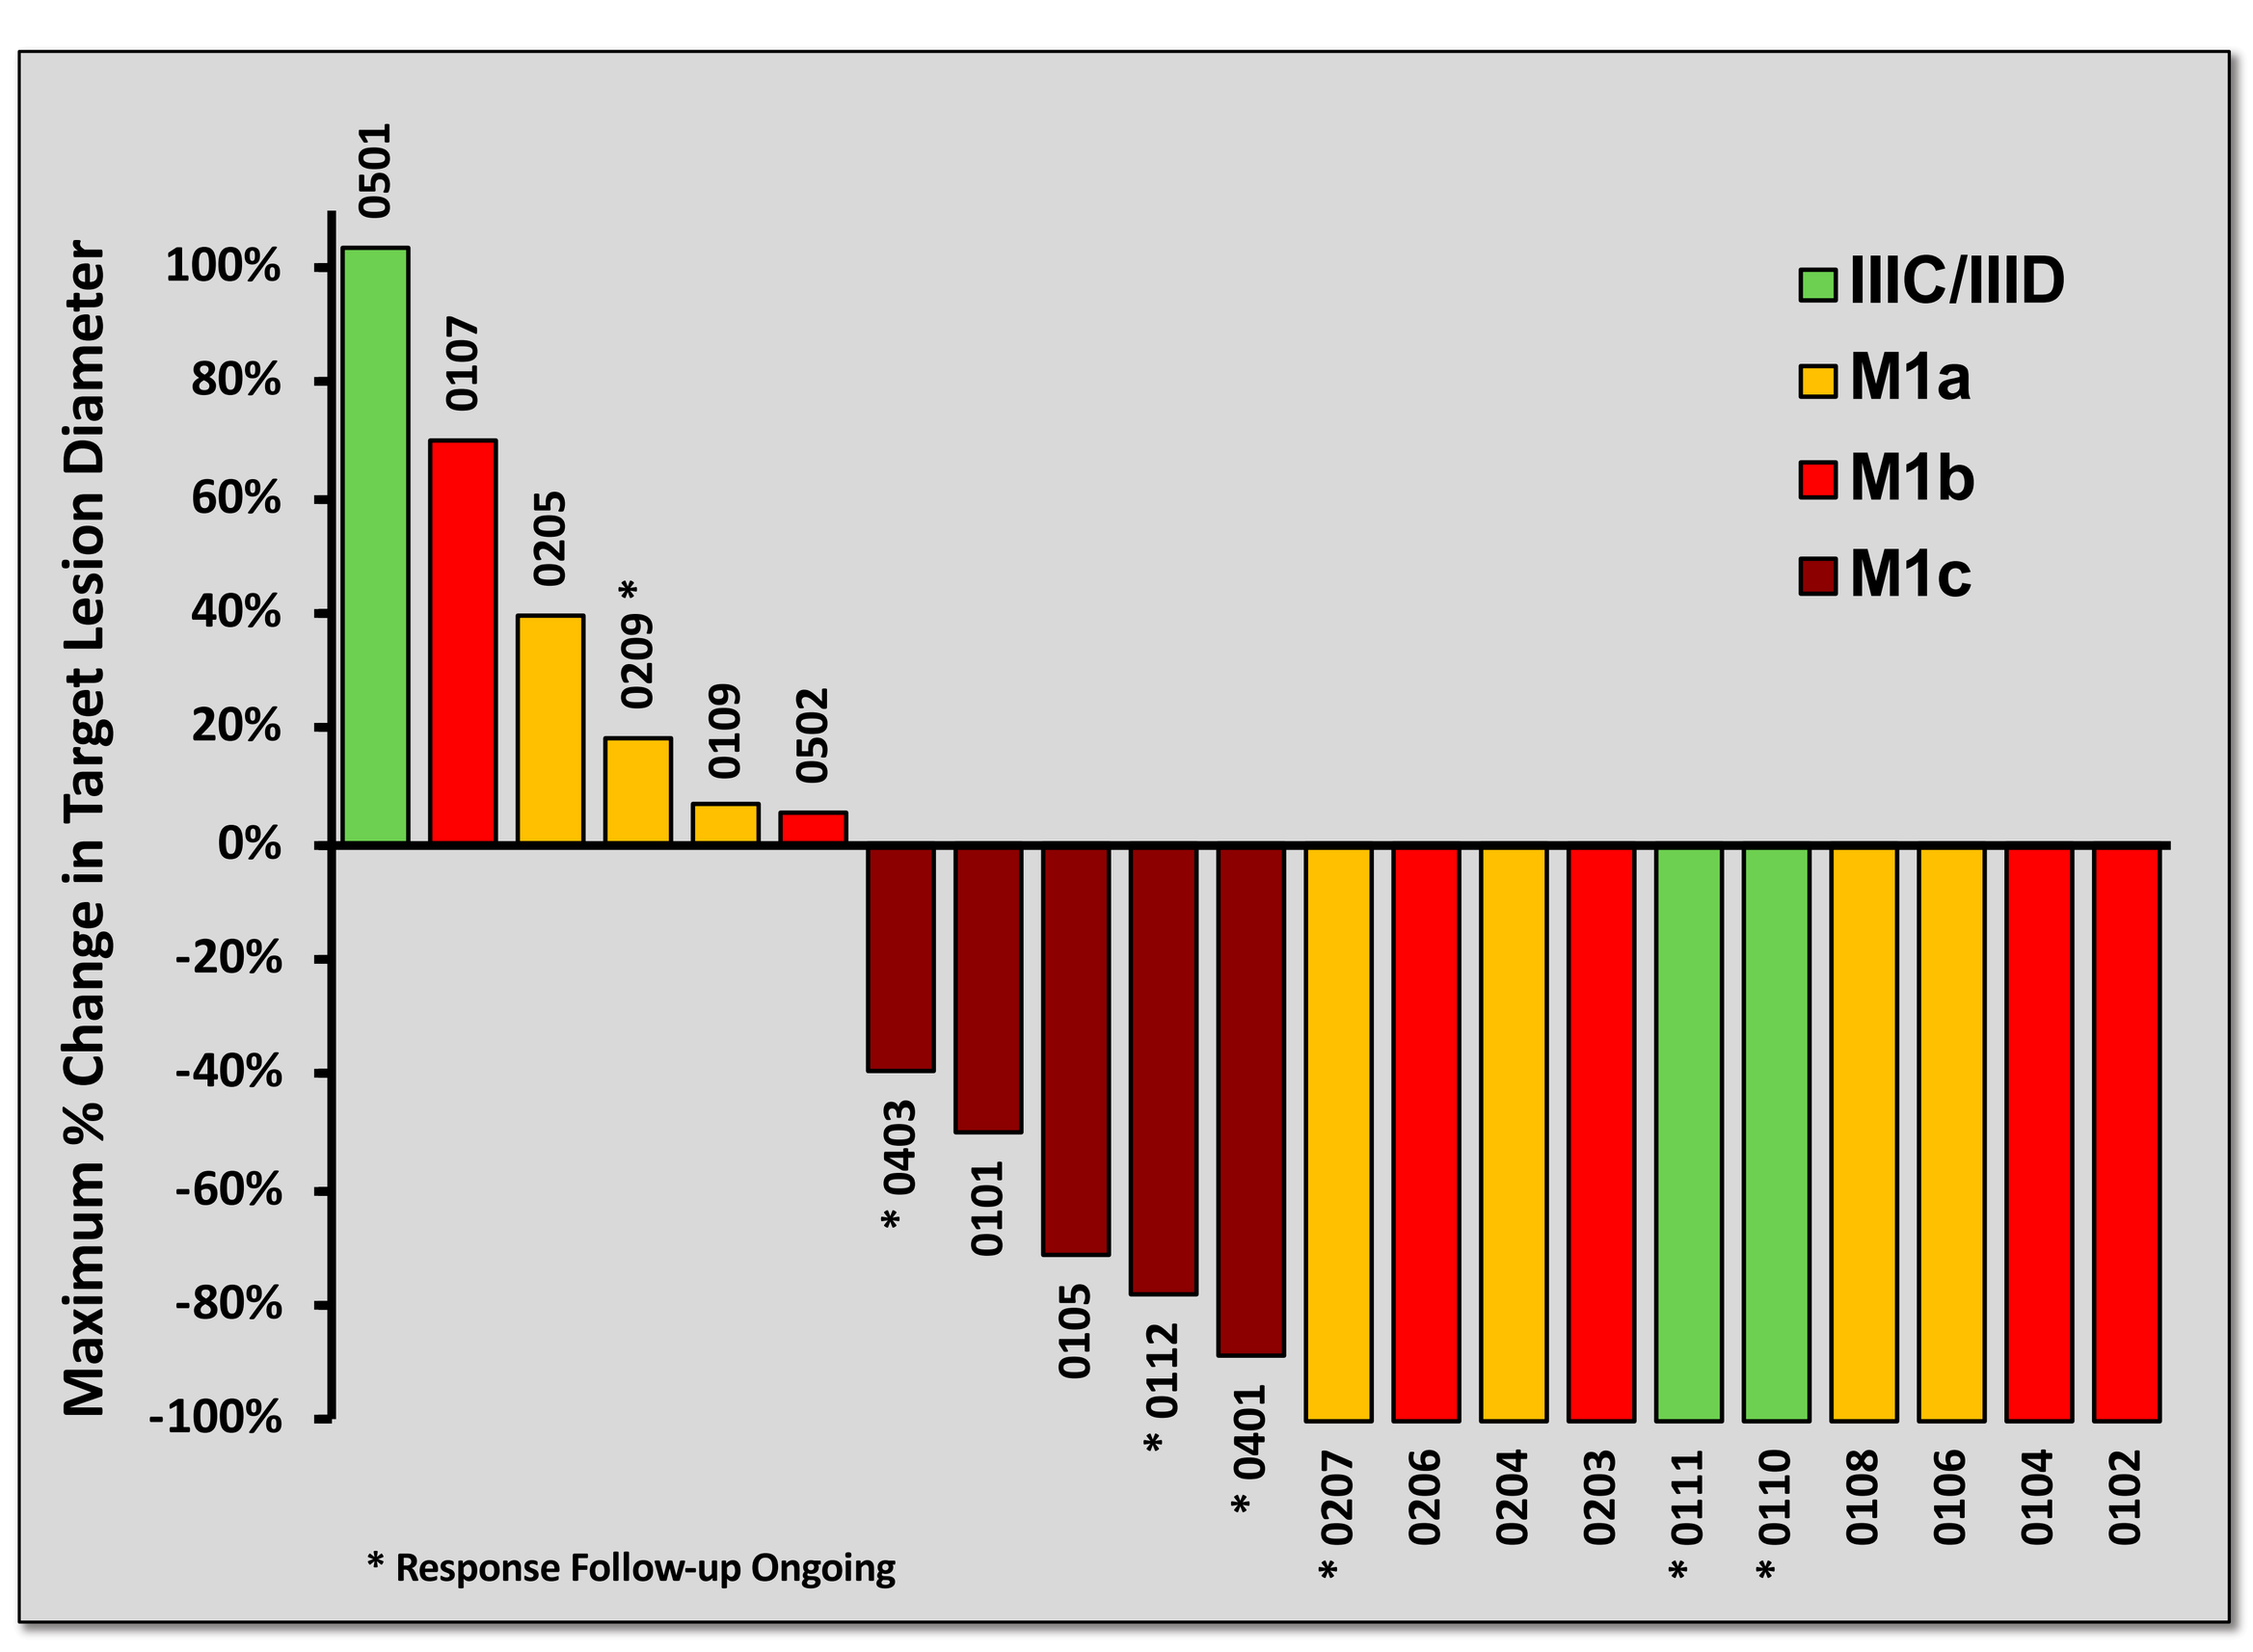

Maximum % Change in Target Lesion Diameter
100%
80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
0501
0107
0205
0209 *
0109
0502
* 0403
0101
0105
* 0112
* 0401
* 0207
0206
0204
0203
* 0111
* 0110
0108
0106
0104
0102
IIIC/IIID
M1a
M1b
M1c
* Response Follow-up Ongoing

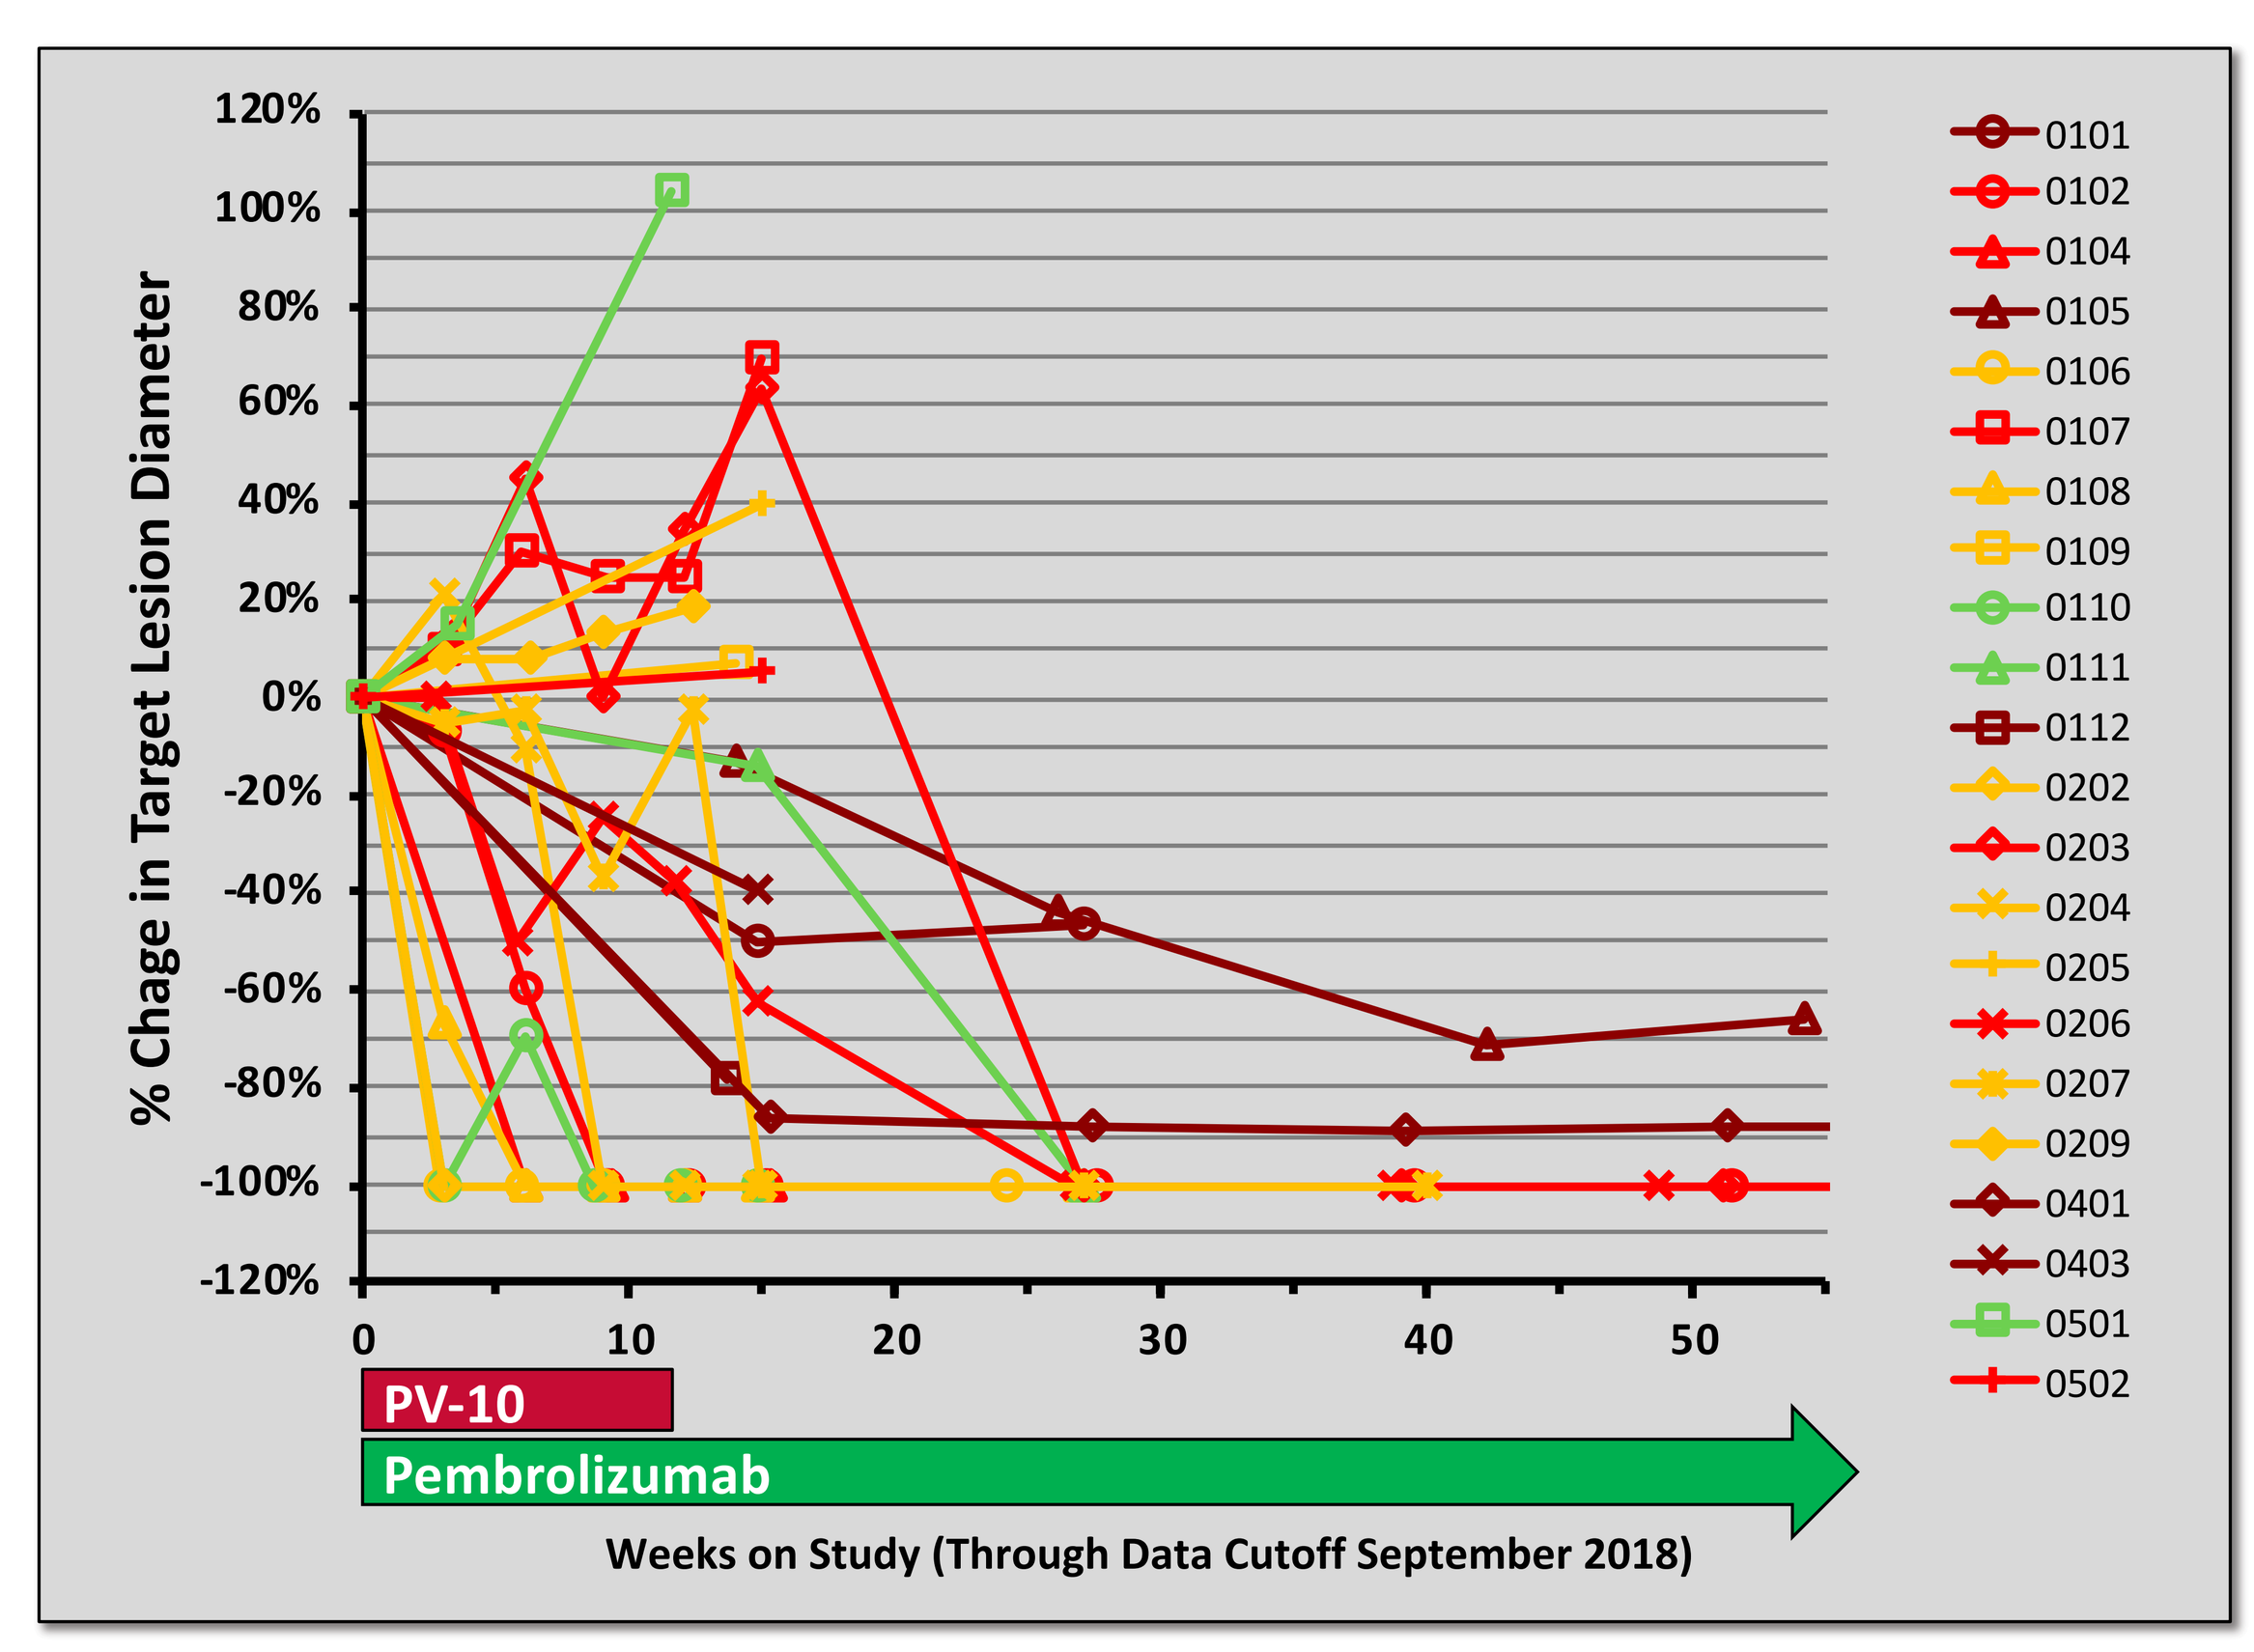

% Chage in Target Lesion Diameter
120%
100%
80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
-120%
0
10
20
30
40
50
PV-10
Pembrolizumab
