Presentation Number:
P15-1

Preliminary Results of a Phase 1b/2 Study of PV-10 and Pembrolizumab in Stage IV Melanoma

Sanjiv S. Agarwala [1], Merrick Ross [2], Jonathan Zager [3], Richard Essner [4], Keisuke Shirai [5], B.M. Smithers [6], Victoria Atkinson [6] and Eric Wachter [7]

[1] St Luke's University Hospital and Health Network, Easton, PA USA; [2] MD Anderson Cancer Center, Houston, TX USA; [3] Moffitt Cancer Center, Tampa, FL USA; [4] Cedars-Sinai Medical Center, Los Angeles, CA USA; [5] Dartmouth-Hitchcock Medical Center, Lebanon, NH USA; [6] Princess Alexandra Hospital, Brisbane, QLD AUS; and [7] Provectus Biopharmaceuticals, Inc., Knoxville, TN USA

WCM/SMR 2017 – 9th World Congress of Melanoma / 14th International Congress of the Society for Melanoma Research
Brisbane 18-21 October 2017

For additional information:
info@pvct.com

Updated Abstract and Background

PV-10 is an injectable solution of rose bengal disodium. As the first small molecule oncolytic immunotherapy, PV-10 induces immunogenic cell death resulting in tumor-specific reactivity in circulating CD8+ T cells after intralesional (IL) injection [1-3]. PV-10 has been administered to 130 melanoma patients in Phase 1 and 2, with another 180 melanoma patients receiving PV-10 under a compassionate use protocol [4-7]. It is currently in Phase 3 as a single agent for locally advanced cutaneous melanoma (Stage IIIB–IV M1a, NCT02288897). To address patients with visceral disease, PV-10 is being investigated in combination with checkpoint inhibition.

Study PV-10-MM-1201 (NCT02557321) is an international, sequential phase 1b/2 study of PV-10 in combination with pembrolizumab. Stage IV melanoma patients with at least 1 injectable lesion who are candidates for pembrolizumab are eligible. In Phase 1b, up to 24 subjects will receive the combination q3w for 5 cycles, followed by pembrolizumab q3w for up to 24 months. The primary endpoint is safety and tolerability; objective response rate and progression-free survival are key secondary endpoints (both assessed via RECIST 1.1 after 5 treatment cycles then q12w thereafter).

Among the initial 12 patients receiving PV-10 and pembrolizumab (safety population as of a September 2017 data cutoff), all Treatment-Emergent AEs (TEAEs) at least possibly related to study treatment were consistent with established patterns for each drug, with no unexpected toxicities attributable to the combination.

Initial testing indicates acceptable tolerability of the combination with no unexpected safety issues.

1. Wachter et al., Proceedings of SPIE 2002; 4620: 143. **2.** Liu et al., Oncotarget 2016; 7: 37893. **3.** Qin et al., Cell Death and Disease 2017; 8: e2584. **4.** Thompson et al., Melanoma Research 2008; 18: 405. **5.** Thompson et al., Annals Surg Oncol 2015; 22: 2135. **6.** Lippey et al., J Surg Oncol 2016; 114: 380. **7.** Foote et al., J Surg Oncol 2017; 115: 891. **8**. Weber et al. AACR 2016.

Small Molecule Oncolytic Immunotherapy

Primary Oncolytic Mechanism

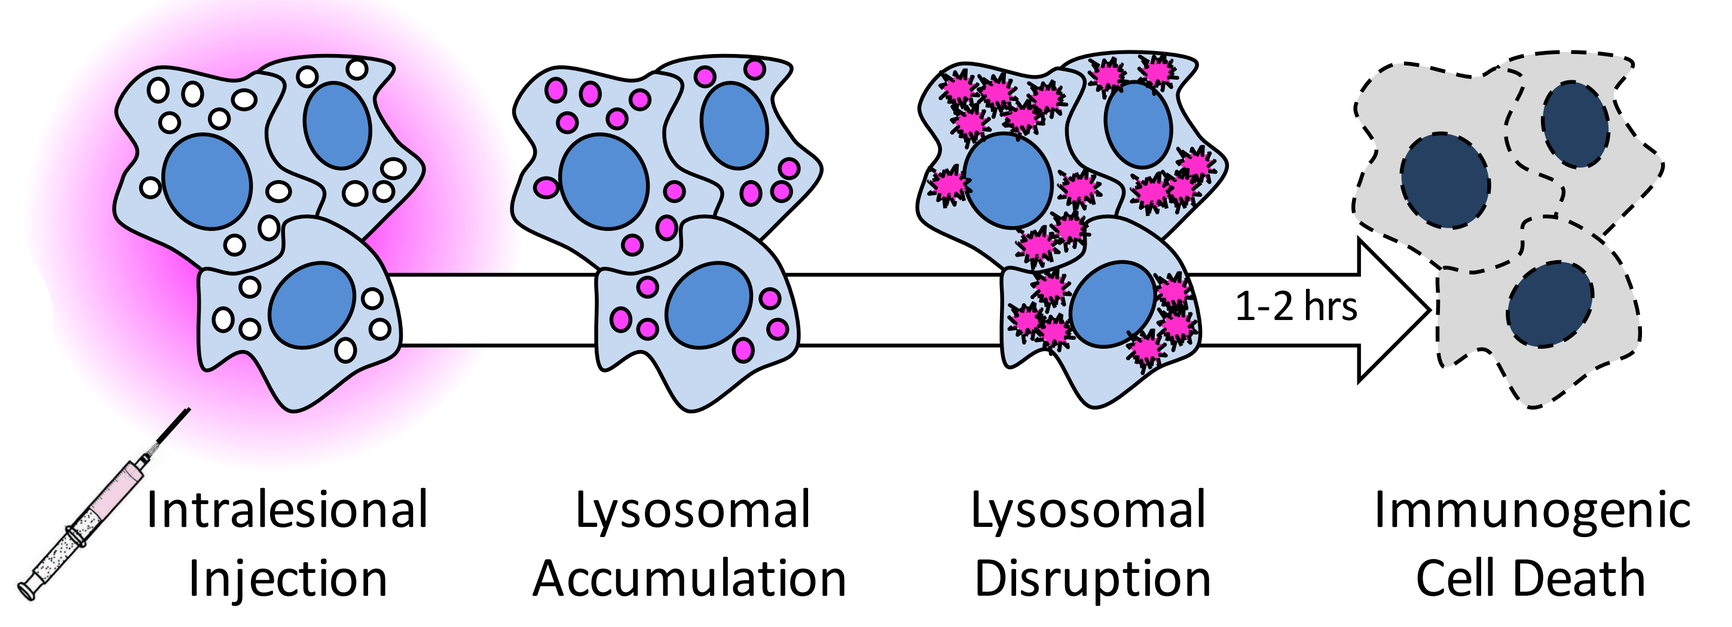

1-2 hrs
Intralesional Injection
Lysosomal Accumulation
Lysosomal Disruption
Immunogenic Cell Death