Weeks on Study (Through Data Cutoff September 2018)
0101
0102
0104
0105
0106
0107
0108
0109
0110
0111
0112
0202
0203
0204
0205
0206
0207
0209
0401
0403
0501
0502

- Subjects received a median of 4.0 cycles of PV-10 (mean 3.7, range 1 – 5)
- Subjects received a median of 5.0 injections of PV-10 (range 1 – 82)

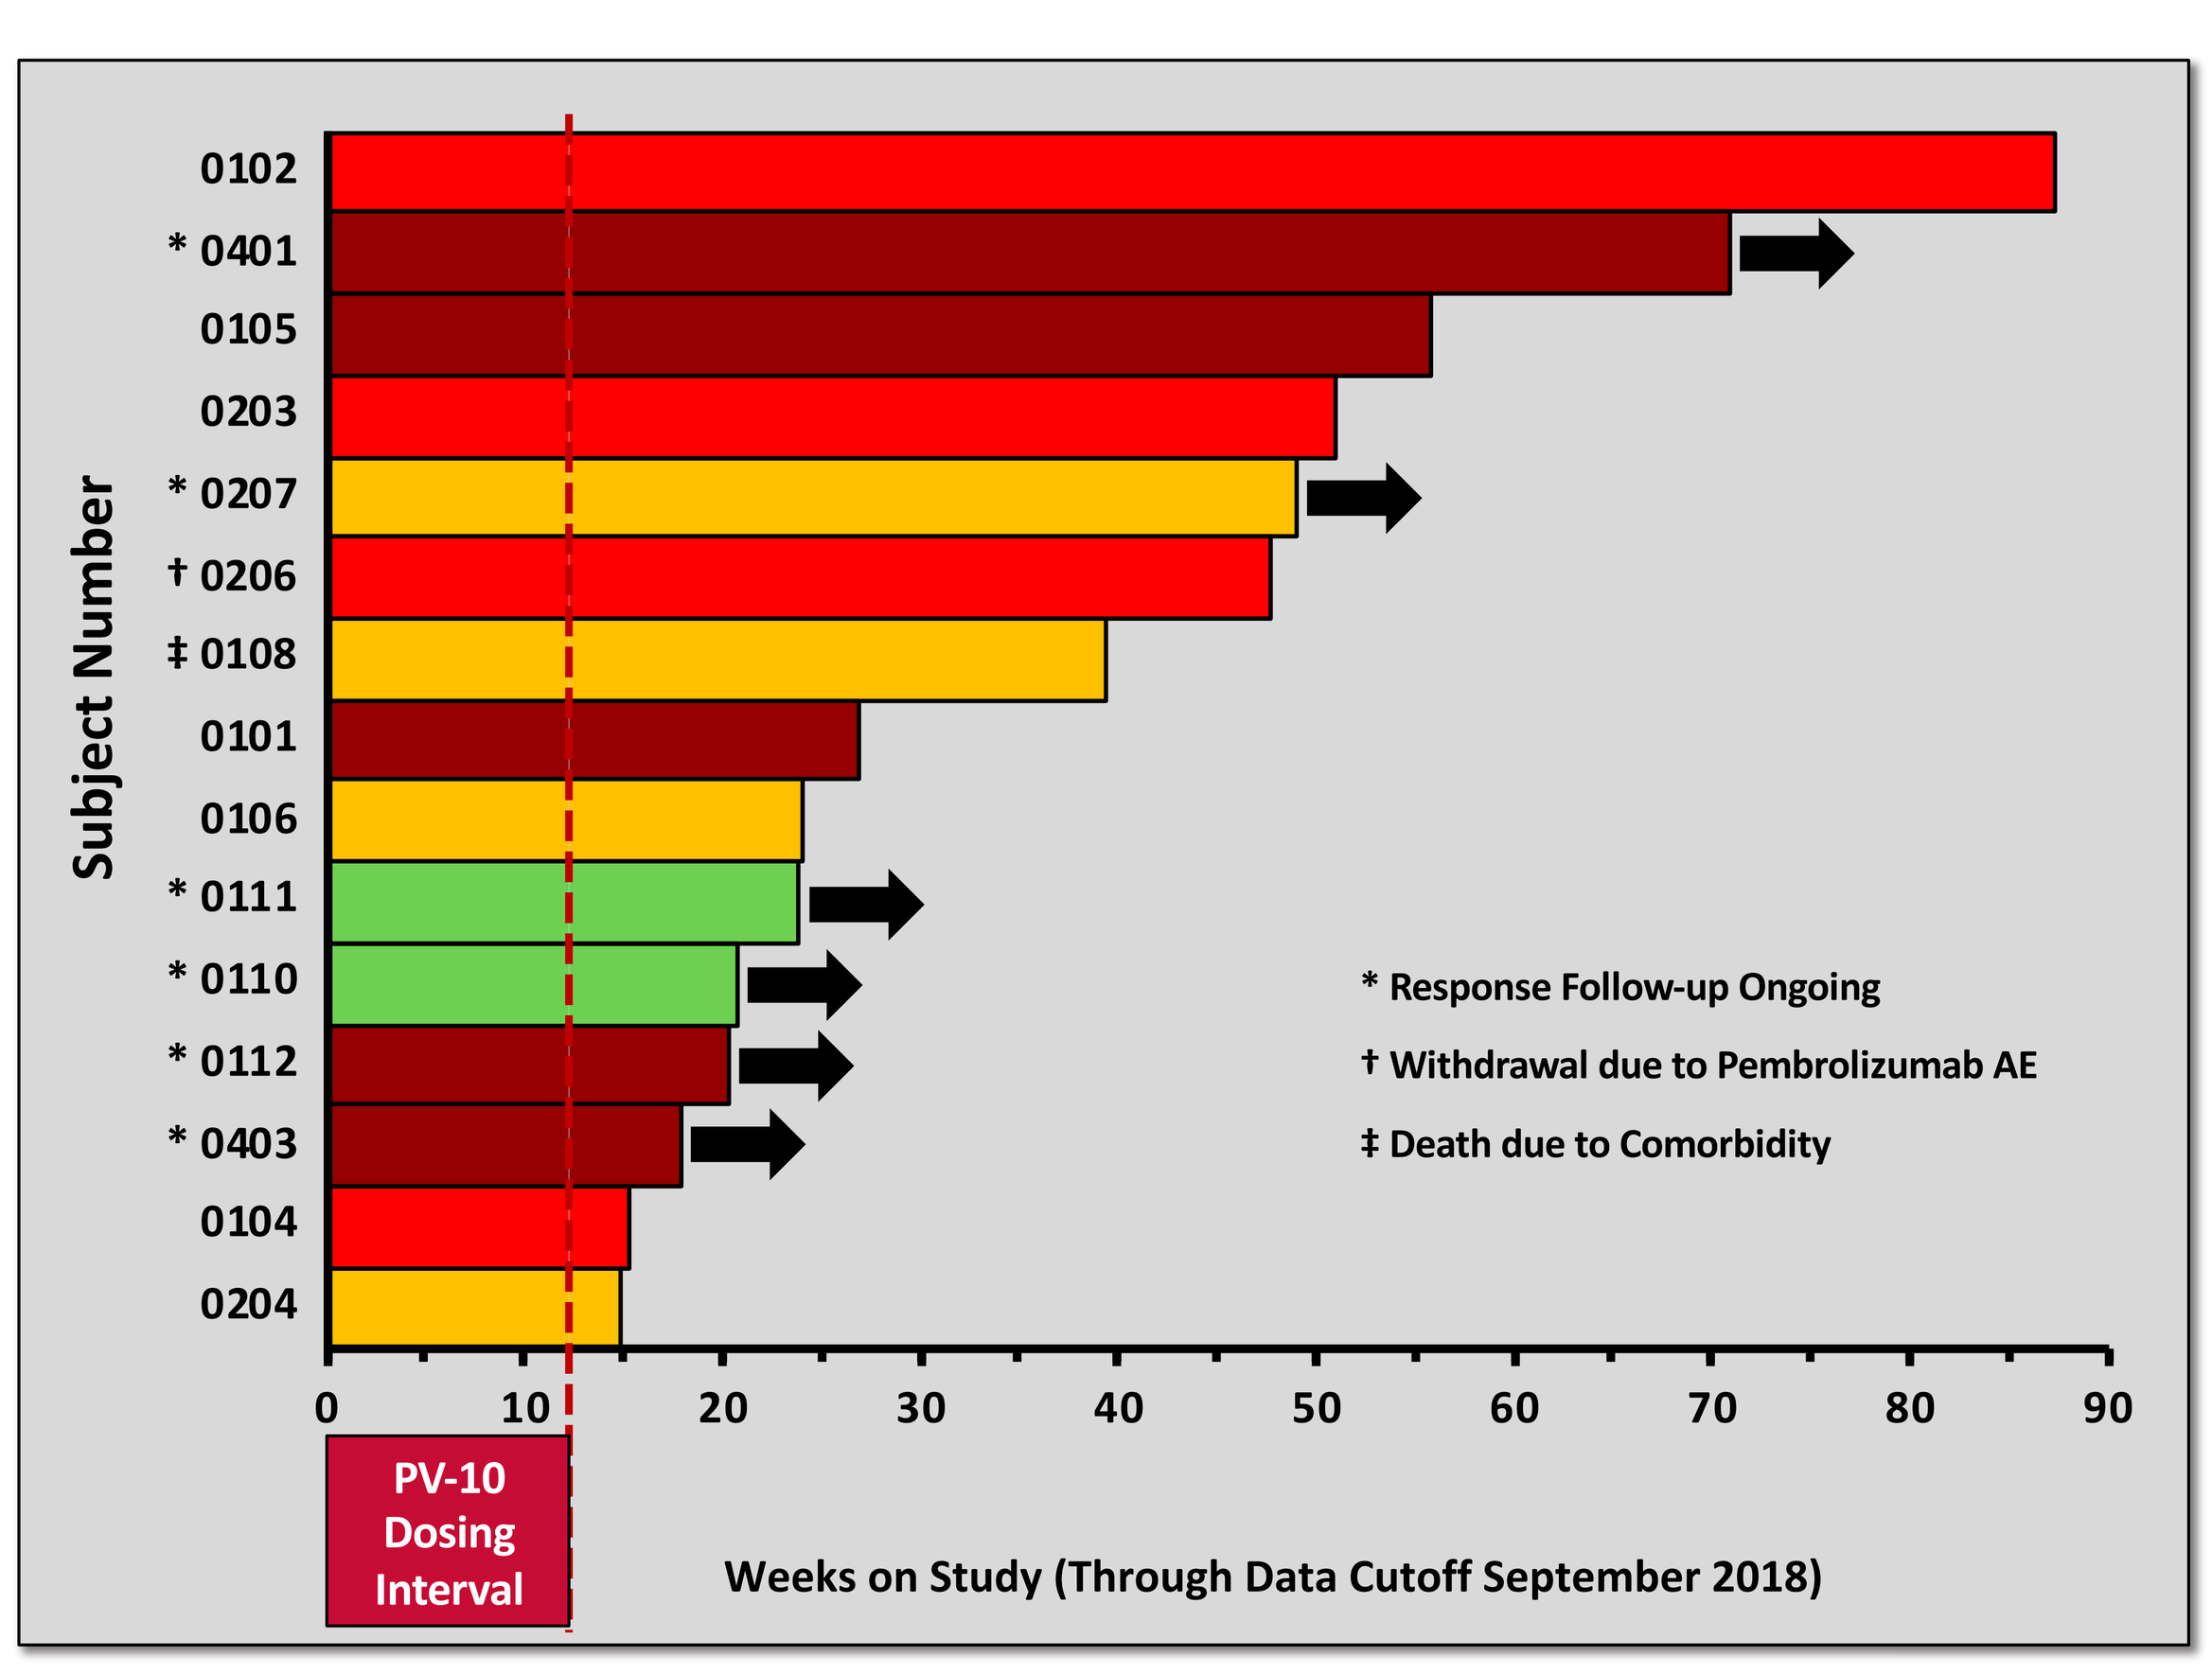

Subject Number
0102
* 0401
0105
0203
* 0207
† 0206
‡ 0108
0101
0106
* 0111
* 0110
* 0112
* 0403
0104
0204
0
10
20
30
40
50
60
70
80
90
PV-10 Dosing Interval
Weeks on Study (Through Data Cutoff September 2018)
* Response Follow-up Ongoing
† Withdrawal due to Pembrolizumab AE
‡ Death due to Comorbidity

Best Overall Response by RECIST 1.1 (Phase 1b Main Cohort ITT Population, N = 23)	All Participants (N = 23)	IIIC/IIID (N = 3)	M1a (N = 8)	M1b (N = 6)	M1c (N = 6)
Complete Response (CR)	2 (9%)	0	1	1	0
Partial Response (PR)	13 (57%)	2	3	3	5
Stable Disease (SD)	1 (4%)	0	1	0	0
Progressive Disease (PD)	5 (22%)	1	2	2	0
Not Evaluable (NE)	2 (9%)	0	1	0	1
Objective Response (CR + PR)	15 (65%)				
Clinical Benefit (CR + PR + SD)	16 (70%)				

Interim Safety Results

Treatment-Emergent Adverse Events (TEAEs) Occurring in >1 Subject, or Any Grade 3 or Higher (Phase 1b Main Cohort ITT Population, N = 23)

	TEAEs Related to PV-10: All	≥ G3	TEAEs Related to Pembrolizumab: All	≥ G3	TEAEs Related to Combination: All	≥ G3
GENERAL DISORDERS AND ADMINISTRATION SITE CONDITIONS						
Injection site pain	18	*1*	0	0	0	0
Fatigue	0	0	8	0	1	0
Injection site discharge	8	0	0	0	0	0
Injection site oedema	5	0	0	0	1	0