Secondary Immunomodulatory Mechanism

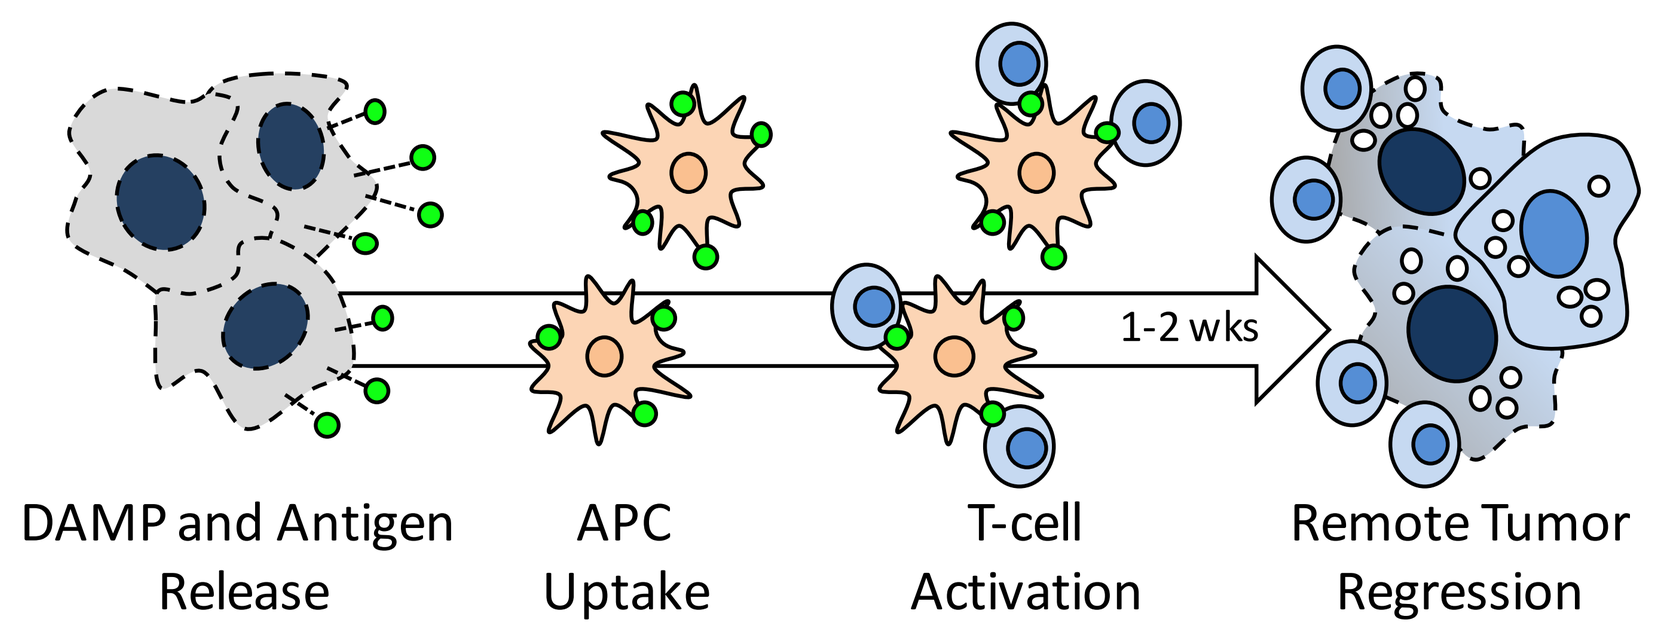

1-2 wks
DAMP and Antigen Release
APC Uptake
T-cell Activation
Remote Tumor Regression

- Functional T cell Activation in Peripheral Blood of Melanoma Patients [2]
- Immunologic Priming of PV-10 Complementary to Checkpoint Inhibition in Murine Melanoma Models [8]

Study Overview and Preliminary Clinical Data

Study Participants

Baseline Characteristics (Safety Population, N = 12)		
Gender	Male Female	11 (92%) 1 (8%)
Age	Median (Range)	71.5 (28 – 81)
Race	Black or African American White	1 11
AJCC Disease Stage	IV M1a IV M1b IV M1c	4 (33%) 4 (33%) 4 (33%)
Number of Cutaneous / Subcutaneous Lesions	Median (Range)	2.5 (1 – 40)
Prior Therapy	Checkpoint Inhibition Chemotherapy Radiotherapy Surgery Other	1 (8%) 1 (8%) 1 (8%) 11 (92%) 5 (42%)
BRAF Mutation Status	Mutant Wild-Type Not Reported	3 (25%) 2 (17%) 7 (58%)

Subject Characteristics (Safety Population, N = 12)

ID / Age / Gender	Stage	Number of CUT/SQ Lesions	Location(s) of Target Lesions	Site(s) of Non-Target Lesions
0101 / 81 / M	M1c	3	SQ (x2), Axillary LN	Liver, Lung
0102 / 47 / M	M1b	4	Scalp	Lung
0104 / 79 / M	M1b	3	LLE	Bilateral Lung
0105 / 69 / M	M1c	4	RUE	Bone, Liver, Lung
0106 / 78 / M	M1a	1	RLE (x2)	In-transit or satellite with nodal mets (N3)
0202 / 52 /M	M1a	1	Chest Wall	Chest Wall, Axillary LN
0203 / 76 / M	M1b	2	RLE (x2)	Lower Extremity and Lung
0204 / 28 / M	M1a	40	Jaw, LUE	Scalp, Face, Neck, Torso, Upper Extremity, LN (x20)
0205 / 50 / F	M1a	3	Chest Wall	Back
0206 / 73 / M	M1b	1	RUE	Lung
0401 / 70 / M	M1c	1	Lung, Shoulder	Liver, Lung
0402 / 79 / M	M1c	2	Axilla, Flank	Liver

- Safety population is all subjects receiving at least one dose of PV-10 and pembrolizumab
- PV-10 injection to all cutaneous and subcutaneous lesions (not to nodal or visceral lesions)
- All subjects had measurable Target Lesions assigned per RECIST (Subjects 0101, 0205 and 0401 had non-injected Target Lesions)
- Subjects had substantial systemic disease burden in addition to their injectable lesions

Preliminary Efficacy Results

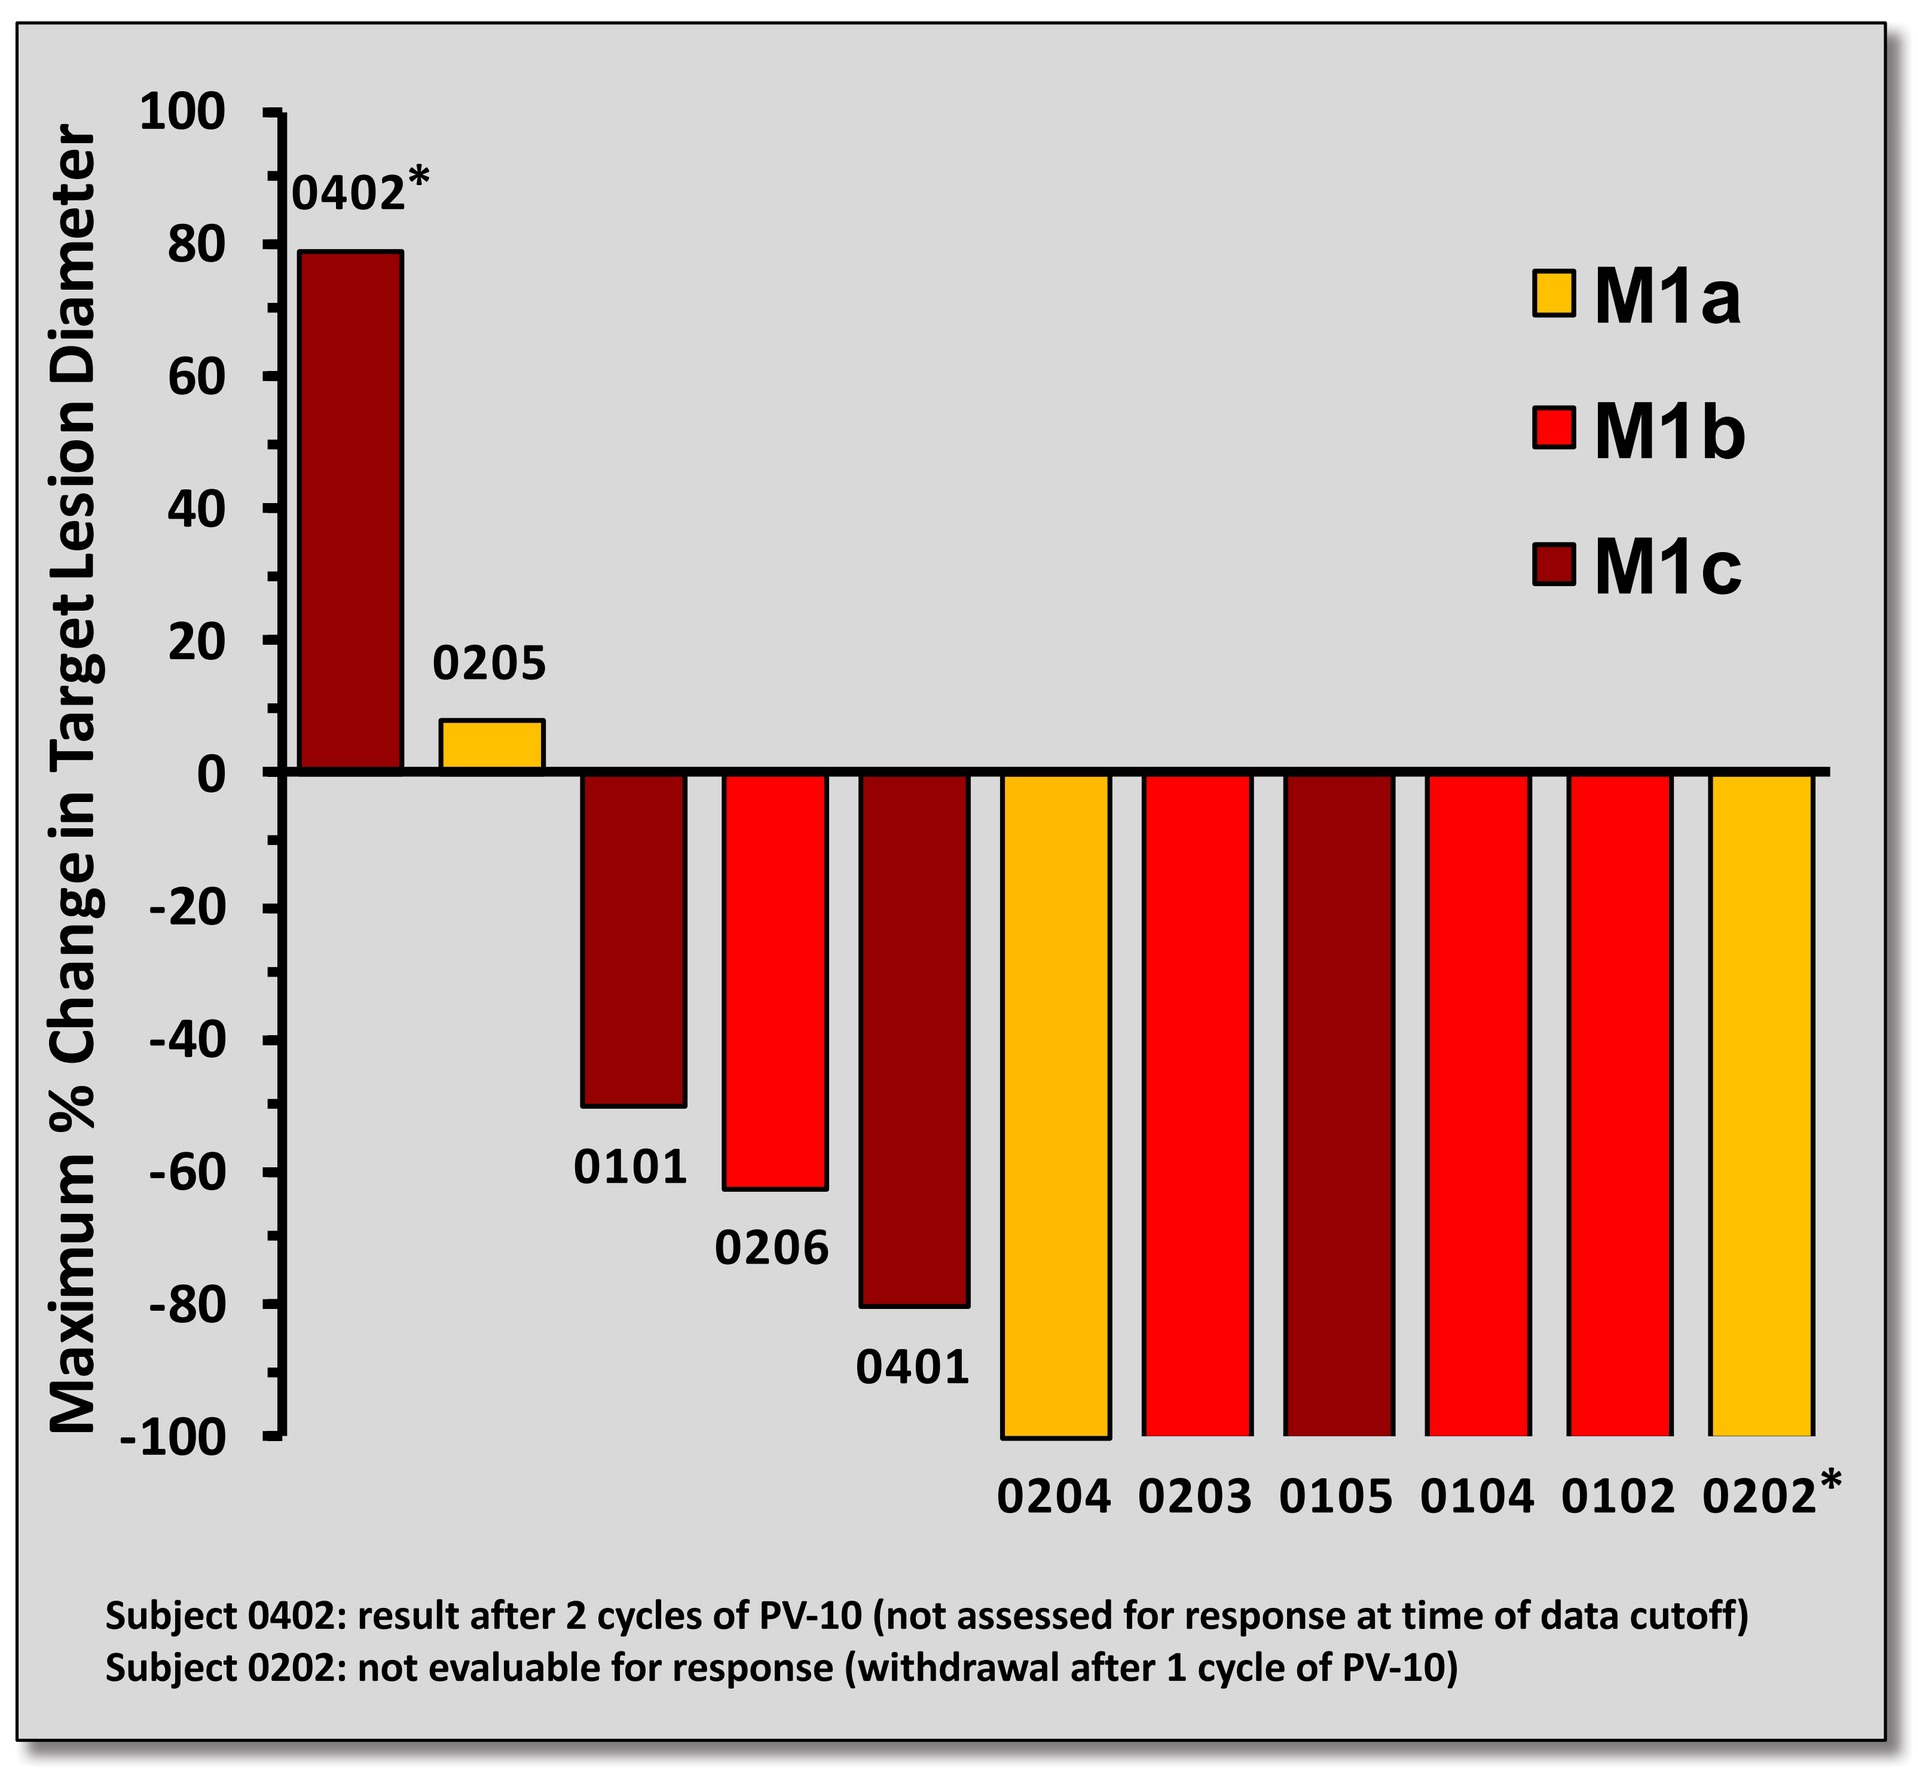

Maximum % Change in Target Lesion Diameter
100
80
60
40
20
0
-20
-40
-60
-80
-100
0402*
0205
0101
0206
0401
0204 0203 0105 0104 0102 0202*
M1a
M1b
M1c
Subject 0402: result after 2 cycles of PV-10 (not assessed for response at time of data cutoff)
Subject 0202: not evaluable for response (withdrawal after 1 cycle of PV-10)