Injection site photosensitivity reaction	5	0	0	0	0	0
Injection site discolouration	4	0	0	0	0	0
Injection site ulcer	3	0	0	0	0	0
Injection site pruritus	2	0	0	0	2	0
Injection site haemorrhage	2	0	0	0	0	0
Injection site erythema	2	0	0	0	1	0
Influenza like illness	0	0	1	0	1	0
INJURY, POISONING AND PROCEDURAL COMPLICATIONS						
Eschar	2	0	0	0	0	0
SKIN AND SUBCUTANEOUS TISSUE DISORDERS						
Pruritus	0	0	4	*1*	0	0
Rash	0	0	4	0	0	0
Rash maculo-papular	0	0	3	0	0	0
METABOLISM AND NUTRITION DISORDERS						
Hypothyroidism	0	0	6	0	0	0
Hyperthyroidism	0	0	4	0	0	0
Hyperglycaemia	0	0	3	*1*	0	0
Hypocalcaemia	0	0	2	0	0	0
IMMUNE SYSTEM DISORDERS						
Myasthenia gravis	0	0	1	*1*	0	0
Pemphigoid	0	0	1	*1*	0	0
GASTROINTESTINAL DISORDERS						
Diarrhoea	0	0	4	0	0	0
Dry mouth	0	0	2	0	0	0
MUSCULOSKELETAL AND CONNECTIVE TISSUE DISORDERS						
Arthralgia	0	0	6	0	0	0
Neck pain	0	0	2	0	0	0
RESPIRATORY, THORACIC AND MEDIASTINAL DISORDERS						
Dyspnoea	0	0	3	0	0	0
INVESTIGATIONS						
Lymphocyte count decreased	0	0	1	*1*	0	0

AEs coded using MedDRA v21.1 for system organ class (SOC) and preferred term (PT).
Subjects with more than one occurrence of the same AE are counted once based on maximum severity.
All AEs deemed at least possibly related to PV-10 were Grade 1 or 2 except for a single occurrence of Grade 3 injection site pain.
All Grade 3 or higher AEs deemed at least possibly related to pembrolizumab were Grade 3 except for a single occurrence of exacerbation of myasthenia gravis (Grade 5).
All AEs deemed at least possibly related to the combination were Grade 1 except for a single occurrence of Grade 2 increase in TSH.

- AEs consistent with established single-agent patterns for each study drug
- No unexpected toxicities or significant overlapping toxicity

Conclusions

- Single-agent AE profiles of each study drug are maintained
- Clinical benefit upon minimal intervention with PV-10
- High ORR in Stage IV patients
- An Expansion Cohort will extend investigation to patients failing to respond on prior checkpoint inhibition