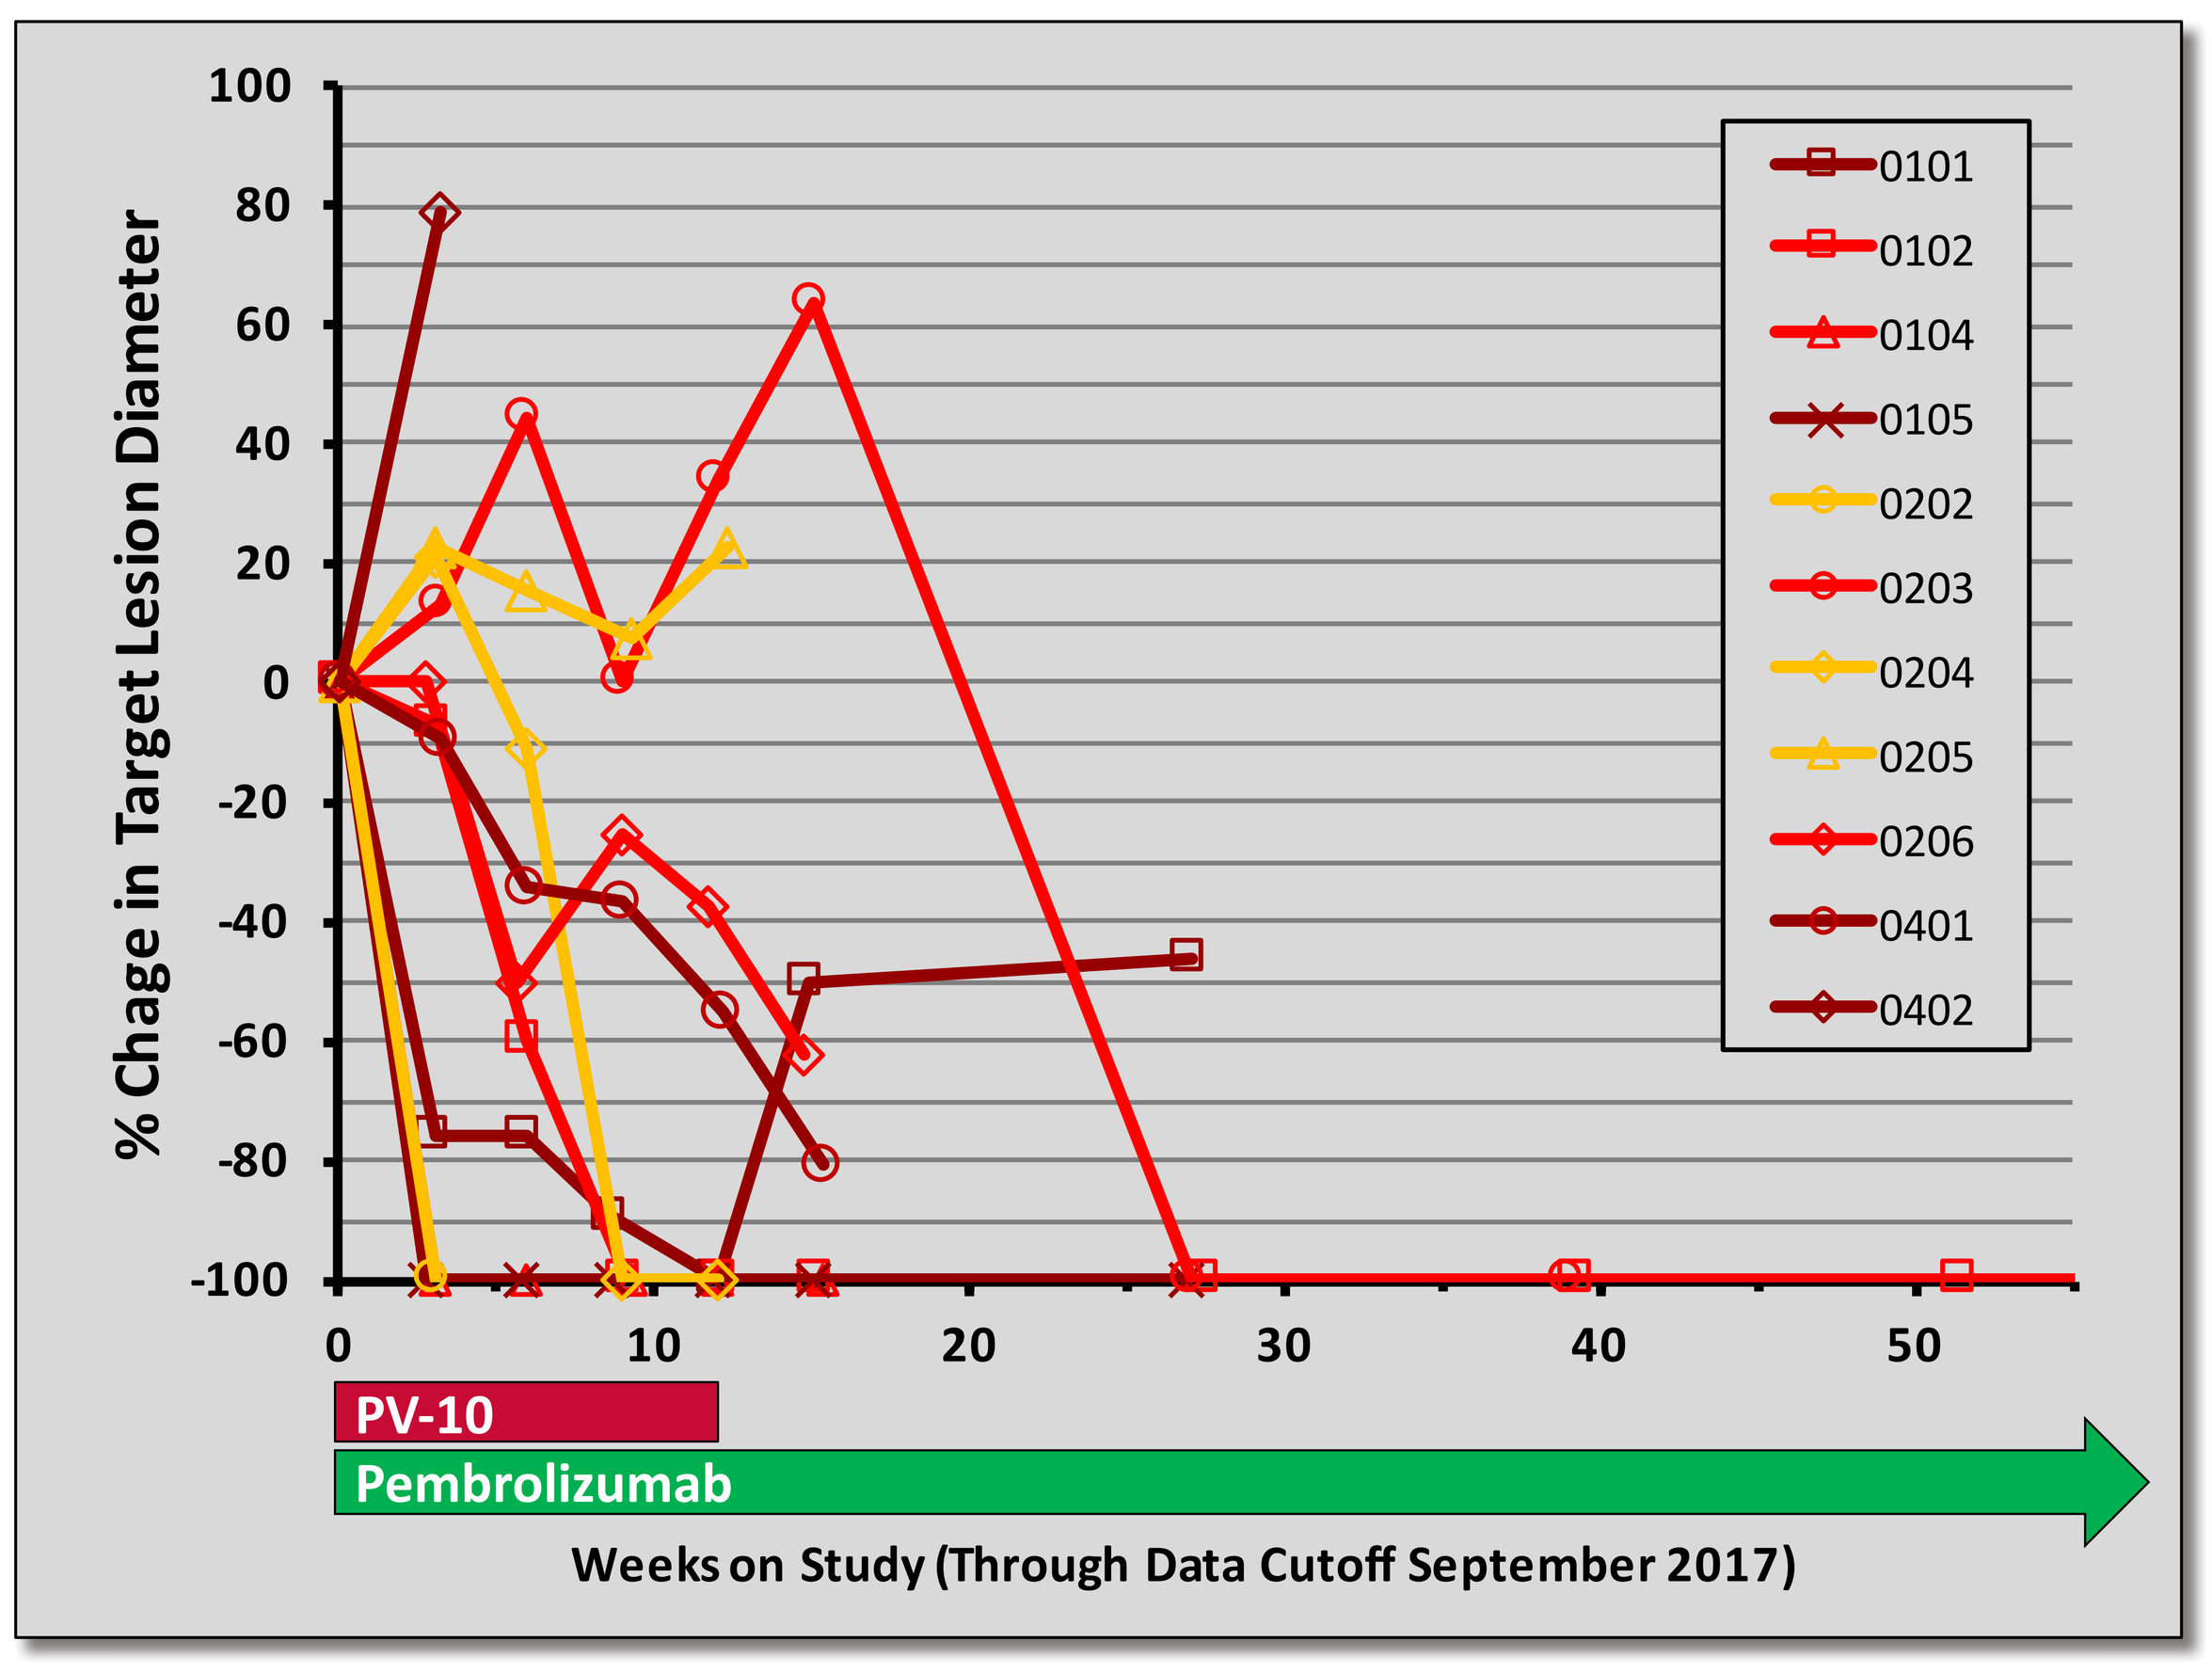

% Chage in Target Lesion Diameter
100
80
60
40
20
0
-20
-40
-60
-80
-100
0
10
20
30
40
50
0101
0102
0104
0105
0202
0203
0204
0205
0206
0401
0402
PV-10
Pembrolizumab
Weeks on Study (Through Data Cutoff September 2017)

- Most subjects had robust response in their Target Lesions
- These included injected cutaneous / subcutaneous lesions and non-injected nodal / visceral lesions

- Response of Target Lesions coincided with combination regimen
- Subjects received a median of 5 cycles of PV-10 (mean 3.8, range 1 – 5)

Best Overall Response by RECIST 1.1 (Efficacy Evaluable Population, N = 10)	All Participants (N = 10)	M1a (N = 3)	M1b (N = 4)	M1c (N = 3)
Complete Response (CR)	1 (10%)	0	0	1
Partial Response (PR)	4 (40%)	0	2	2
Stable Disease (SD)	1 (10%)	0	1	0
Progressive Disease (PD)	3 (30%)	2	1	0
Not Evaluable (NE)	1 (10%)	1	0	0
Objective Response (CR + PR)	5 (50%)			
Clinical Benefit (CR + PR + SD)	6 (60%)			

- Efficacy evaluable population is all subjects with Target and Non-Target Lesion follow-up data
- Overall response by RECIST 1.1 consistent with Target Lesion response
- Highest response observed in M1c patients

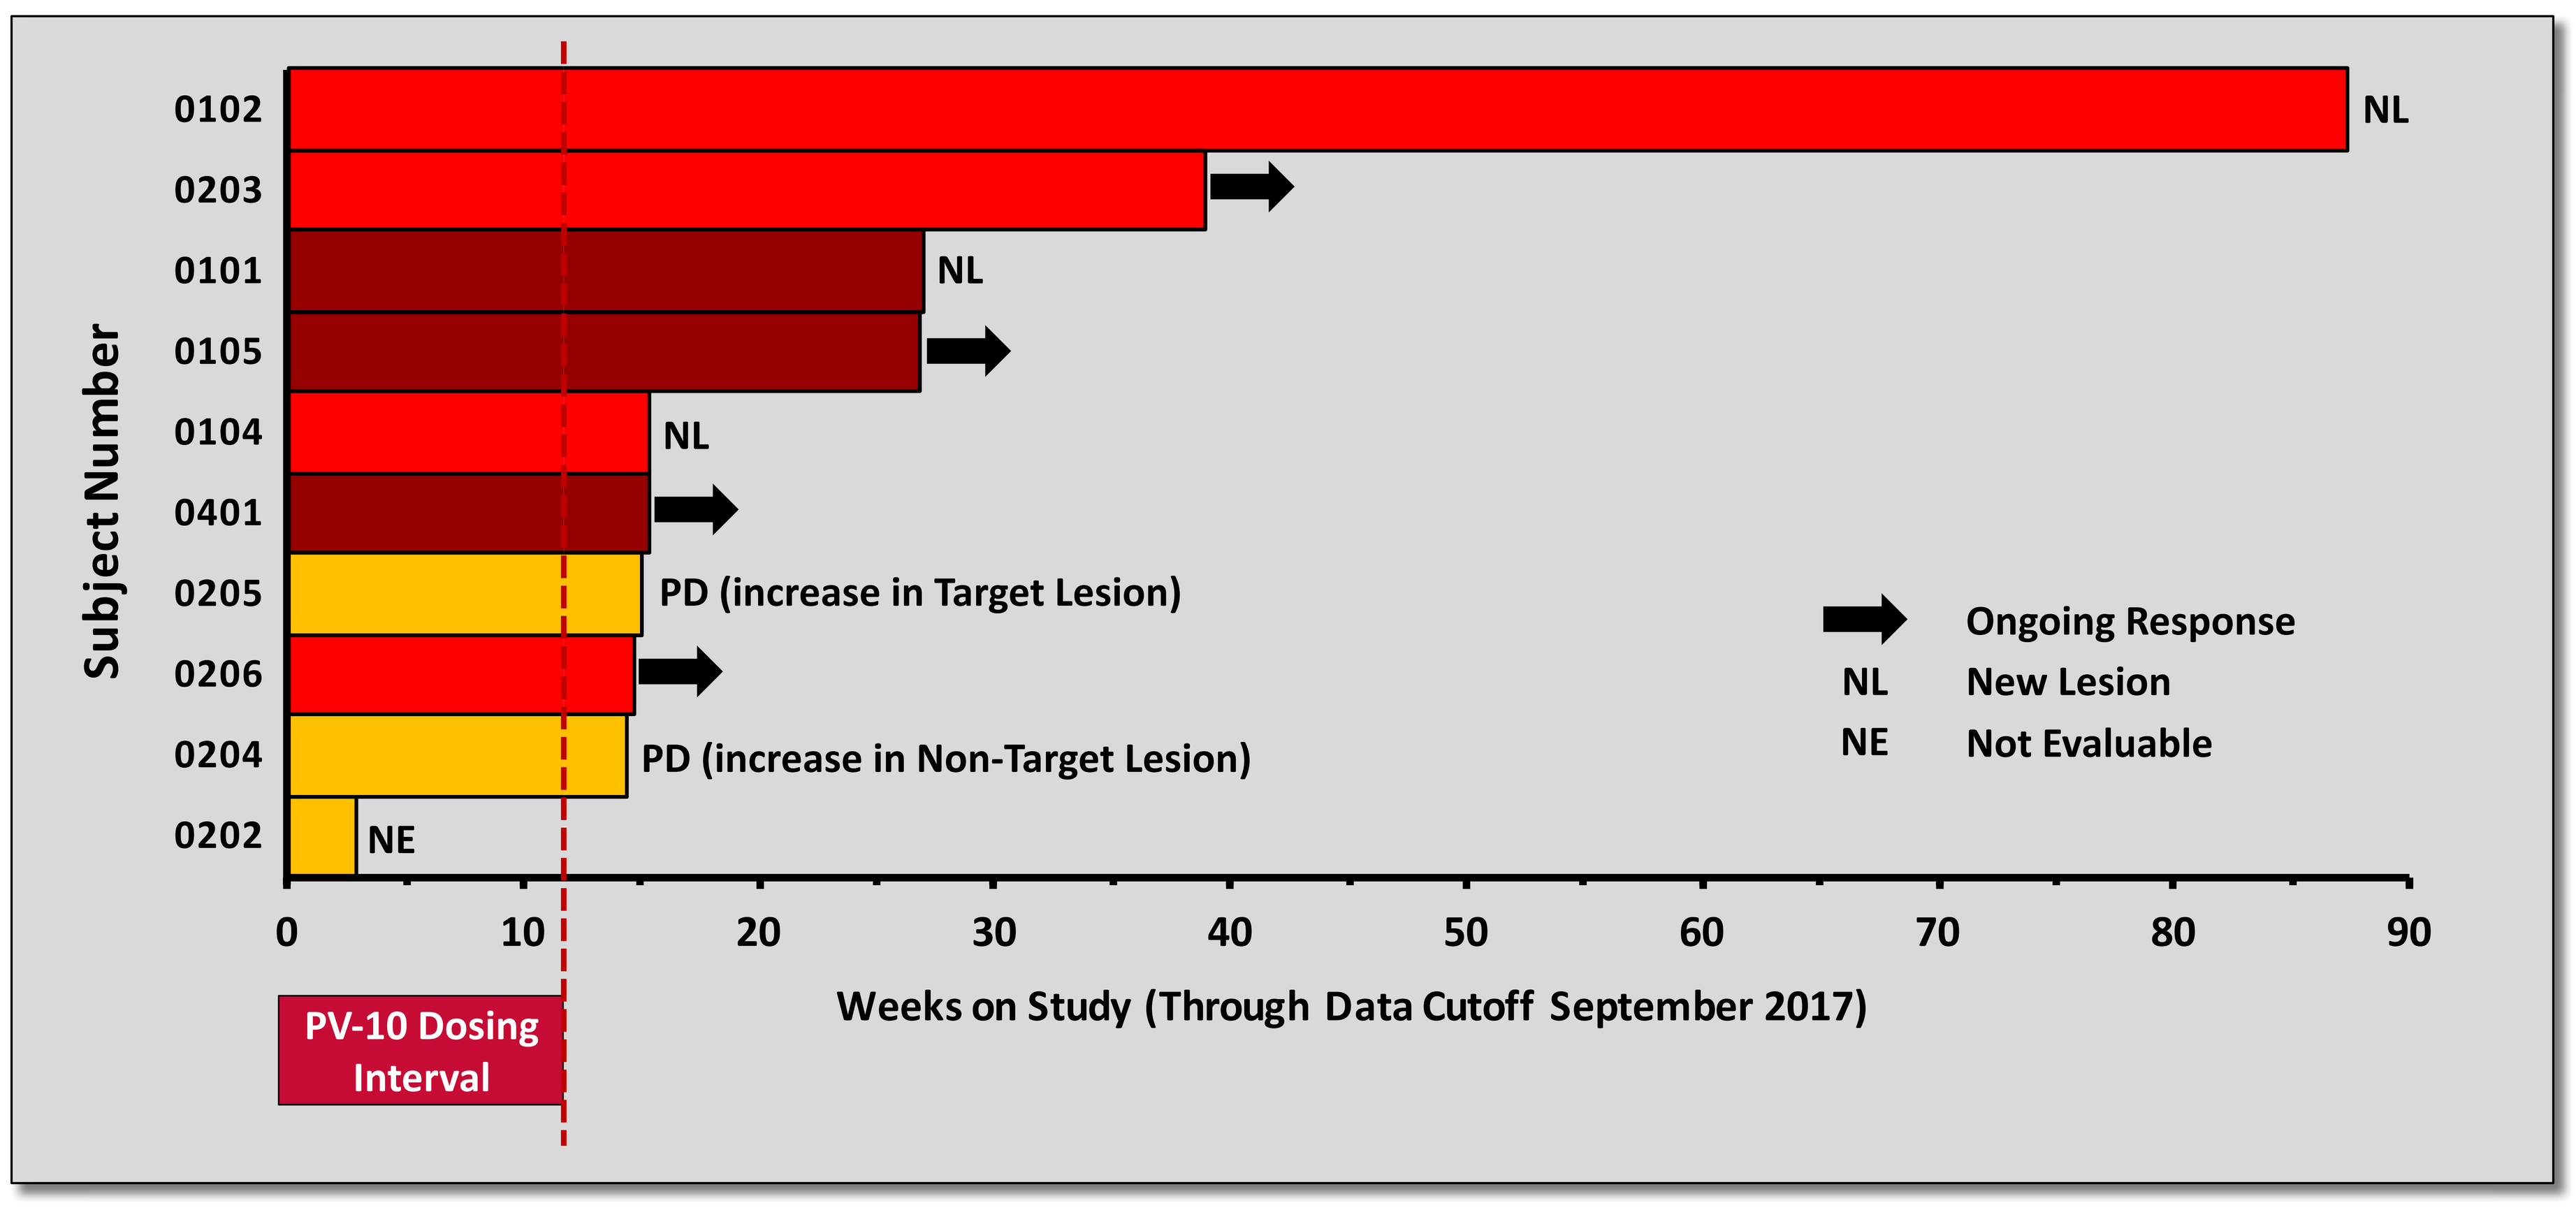

Subject Number
0102
0203
0101
0105
0104
0401
0205
0206
0204
0202
NL
NL
NL
PD (increase in Target Lesion)
PD (increase in Non-Target Lesion)
NE
Ongoing Response
NL New Lesion
NE Not Evaluable
0
10
20
30
40
50
60
70
80
90
Weeks on Study (Through Data Cutoff September 2017)
PV-10 Dosing Interval

- Primary etiology of progression was occurrence of a new lesion (NL) after final allowed cycle of PV-10
- Phase 1b study format does not permit additional PV-10 administration after week 12, thus new lesions could not be injected after this initial dosing interval

Preliminary Safety Results

Treatment-Emergent Adverse Events (TEAEs) Occurring in >1 Subject, or Any Grade 3 or Higher (Safety Population, N = 12)	All TEAEs		TEAEs Related to PV-10		TEAEs Related to Pembrolizumab	
	All	≥ G3	All	≥ G3	All	≥ G3
GENERAL DISORDERS AND ADMINISTRATION SITE CONDITIONS						
Injection site pain	7	0	7	0	0	0
Fatigue	4	0	0	0	3	0
Injection site discolouration	3	0	3	0	0	0
Chest pain	2	0	0	0	0	0
Injection site discharge	2	0	2	0	0	0
Injection site haemorrhage	2	0	2	0	0	0
Injection site photosensitivity reaction	2	0	2	0	0	0
Oedema peripheral	2	0	2	0	0	0
SKIN AND SUBCUTANEOUS TISSUE DISORDERS						
Rash	2	0	0	0	2	0
Rash maculo-papular	2	0	0	0	2	0
ENDOCRINE DISORDERS						
Hypothyroidism	2	0	0	0	2	0
GASTROINTESTINAL DISORDERS						
Diarrhoea	3	0	0	0	2	0
MUSCULOSKELETAL AND CONNECTIVE TISSUE DISORDERS						
Arthralgia	3	0	0	0	2	0
METABOLISM AND NUTRITION DISORDERS						
Hyperglycaemia	1	1	0	0	1	1 [a]
NERVOUS SYSTEM DISORDERS						
Myasthenia gravis	1	1	0	0	1	1 [a]

AEs coded using MedDRA v19.0 for system organ class (SOC) and preferred term (PT).
Subjects with more than one occurrence of the same AE are counted once based on maximum severity.
Single occurrences of decreased appetite (Grade 1) and fatigue (Grade 1) were deemed possibly related to the combination.
[a] Single occurrences of hyperglycemia (Grade 3) and exacerbation of myasthenia gravis (Grade 5) were deemed possibly related and certain related to pembrolizumab, respectively.

- AEs consistent with established patterns for each drug
- No unexpected toxicities or evidence of compounded toxicity
- One serious adverse reaction (exacerbation of myasthenia gravis) occurred after second cycle of PV-10 + pembrolizumab and was deemed related to pembrolizumab; subject had prior treatment with ipilimumab

Conclusions

- Orthogonal AE profiles of each study drug are maintained when used in combination
- Clinical benefit was observed upon minimal intervention with PV-10
- Observed response rate higher than either drug as monotherapy (especially in M1c patients)
- The data support advanced study of PV-10 with checkpoint inhibition

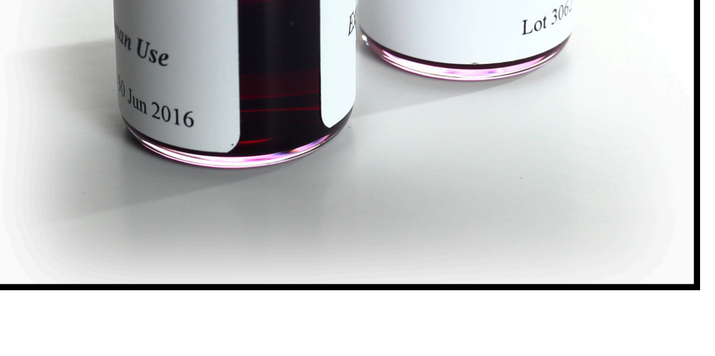